CD27 a new immuno-oncology target shaping innate and adaptive anti-tumor immune responses

5588

Yulia Ovechkina, Jessica Cross, Nathan Eyde, Emily Frazier, Neda Kabi, Chen Katz, Remington Lance, Kurt Lustig, David Peckham, Shaarwari Sridhar, Carla Talbaux, Isabelle Tihista, Mei Xu, Shawn Iadonato and Thierry Guillaudeux

Kineta Inc. 219 Terry Av. North, Seattle, WA 98109.

tguillaudeux@kineta.us

Introduction

- CD27 is highly expressed on CD4+ and CD8+ T cells as well as NK and NKT cells.
- CD27 plays a key role in T cell co-activation, proliferation and differentiation after stimulation with CD70.
- The costimulatory signal of CD27 on T cells is mediated via the NFκB pathway but also via the phosphatidylinositol 3 kinase and the protein kinase B pathways.
- CD27/CD70 co-stimulation has the potential to boost anti-tumor immunity via T-cell activation, clonal expansion and enhanced differentiation into antigen specific cytotoxic and memory T cells.
- CD27/CD70 also has the capability to influence the innate immune response via direct activation of NK cells and a subsequent secretion of interferon-gamma (IFN-γ).
- CD27 is a promising target for antitumor therapy to drive specific T cell anti-tumor responses.

Objectives

- Select 1 or 2 antibodies per clade from our library of 147 highly diverse fully human anti-CD27 monoclonal antibodies (Cancer Immunol Res; 2022; 10; 1_Supplement: P035).
- Measure human anti-CD27 mAb affinity to human CD27.
- Evaluate anti-CD27 mAbs cross-reactivity to human, non-human primate and murine CD27.
- Assess anti-CD27 mAbs agonist activity using NFκB-Luc2/CD27 Jurkat reporter assay.
- Characterize the stimulatory effects of agonist anti-CD27 mAbs on human T and NK cells.

Kineta's anti-CD27 mAbs demonstrate a range of binding affinities to human CD27 by ELISA

Antibody	EC50 (ug/mL)	EC50 (nM)
CD27_70.10	0.04	0.28
CD27_93.10	0.04	0.29
CD27_53.10	0.04	0.29
CD27_29.10	0.04	0.29
CD27_30.10	0.05	0.30
CD27_106.10	0.05	0.35
CD27_62.10	0.05	0.35
CD27_10.10	0.06	0.37
CD27_8.10	0.06	0.38
CD27_88.10	0.06	0.38
CD27_18.10	0.06	0.41
CD27_26.10	0.06	0.42
CD27_6.10	0.07	0.43
Varlilumab	0.07	0.43
CD27_7.10	0.08	0.53
CD27_23.10	0.08	0.54
CD27_33.10	0.09	0.57
CD27_132.10	0.09	0.57
CD27_34.10	0.09	0.60
CD27_92.10	0.12	0.80
CD27_123.10	1.9	13
CD27_20.10	5.8	39
CD27_79.10	23	150
CD27_115.10	32	210
CD27_2.10	97	>600
CD27_122.10	>100	>600
Cont. Isotype	>100	>600

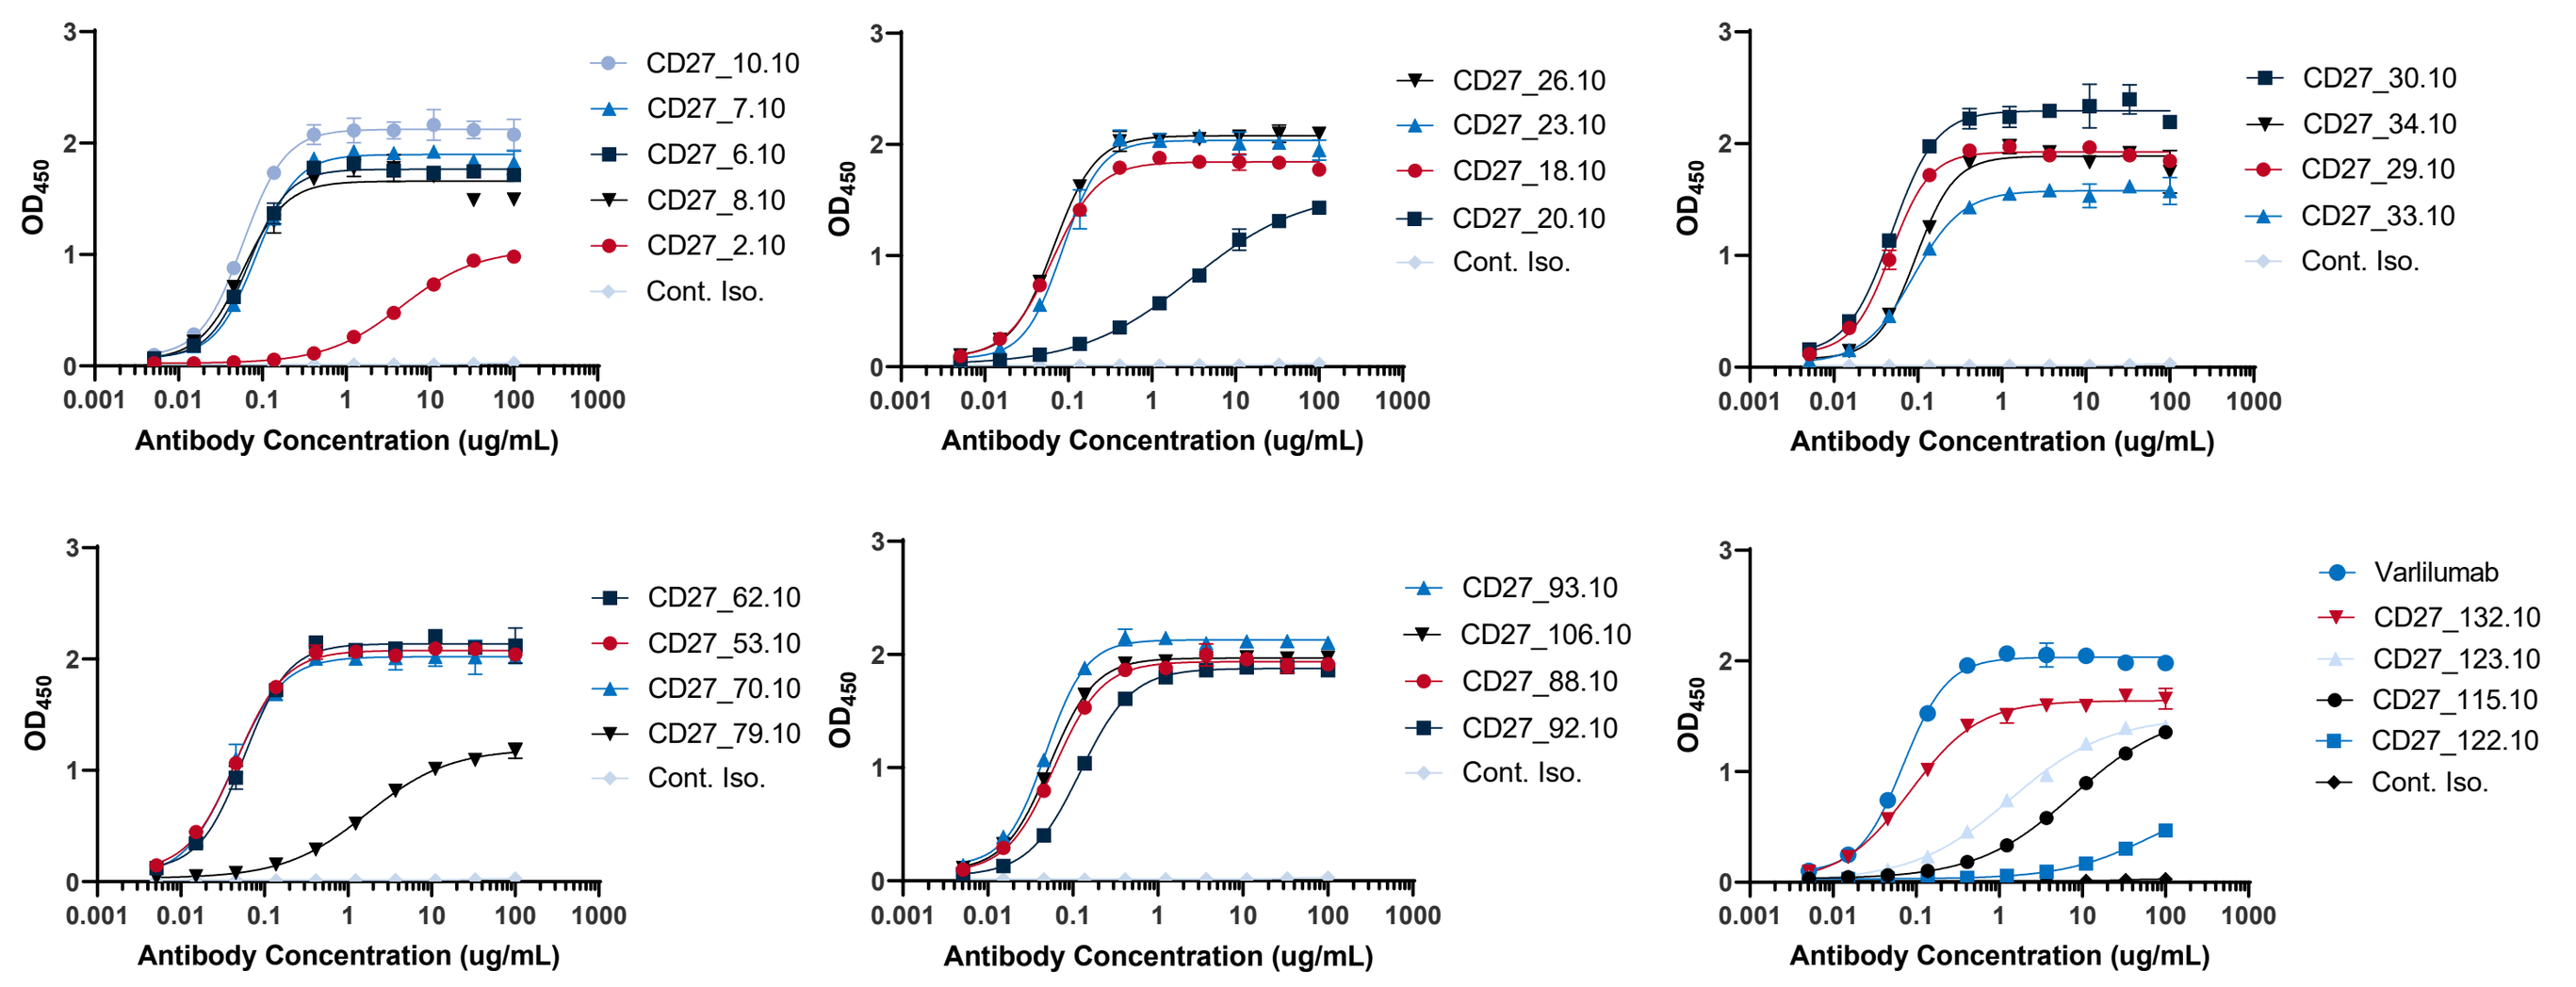
OD450
Antibody Concentration (ug/mL)
CD27_10.10
CD27_7.10
CD27_6.10
CD27_8.10
CD27_2.10
CD27_26.10
CD27_23.10
CD27_18.10
CD27_20.10
CD27_30.10
CD27_34.10
CD27_29.10
CD27_33.10
CD27_62.10
CD27_53.10
CD27_70.10
CD27_79.10
CD27_93.10
CD27_106.10
CD27_88.10
CD27_92.10
Varlilumab
CD27_132.10
CD27_123.10
CD27_115.10
CD27_122.10
Cont. Iso.

