Highly Potent Fully Human Anti-VISTA Antibodies Efficiently Abrogate the Interaction of VISTA to Its Putative Receptors at Different pH.

Shawn Iadonato, Eric Tarcha, Robert Bader, Benjamin Dutzar, Nathan Eyde, Emily Frazier, David Jurchen, Remington Lance, Cristina Loomis, Kurt Lustig, Yulia Ovechkina, David Peckham, Jeff Posakony, Shaarwari Sridhar, Mei Xu and Thierry Guillaudeux.

Kineta Inc. 219 Terry Av. North, Seattle, WA 98109.

Background

- VISTA (V-domain Ig Suppressor of T cell Activation) is a unique CD28/B7 family member with poorly defined receptors. However, VISTA itself, PSGL-1, VSIG3, VSIG8 and LRIG1 have been suggested as putative receptors.
- VISTA is highly expressed on circulating and intratumoral myeloid cells as well as NK cells and Treg.
- VISTA is a negative regulator that directly suppresses T cell activation and proliferation.
- High VISTA expression correlates with poor survival in cancer patients.
- VISTA is a unique immune checkpoint inhibitor for tumor immunotherapy.

Objectives

- Various research groups have demonstrated that VISTA can interact with at least 5 receptors: VISTA itself, PSGL-1 in acidic conditions, VSIG3, VSIG8 and LRIG1.
 - Identify and/or confirm the putative receptors for VISTA.
 - Describe the expression of these potential receptors on the different blood cell populations.