Figure 1. ELISA binding studies were conducted with immobilized human CD27 (rabbit-Fc). Binding of anti-CD27 mAbs was detected using biotinylated anti-human IgG (light chain) followed by streptavidin HRP.

Kineta's anti-CD27 mAbs demonstrate a range of binding affinities to human CD27 by Octet

Figure 2. Kinetic studies were performed using 1xKB (kinetic buffer, pH 7.2) and human CD27-ECD-His-tag loaded onto HIS1K dip and read biosensors at a concentration of 2 ug/mL with the Sartorius Octet K2 system. The loaded biosensors were transferred to wells containing investigational CD27 mAbs at a 30 nM concentration for 180 seconds to measure the affinity between CD27 and anti-CD27 mAbs. Associated biosensors were then dipped in 1xKB for 500 seconds to measure dissociation. A double reference subtraction was performed to measure non-specific binding to each biosensor with 1xKB and isotype control mAb to a CD27 loaded biosensor. A 1:1 global curve fitting analysis was performed to determine ka, kdis, and KD.

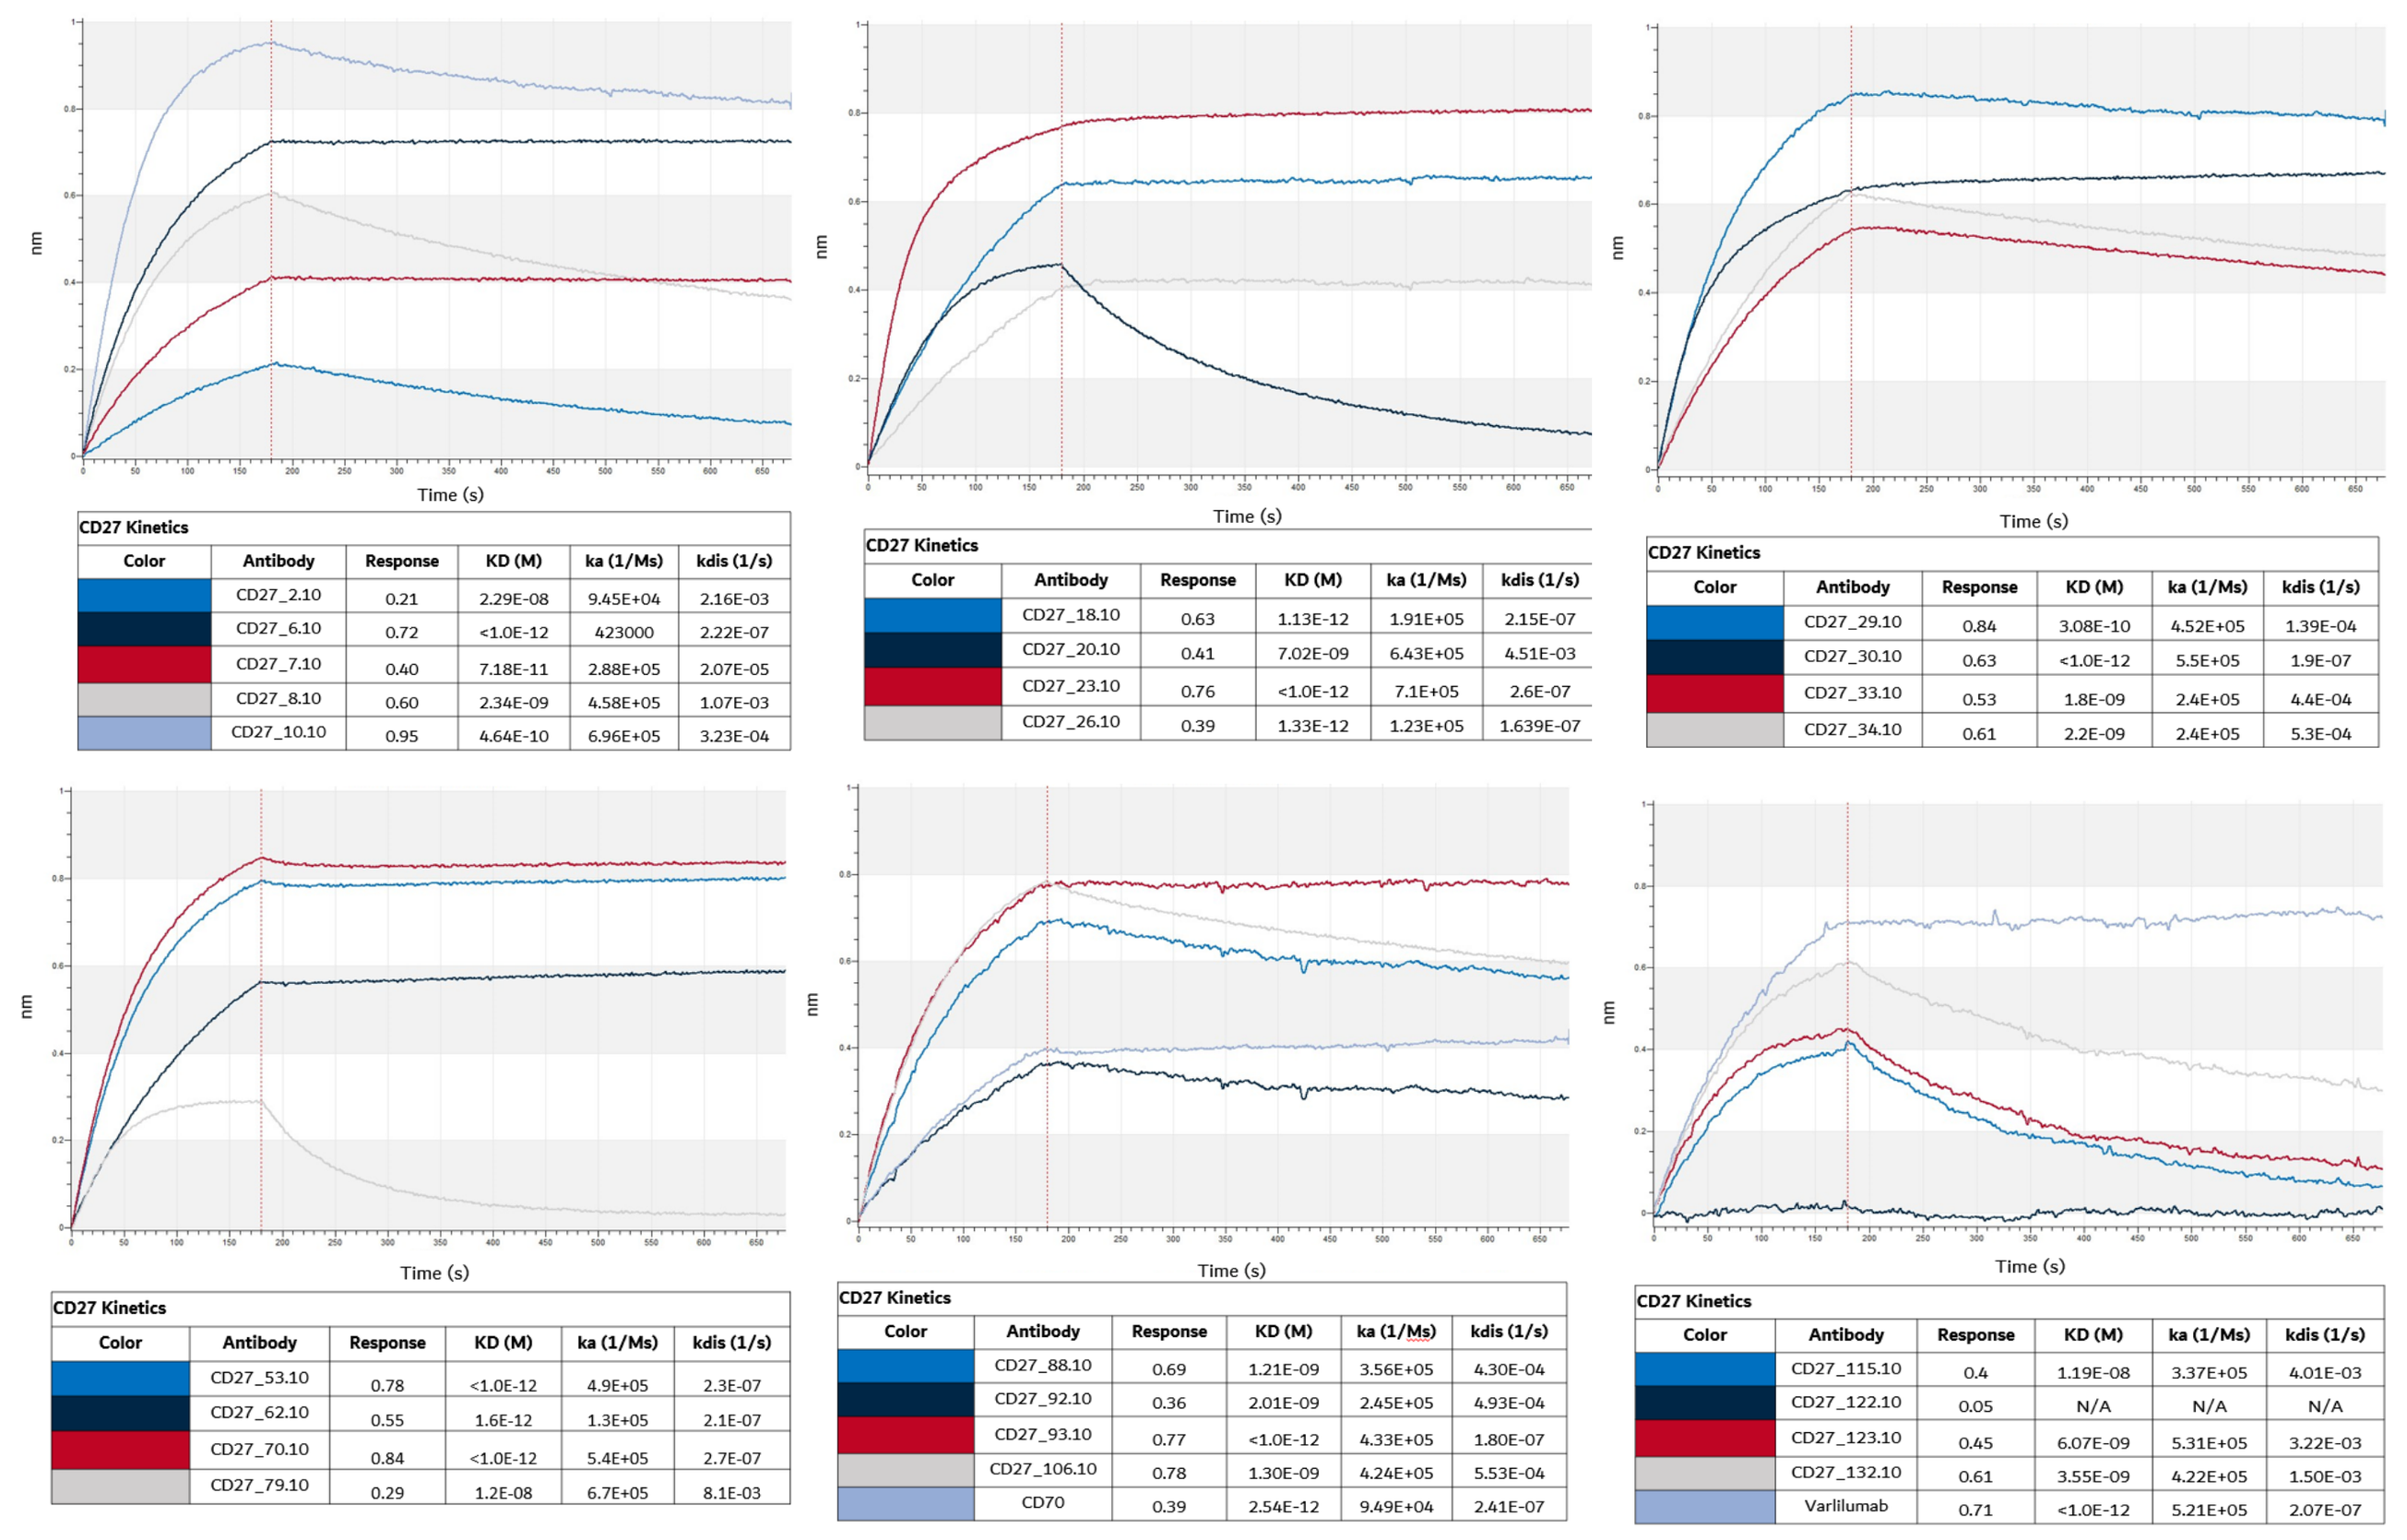
nm
Time (s)

CD27 Kinetics

Color	Antibody	Response	KD (M)	ka (1/Ms)	kdis (1/s)
	CD27_2.10	0.21	2.29E-08	9.45E+04	2.16E-03
	CD27_6.10	0.72	<1.0E-12	423000	2.22E-07
	CD27_7.10	0.40	7.18E-11	2.88E+05	2.07E-05
	CD27_8.10	0.60	2.34E-09	4.58E+05	1.07E-03
	CD27_10.10	0.95	4.64E-10	6.96E+05	3.23E-04

CD27 Kinetics

Color	Antibody	Response	KD (M)	ka (1/Ms)	kdis (1/s)
	CD27_18.10	0.63	1.13E-12	1.91E+05	2.15E-07
	CD27_20.10	0.41	7.02E-09	6.43E+05	4.51E-03
	CD27_23.10	0.76	<1.0E-12	7.1E+05	2.6E-07
	CD27_26.10	0.39	1.33E-12	1.23E+05	1.639E-07

CD27 Kinetics

Color	Antibody	Response	KD (M)	ka (1/Ms)	kdis (1/s)
	CD27_29.10	0.84	3.08E-10	4.52E+05	1.39E-04
	CD27_30.10	0.63	<1.0E-12	5.5E+05	1.9E-07
	CD27_33.10	0.53	1.8E-09	2.4E+05	4.4E-04
	CD27_34.10	0.61	2.2E-09	2.4E+05	5.3E-04

CD27 Kinetics

Color	Antibody	Response	KD (M)	ka (1/Ms)	kdis (1/s)
	CD27_53.10	0.78	<1.0E-12	4.9E+05	2.3E-07
	CD27_62.10	0.55	1.6E-12	1.3E+05	2.1E-07
	CD27_70.10	0.84	<1.0E-12	5.4E+05	2.7E-07
	CD27_79.10	0.29	1.2E-08	6.7E+05	8.1E-03

CD27 Kinetics

Color	Antibody	Response	KD (M)	ka (1/Ms)	kdis (1/s)
	CD27_88.10	0.69	1.21E-09	3.56E+05	4.30E-04
	CD27_92.10	0.36	2.01E-09	2.45E+05	4.93E-04
	CD27_93.10	0.77	<1.0E-12	4.33E+05	1.80E-07
	CD27_106.10	0.78	1.30E-09	4.24E+05	5.53E-04
	CD70	0.39	2.54E-12	9.49E+04	2.41E-07

CD27 Kinetics

Color	Antibody	Response	KD (M)	ka (1/Ms)	kdis (1/s)
	CD27_115.10	0.4	1.19E-08	3.37E+05	4.01E-03
	CD27_122.10	0.05	N/A	N/A	N/A
	CD27_123.10	0.45	6.07E-09	5.31E+05	3.22E-03
	CD27_132.10	0.61	3.55E-09	4.22E+05	1.50E-03
	Varlilumab	0.71	<1.0E-12	5.21E+05	2.07E-07

Results

Kineta's anti-CD27 mAbs demonstrate a range of binding affinities on CD27 expressing Raji cells