Results

VISTA and VISTA Receptors Expression on Human Whole Blood Leukocyte Populations

Results representative of 3 healthy donors: Donor # 4903 shown

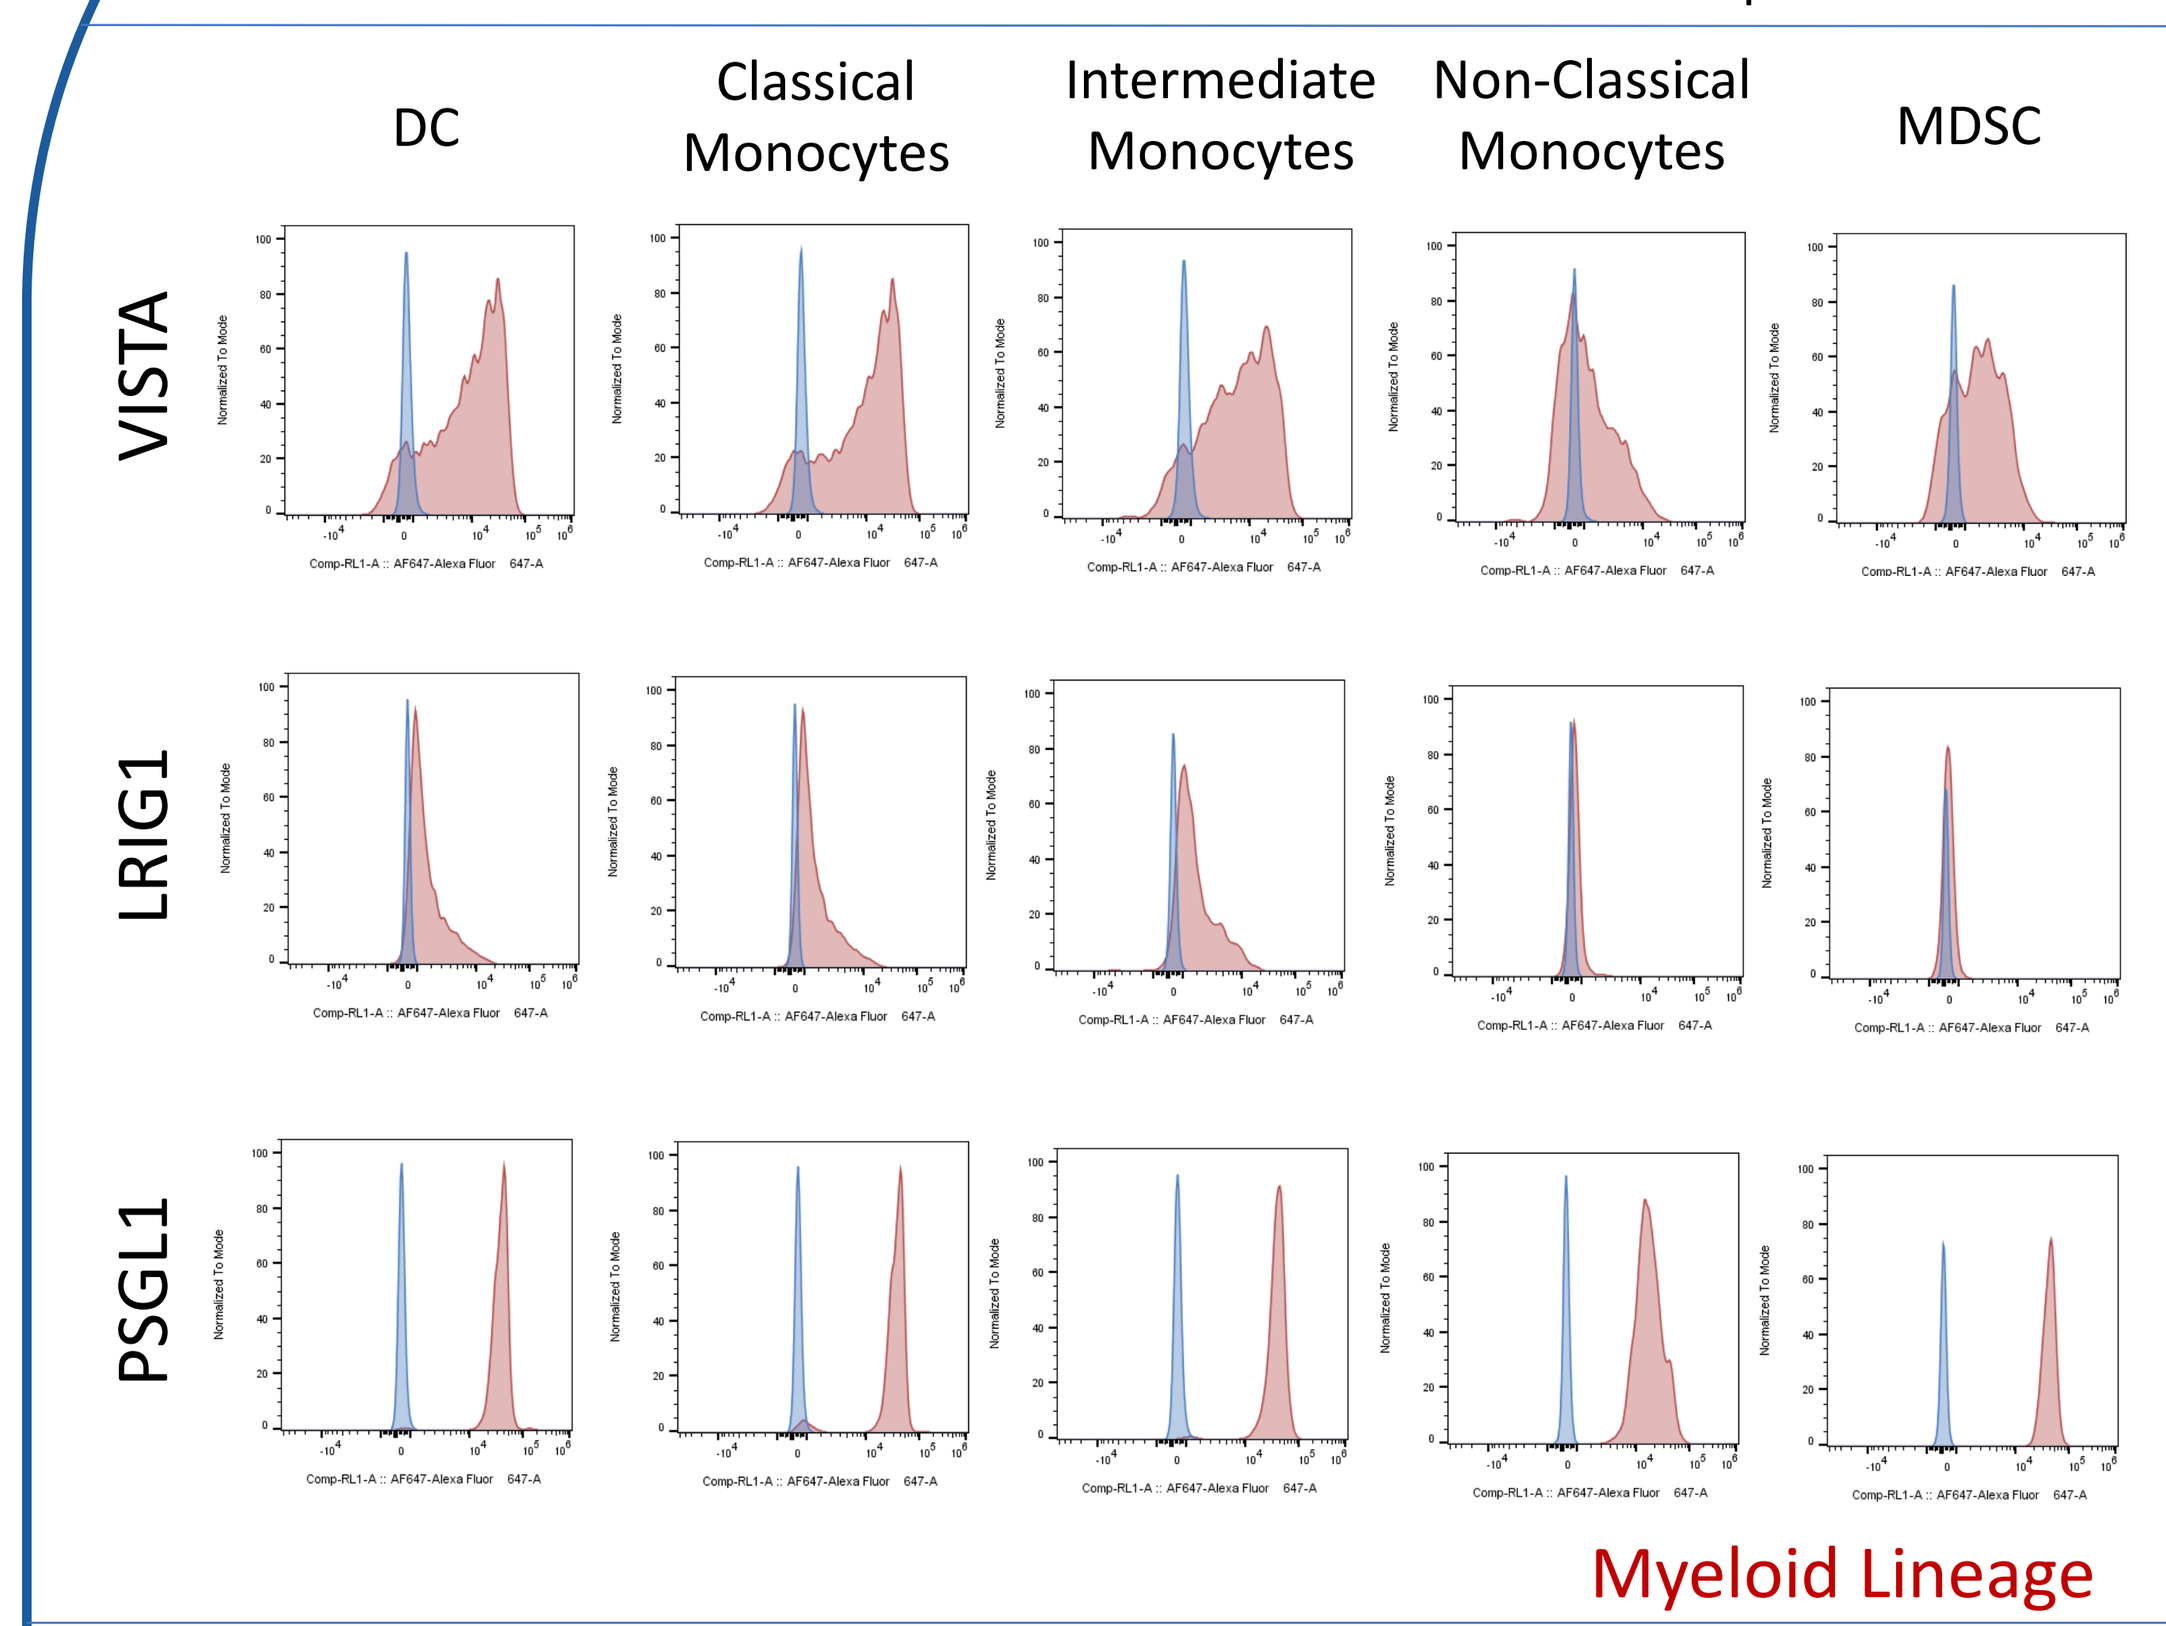
DC
Classical Monocytes
Intermediate Monocytes
Non-Classical Monocytes
MDSC
VISTA
LRIG1
PSGL1
Myeloid Lineage