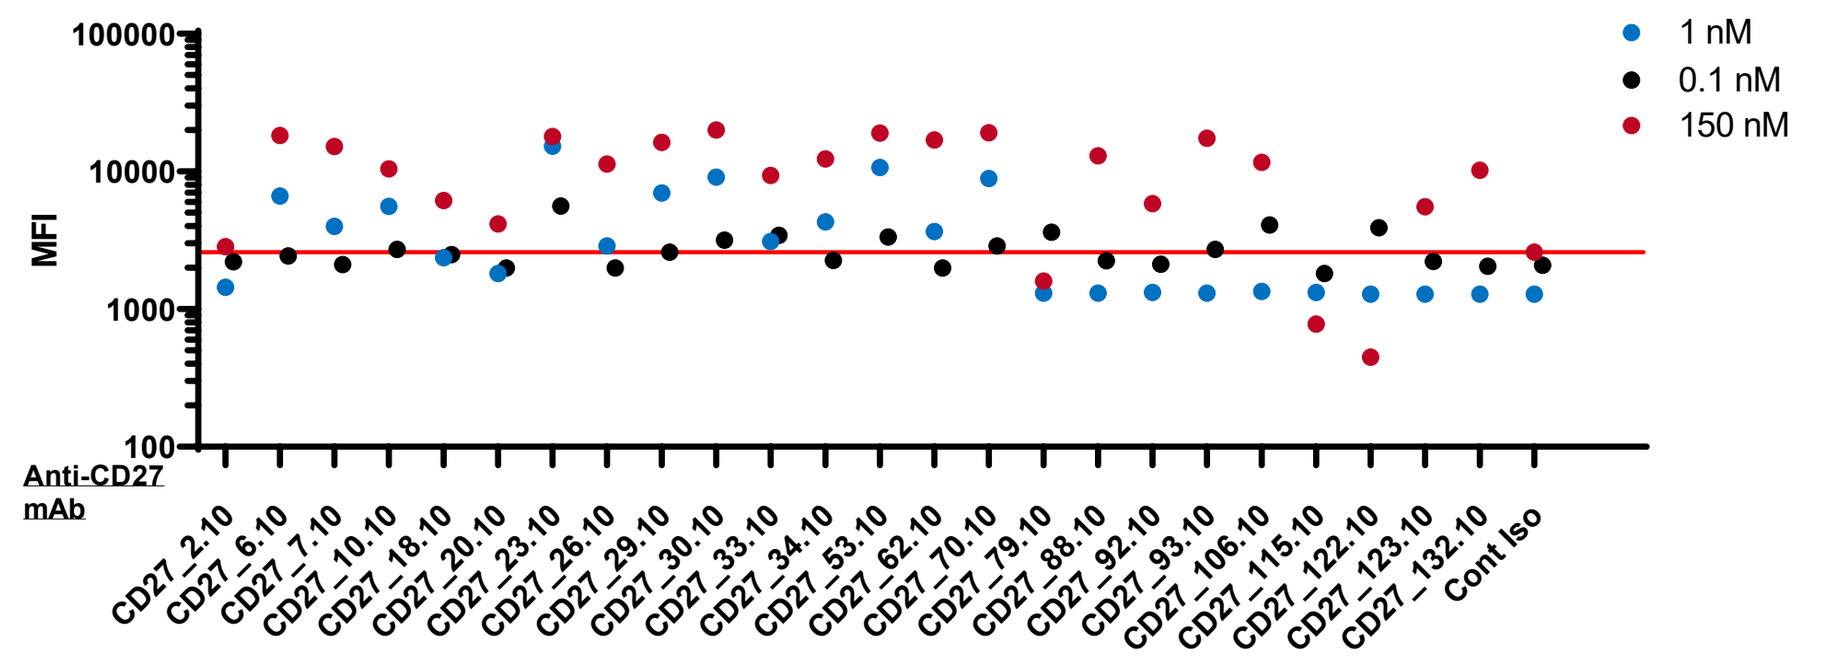
MFI
Anti-CD27 mAb
1 nM
0.1 nM
150 nM
CD27_2.10
CD27_6.10
CD27_7.10
CD27_10.10
CD27_18.10
CD27_20.10
CD27_23.10
CD27_26.10
CD27_29.10
CD27_30.10
CD27_33.10
CD27_34.10
CD27_53.10
CD27_62.10
CD27_70.10
CD27_79.10
CD27_88.10
CD27_92.10
CD27_93.10
CD27_106.10
CD27_115.10
CD27_122.10
CD27_123.10
CD27_132.10
Cont Iso

Figure 3. The anti-CD27 mAb cell binding assay was performed using CD27-positive Raji cells treated with hFc block buffer. Raji cells were incubated with anti-CD27 mAbs at 4°C for 30 min, washed and stained with R-Phycoerythrin AffiniPure F(ab')₂ fragment anti-hIgG. Antibody binding was measured by Attune Nxt Flow Cytometer. Mean fluorescence intensity (MFI) plotted against concentration of antibody.

Kineta's anti-CD27 mAbs cross-react with NHP-CD27 but not with mouse-CD27

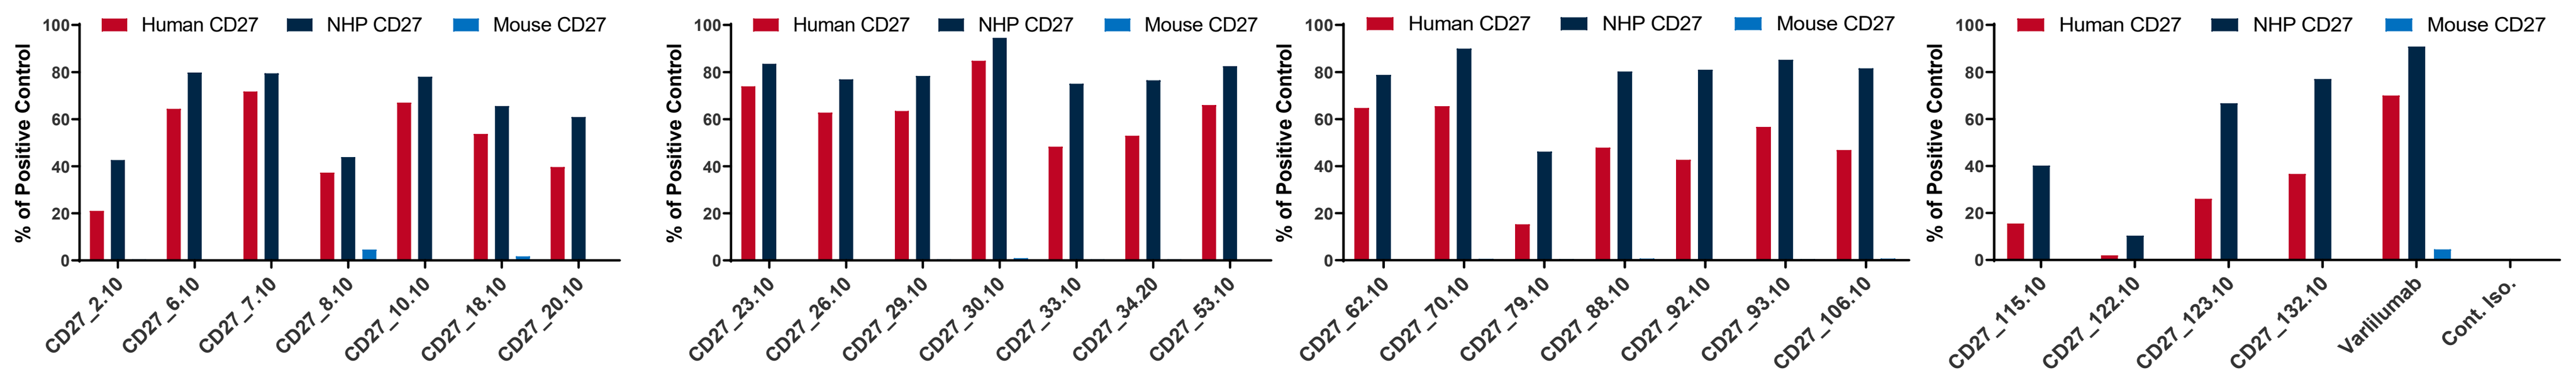
Human CD27
NHP CD27
Mouse CD27
% of Positive Control

Figure 4. Binding of anti-CD27 mAbs to immobilized human CD27 (rabbit-Fc), non-human primate (NHP) CD27 (human-Fc), or mouse CD27 (mouse-Fc). Bound anti-CD27 mAbs were detected using biotinylated anti-human IgG (light chain) followed by streptavidin HRP.

Selected Kineta's anti-CD27 mAbs induce strong agonist activity

Antibody	EC50 (ug/mL)	EC50 (nM)
CD70	0.3	2.2
CD27_88.10	5.5	37
CD27_20.10	9.9	66
CD27_33.10	30	200
CD27_8.10	42	280
CD27_18.10	61	407
CD27_29.10	88	587
CD27_2.10	>104	>693
CD27_6.10	>104	>693
CD27_7.10	>104	>693
CD27_23.10	>104	>693
CD27_30.10	>104	>693
CD27_34.10	>104	>693
CD27_53.10	>104	>693
CD27_62.10	>104	>693
CD27_70.10	>104	>693
CD27_79.10	>104	>693
CD27_92.10	>104	>693
CD27_93.10	>104	>693
CD27_106.10	>104	>693
CD27_115.10	>104	>693
CD27_122.10	>104	>693
CD27_123.10	>104	>693
CD27_132.10	>104	>693
CD27_10.10	>208	>1390
CD27_26.10	>208	>1390
Varlilumab	>208	>1390
Cont. Iso.	>104	>693

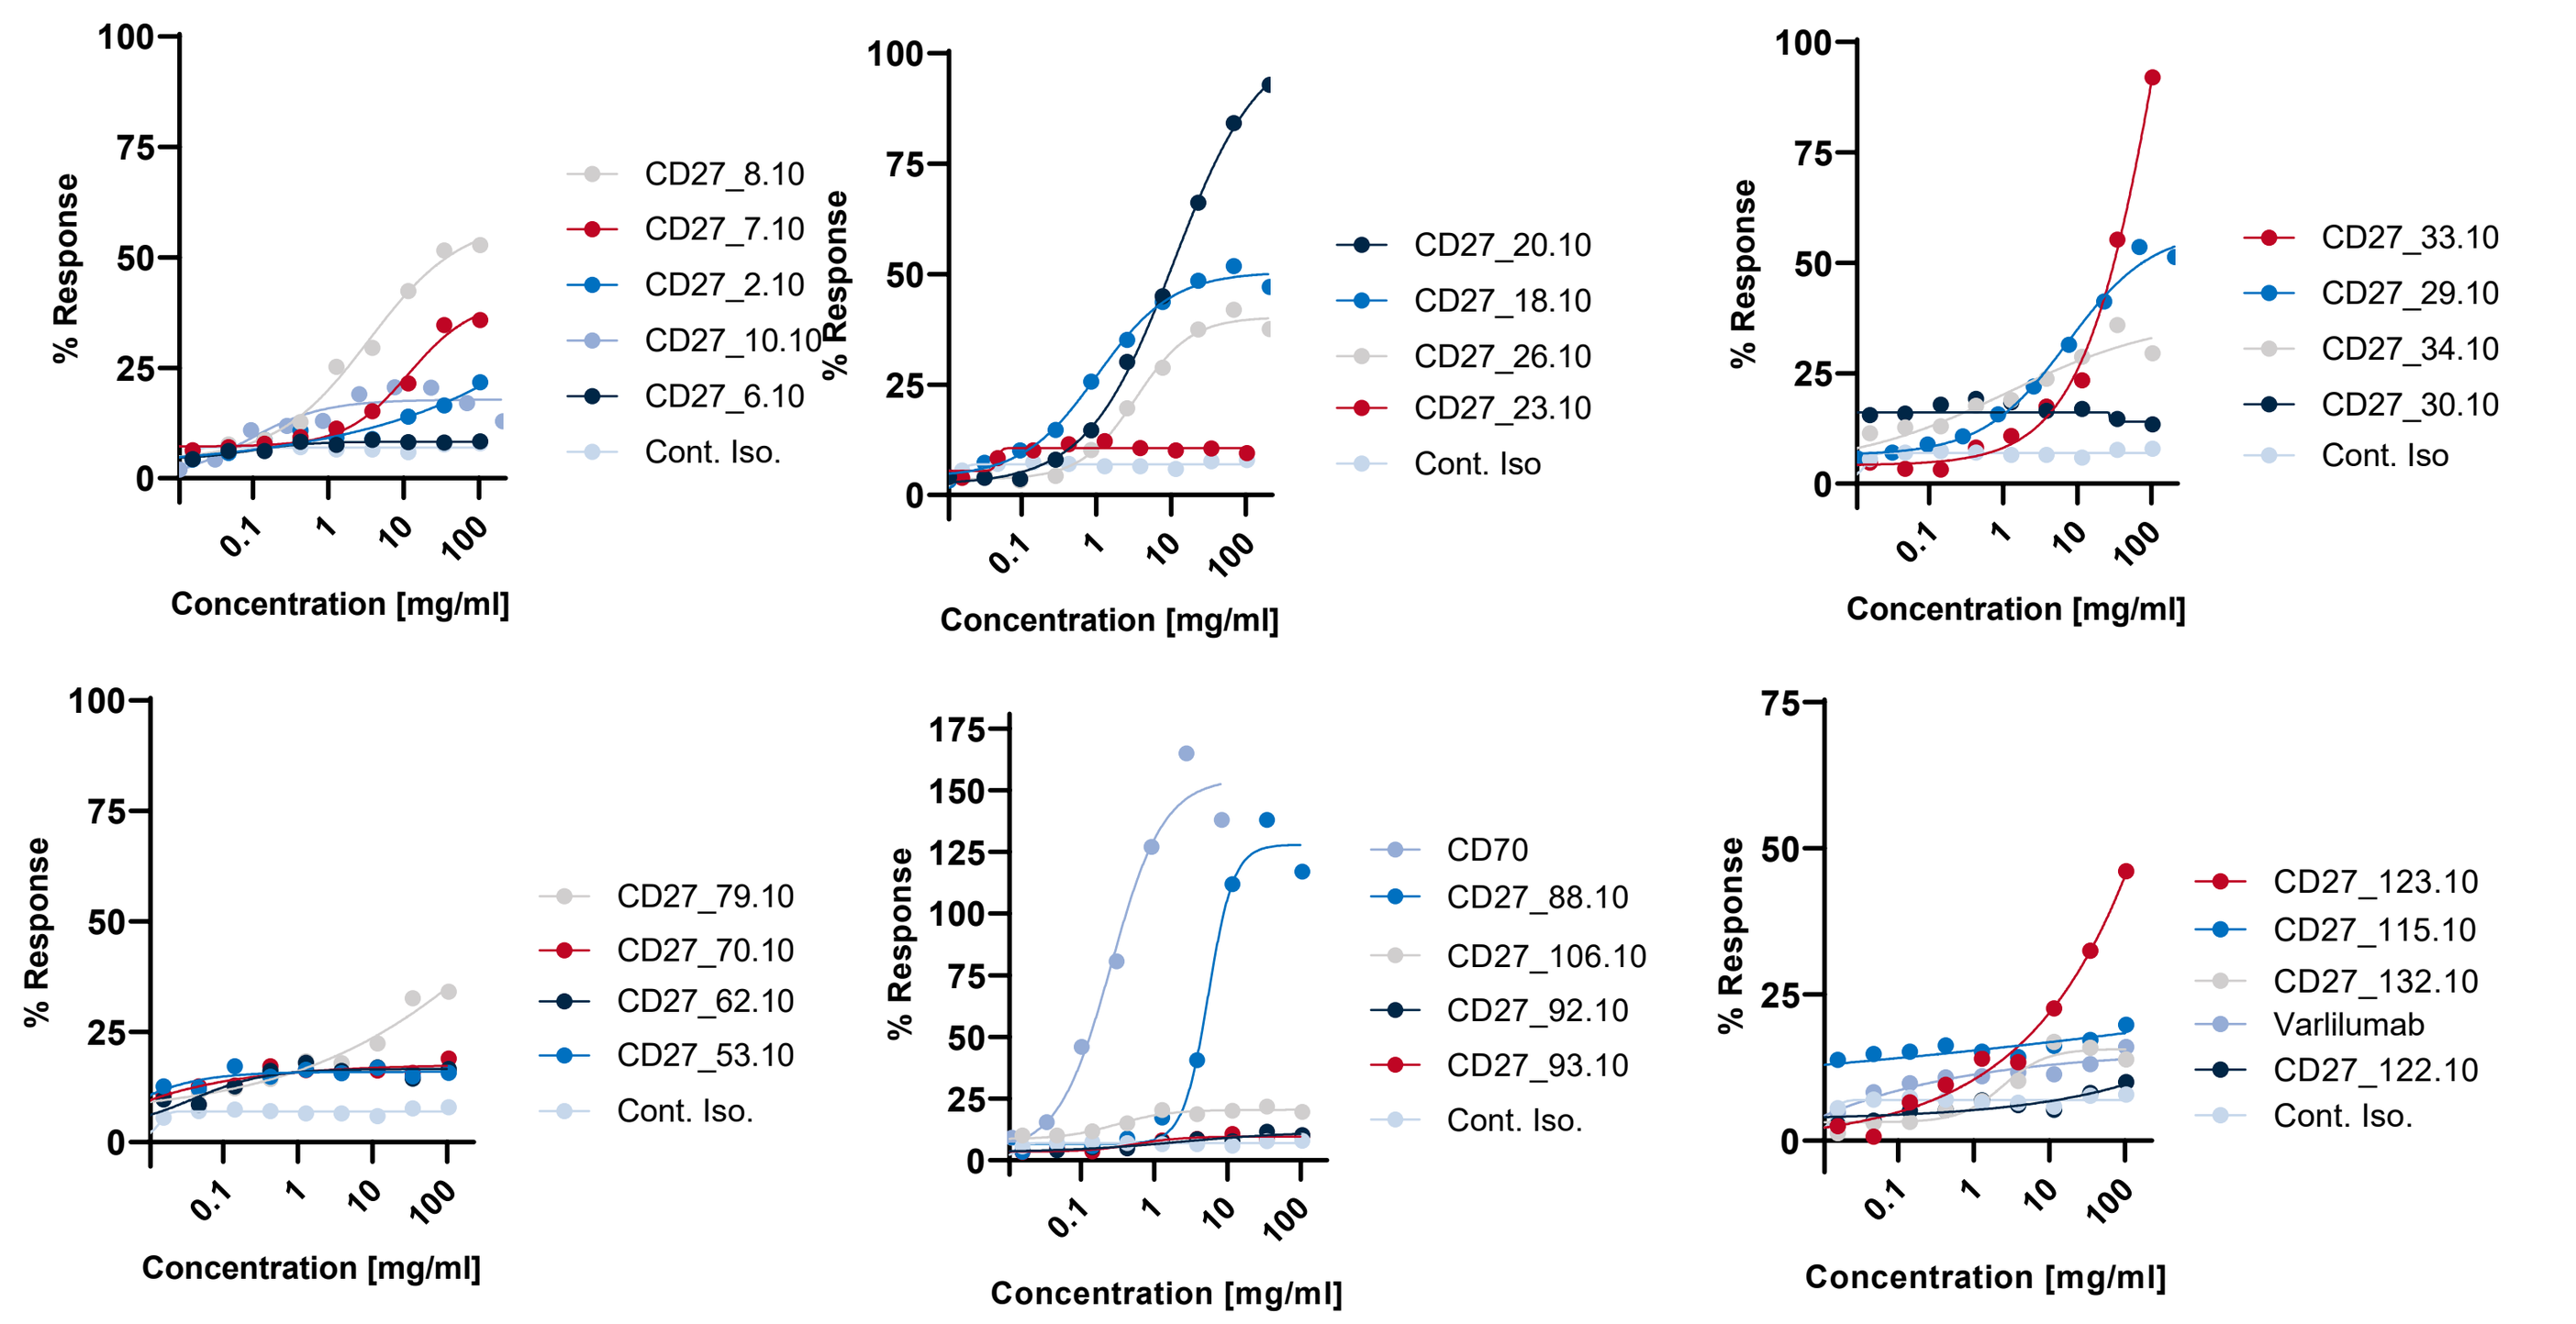
% Response
Concentration [mg/ml]
CD27_8.10
CD27_7.10
CD27_2.10
CD27_10.10
CD27_6.10
CD27_20.10
CD27_18.10
CD27_26.10
CD27_23.10
CD27_33.10
CD27_29.10
CD27_34.10
CD27_30.10
CD27_79.10
CD27_70.10
CD27_62.10
CD27_53.10
CD70
CD27_88.10
CD27_106.10
CD27_92.10
CD27_93.10
CD27_123.10
CD27_115.10
CD27_132.10
Varlilumab
CD27_122.10
Cont. Iso.

Figure 5. Anti-CD27 agonist assay was carried out with NFκB-Luc2/CD27 Jurkat cells seeded at 2e5 cells/well in X-VIVO-15 medium in the 96-well plates pre-coated with 5 µg/mL anti-CD3 mAb. Cells were then incubated with anti-CD27 mAbs or CD27 ligand (CD70-ECD trimer, R&D Systems) for 4 hrs at 37°C, 5% CO_2 humidified incubator. NFκB reporter induction was measured by luminescence in the presence of Bio-Glo reagent.

Three selected Anti-CD27 mAb lead candidates

Figure 6. (A) Anti-CD27 agonist assay was carried out with NFκB-Luc2/CD27 Jurkat cells incubated with anti-CD27 mAbs or CD27 ligand (CD70-ECD trimer, R&D Systems) for 4 hrs. NFκB reporter induction was measured by luminescence in the presence of Bio-Glo reagent. **(B)** ELISA binding studies were conducted with immobilized human CD27 (rabbit-Fc). Binding of anti-CD27 mAbs was detected using biotinylated anti-human IgG (light chain) followed by streptavidin HRP.

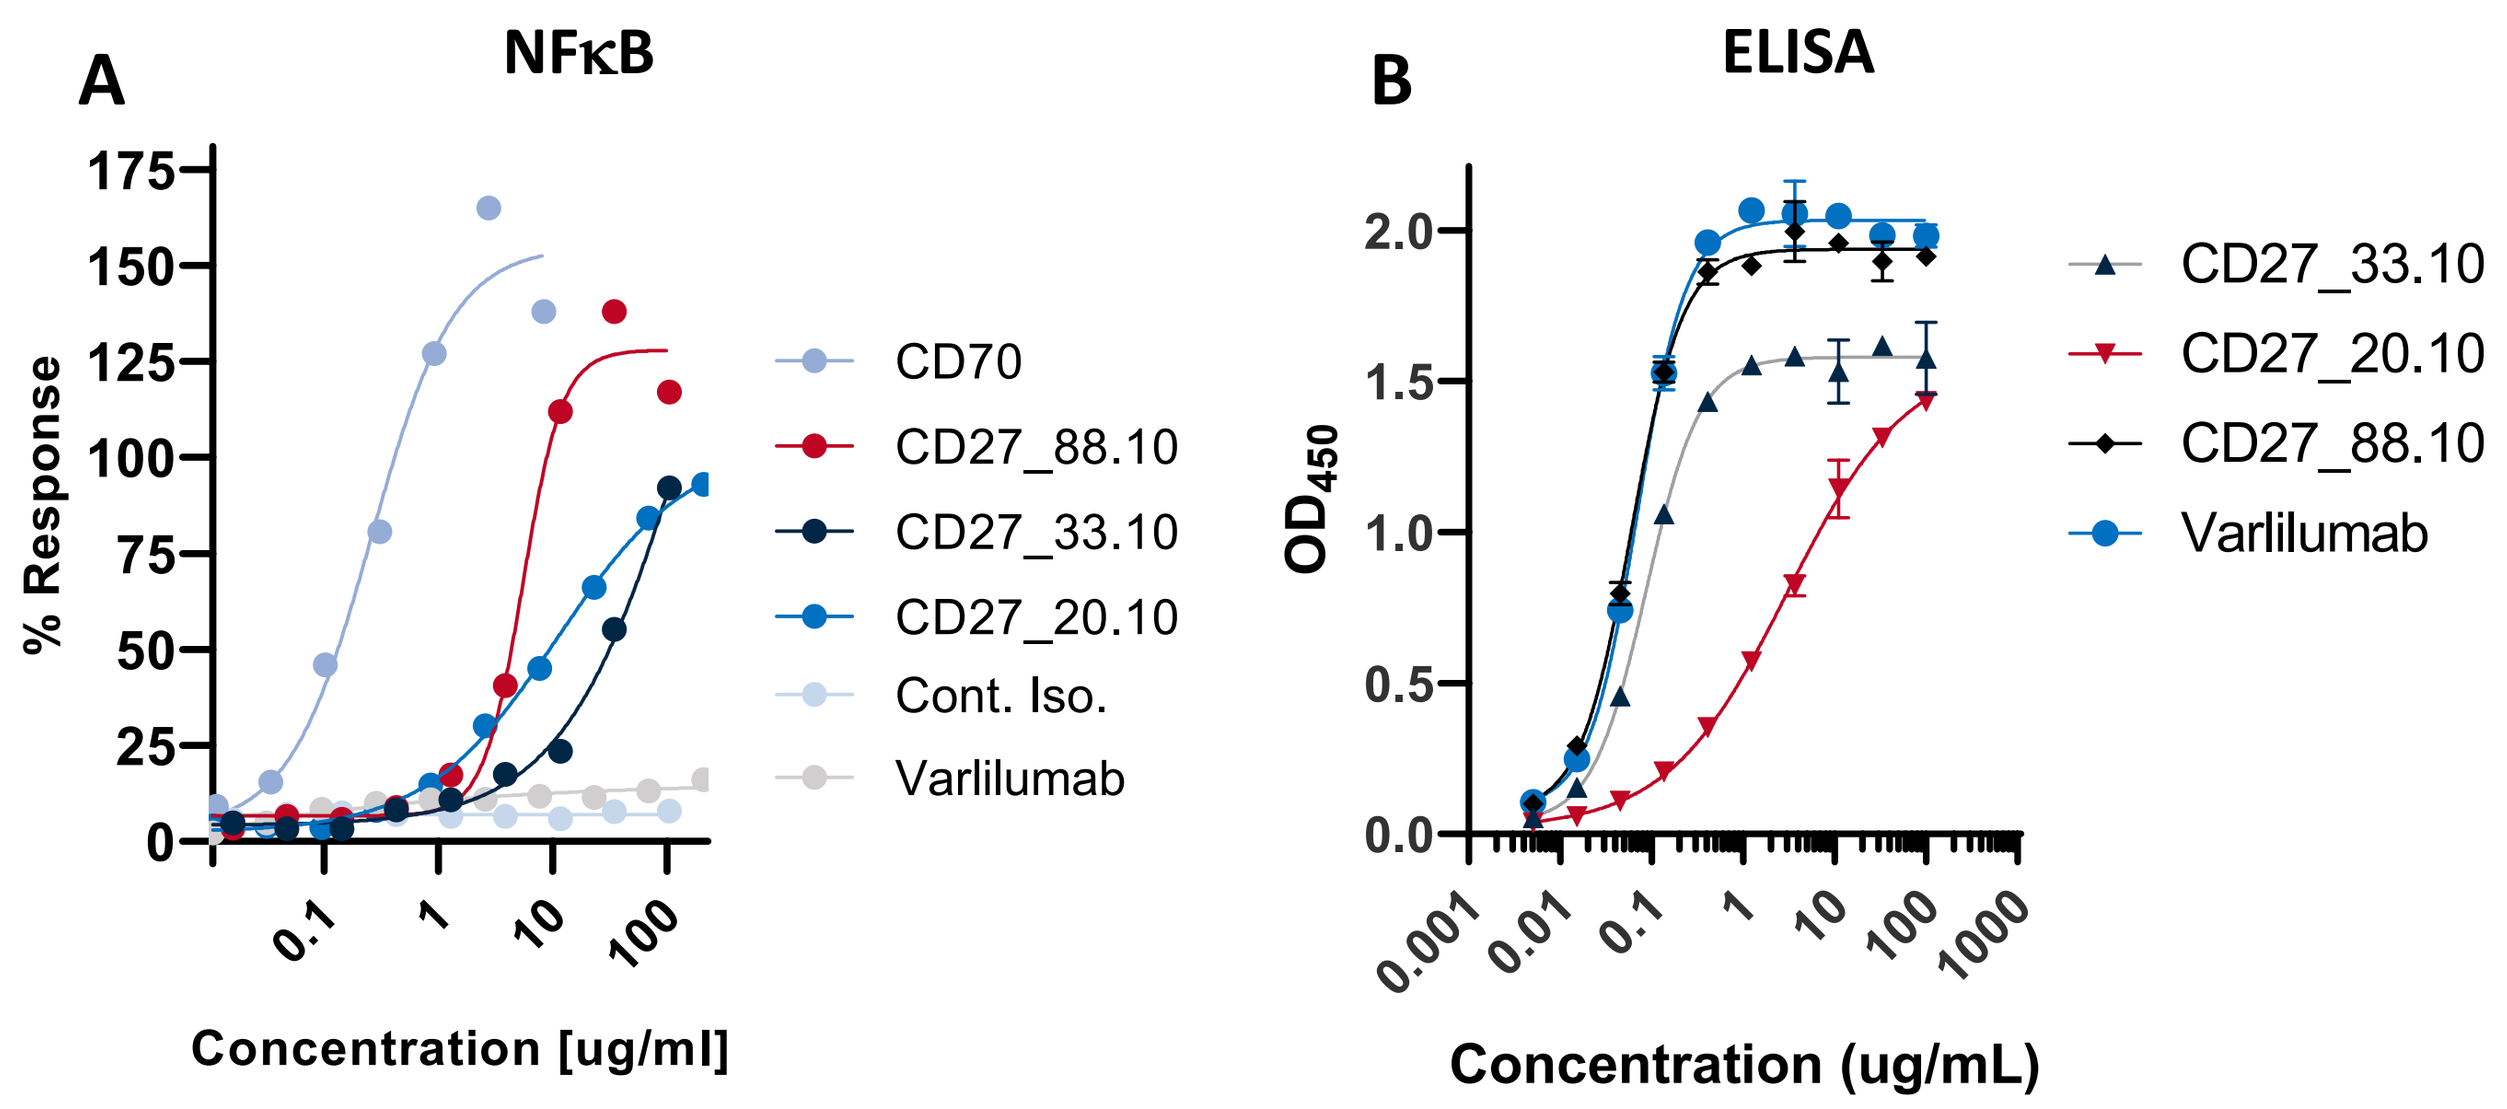
A
NFκB
% Response
Concentration [ug/ml]
CD70
CD27_88.10
CD27_33.10
CD27_20.10
Cont. Iso.
Varlilumab
B
ELISA
OD450
Concentration (ug/mL)
CD27_33.10
CD27_20.10
CD27_88.10
Varlilumab

Anti-CD27 mAb lead candidates induce NK cell activation (CD69+)

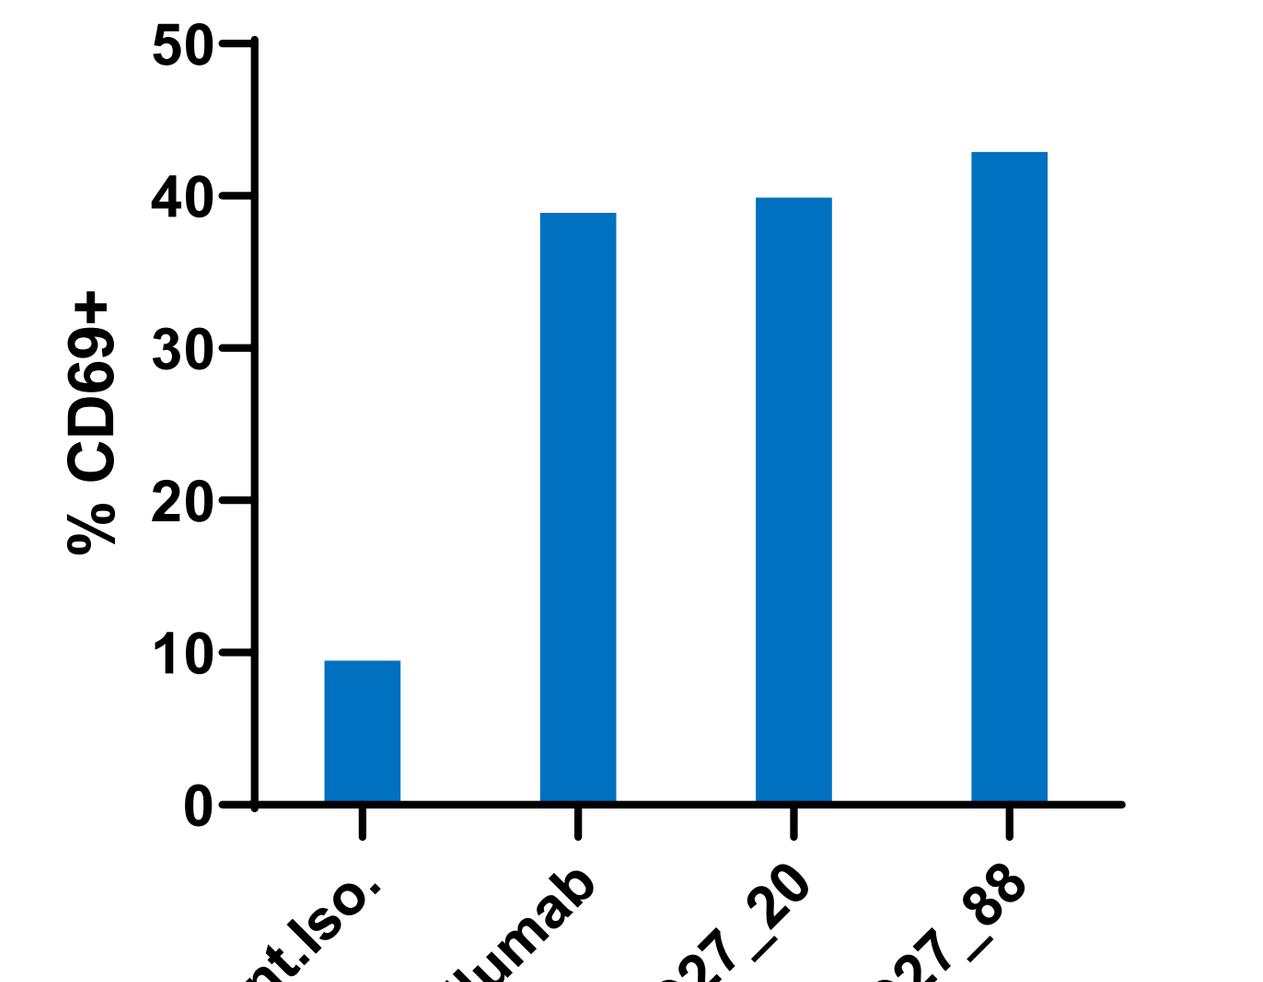
% CD69+
Cont.Iso.
Varlilumab
CD27_20
CD27_88

Figure 7. PBMCs were cultured for four days with 10 ug/mL of HuIgG1, or anti-CD27 mAbs Varlilumab, CD27_20.10, or CD27_88.10. Cells were harvested and immunophenotyped. CD69 activation marker was assessed on CD56-, CD16+ Effector NK cells.

Kineta's anti-CD27 mAb lead candidates show robust T cell activation

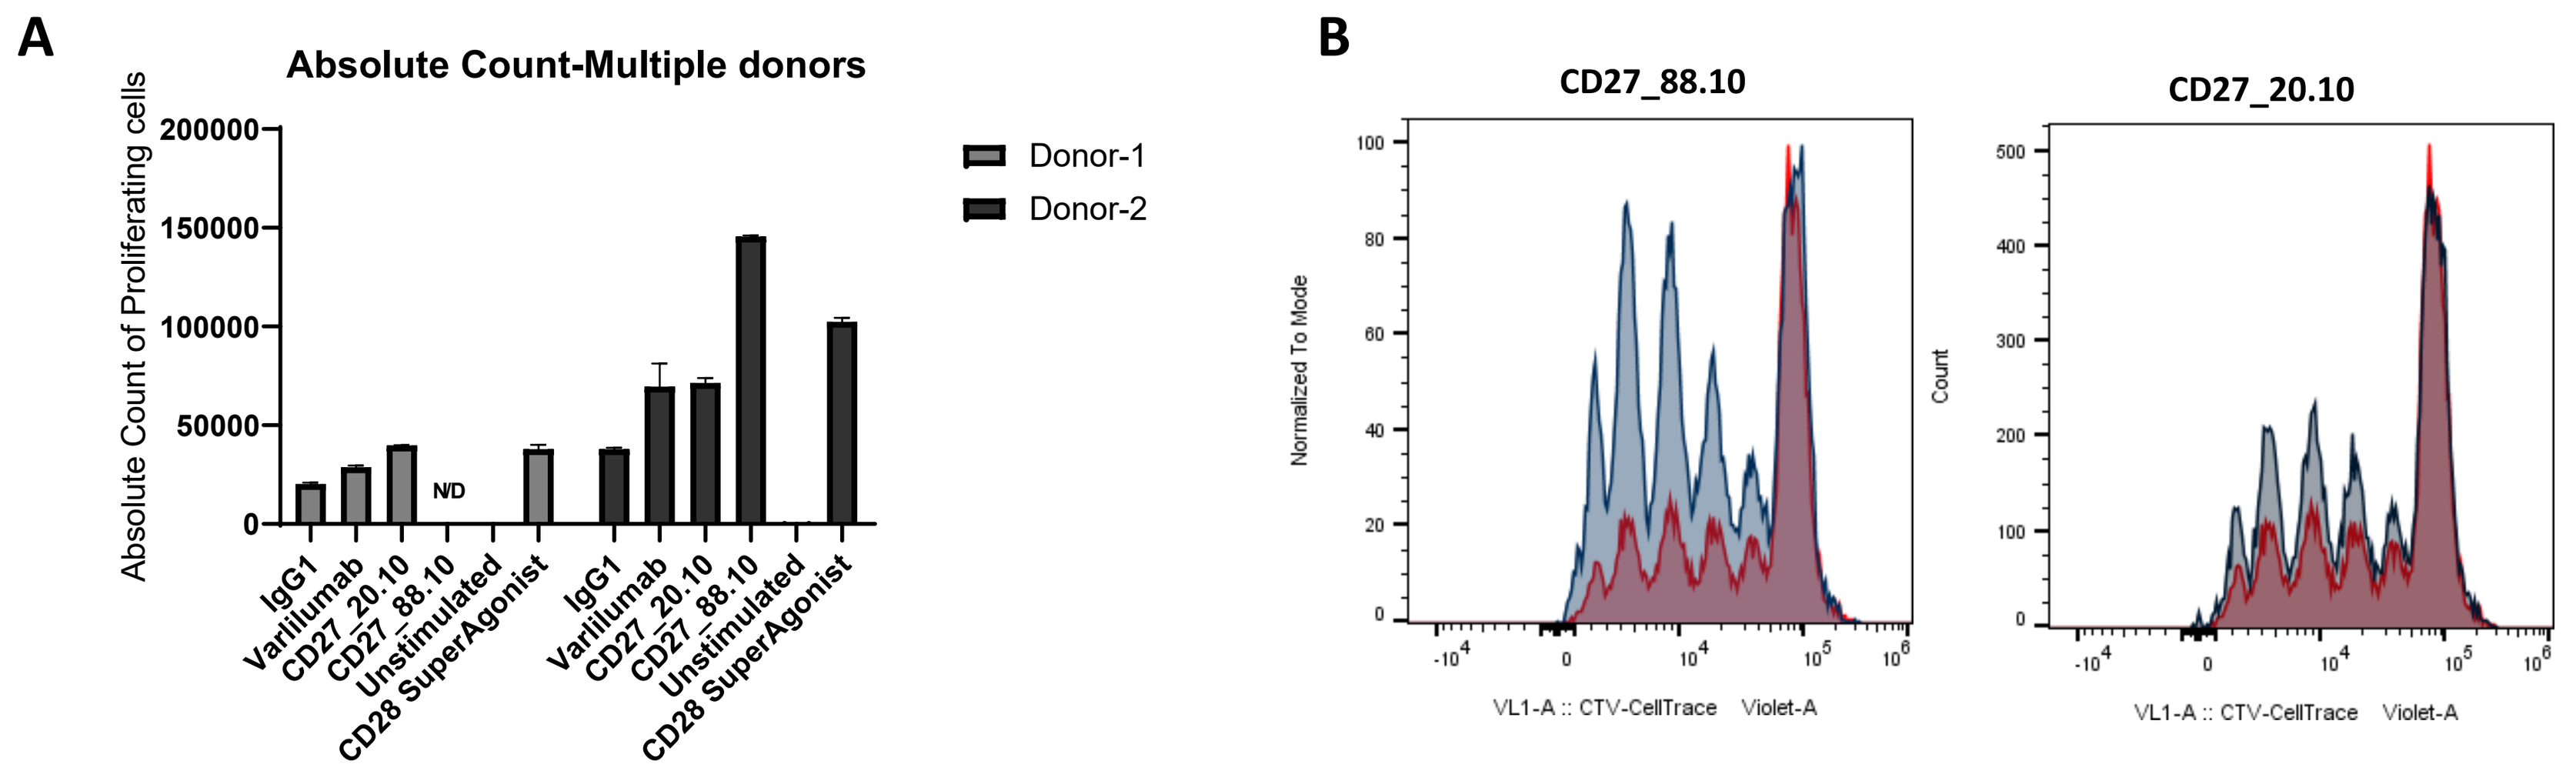
A
Absolute Count-Multiple donors
Absolute Count of Proliferating cells
Donor-1
Donor-2
ND
IgG1
Varlilumab
CD27_20.10
CD27_88.10
Unstimulated
CD28 SuperAgonist
B
CD27_88.10
CD27_20.10
Normalized To Mode
Count
VL1-A :: CTV-CellTrace Violet-A

Figure 8. T-cells from healthy donors were stained with cell trace violet (CTV) and stimulated with 10 ug/mL of anti-CD27 mAbs and T-cell activator (1 ug/mL anti-CD3 and 5 ug/mL anti-CD28). The frequency of CTV labelled cells was examined after 4 days using flow cytometry. **(A)** Absolute count of proliferating cells for each antibody is represented. **(B)** Histogram plots comparing T-cell proliferation for selected anti-CD27 at 10ug/mL (in blue) to Isotype control (in red).

Anti-mCD27 agonistic Ab shows anti-tumor activity alone or in combination with anti-mPD1 or Kineta's anti-VISTA mAb (KVA12123)

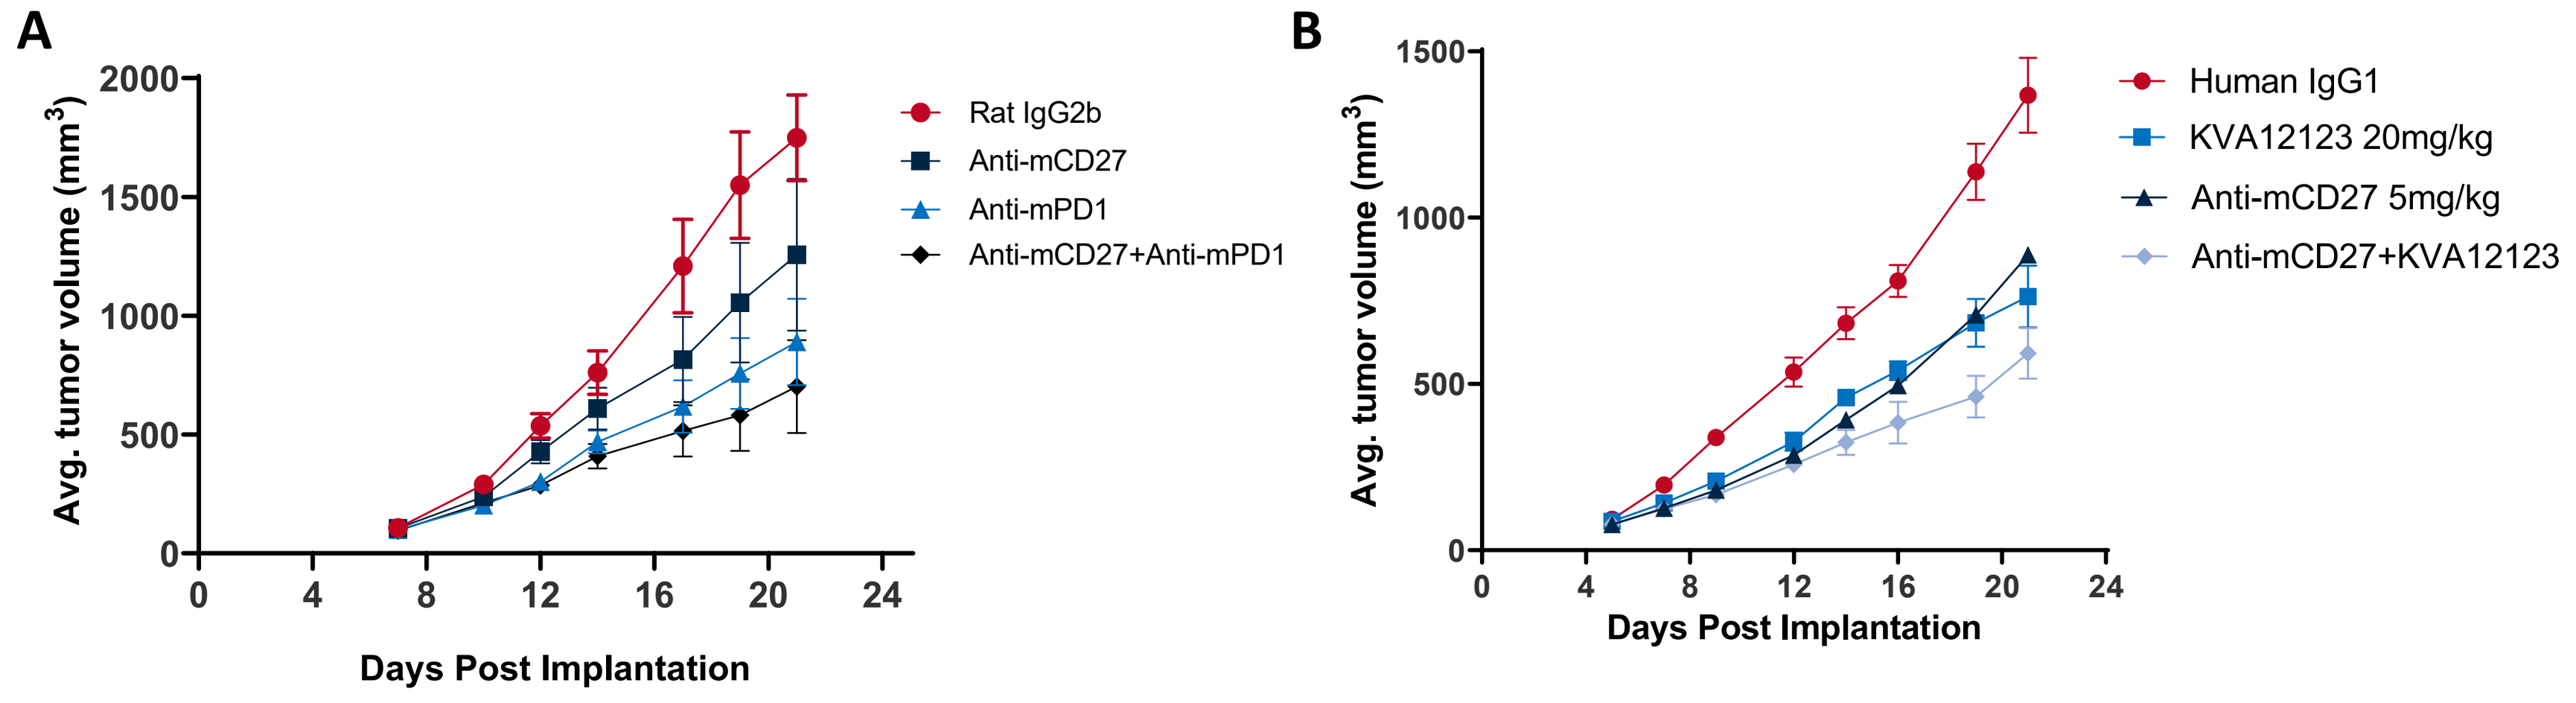
A
Avg. tumor volume (mm3)
Days Post Implantation
Rat IgG2b
Anti-mCD27
Anti-mPD1
Anti-mCD27+Anti-mPD1
B
Human IgG1
KVA12123 20mg/kg
Anti-mCD27 5mg/kg
Anti-mCD27+KVA12123

Figure 9. (A) Plot of mean tumor volumes vs. time following subcutaneous implantation of 10e6 EG7-OVA cells/mouse into female C57BL/6J. Seven days after cell implantation, mice received IP injection of either 10mg/kg of rat IgG2b, 5mg/kg of anti-mPD1, 10mg/kg of anti-mCD27 agonistic antibody (BioXCell BE0348) alone or in combination twice a week for 2.5 weeks. **(B)** Plot of mean tumor volumes vs. time following subcutaneous implantation of 0.5x10e6 MB49 cells/mouse into female hVISTA-KI C57BL/6. Five days after cell implantation, mice received IP injection of either 20mg/kg of hIgG1, 5mg/kg of anti-mCD27 agonistic antibody (BioXCell BE0348), 20mg/kg of KVA12123 alone or in combination twice a week for 3 weeks. Error bars represent SEM.

Conclusions

- We selected 3 lead therapeutic antibodies from our library of 147 fully human anti-CD27 monoclonal antibodies generated in the Trianni mice.
- We confirmed our lead antibodies' binding potency and cross-reactivity with non-human primate CD27 but not with the murine CD27.
- Our lead candidates demonstrate strong agonist proprieties. They induce NK cell activation as well as T cell proliferation and activation after engagement of NFκB.
- Our lead antibodies will be evaluated as a single agent or in combination with other immuno-therapies in vivo in solid and hematological mouse tumor models in hCD27-KI mice.