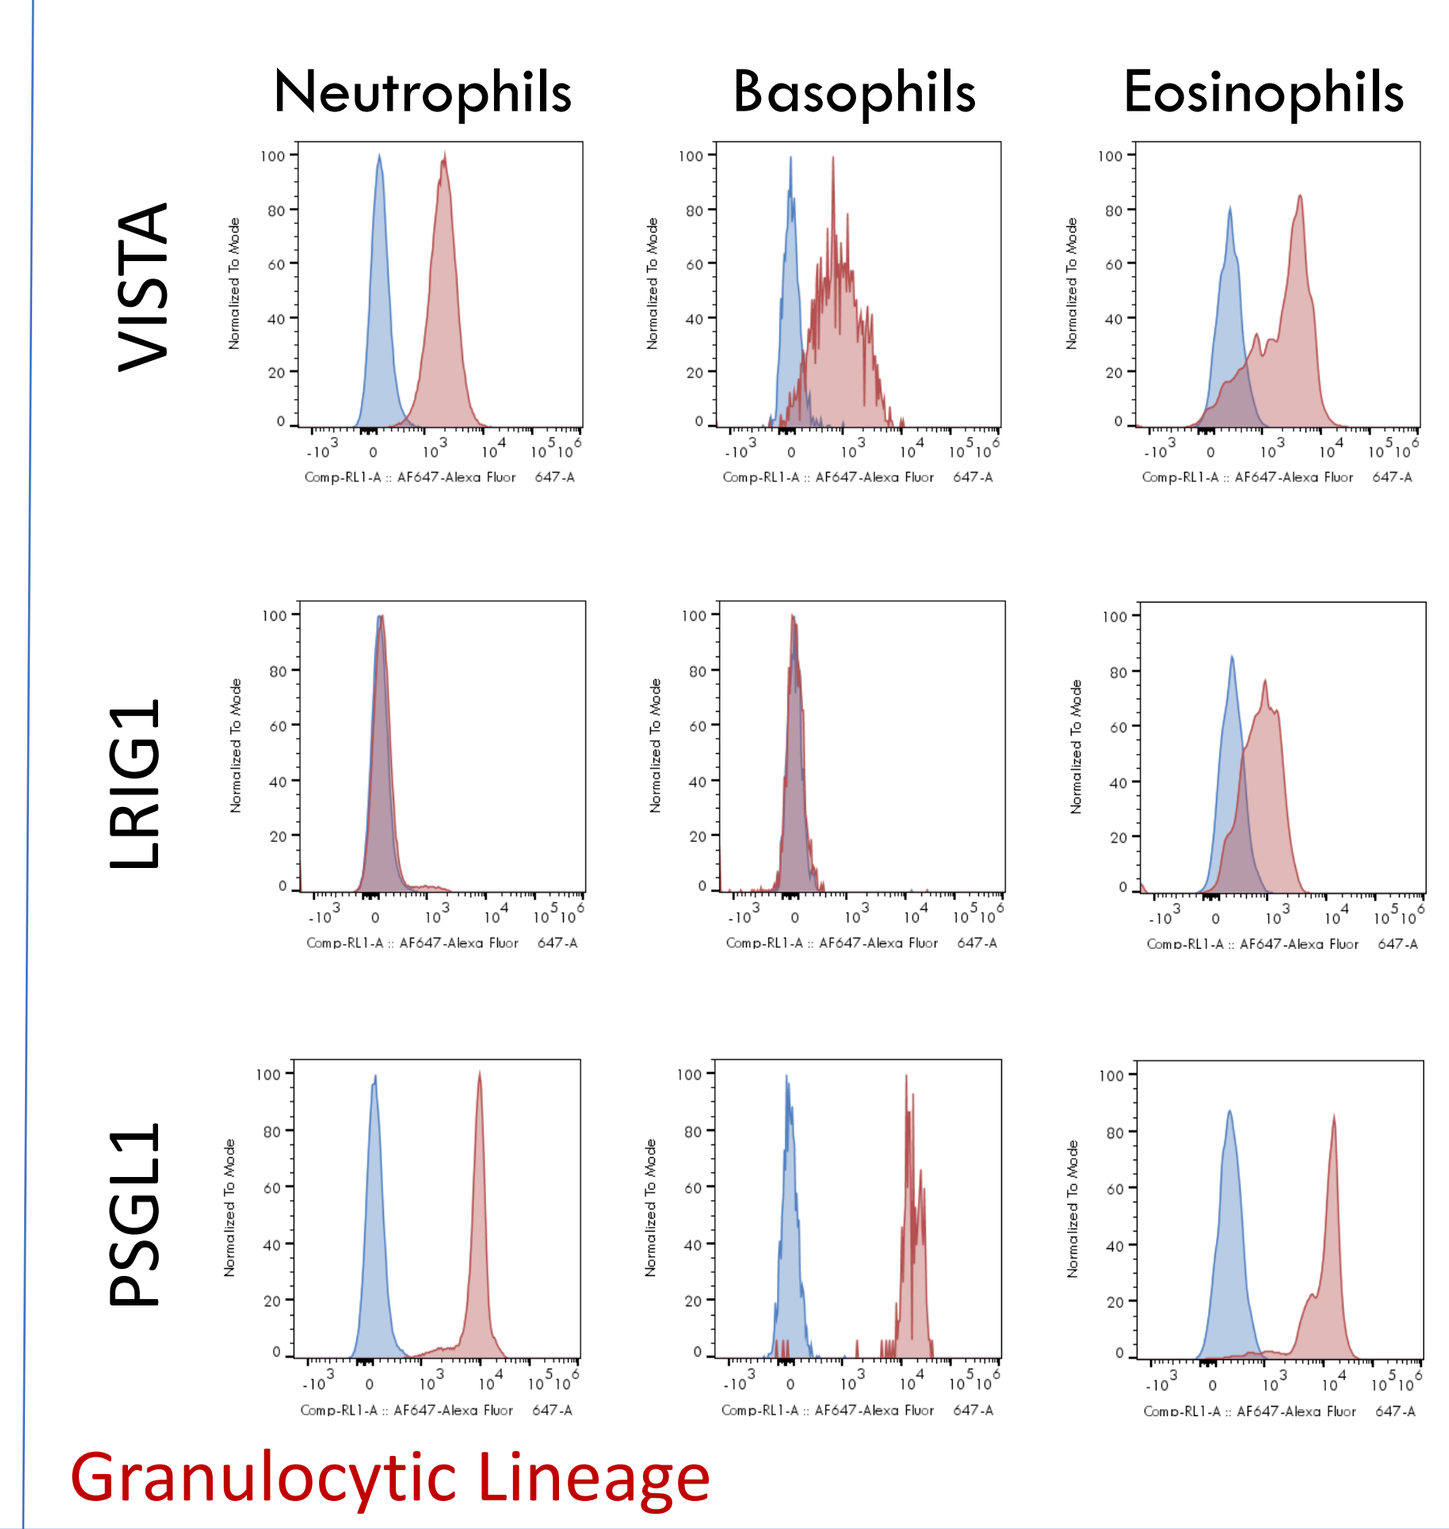
Neutrophils
Basophils
Eosinophils
VISTA
LRIG1
PSGL1
Granulocytic Lineage

Whole peripheral blood from three donors was stained with a four-antibody cocktail against granulocytic, myeloid, NK, and T cell populations and a series of antibodies against VISTA, VISTA ligands, or isotype controls. Samples were fixed and RBC lysed, acquired on an Attune flow cytometer, and analyzed with FlowJo software. Histogram overlays of VISTA or Ligand staining vs. isotype control are shown. Fold Over Background = Anti-VISTA MFI or Anti-Ligand MFI ÷ Isotype Control MFI.

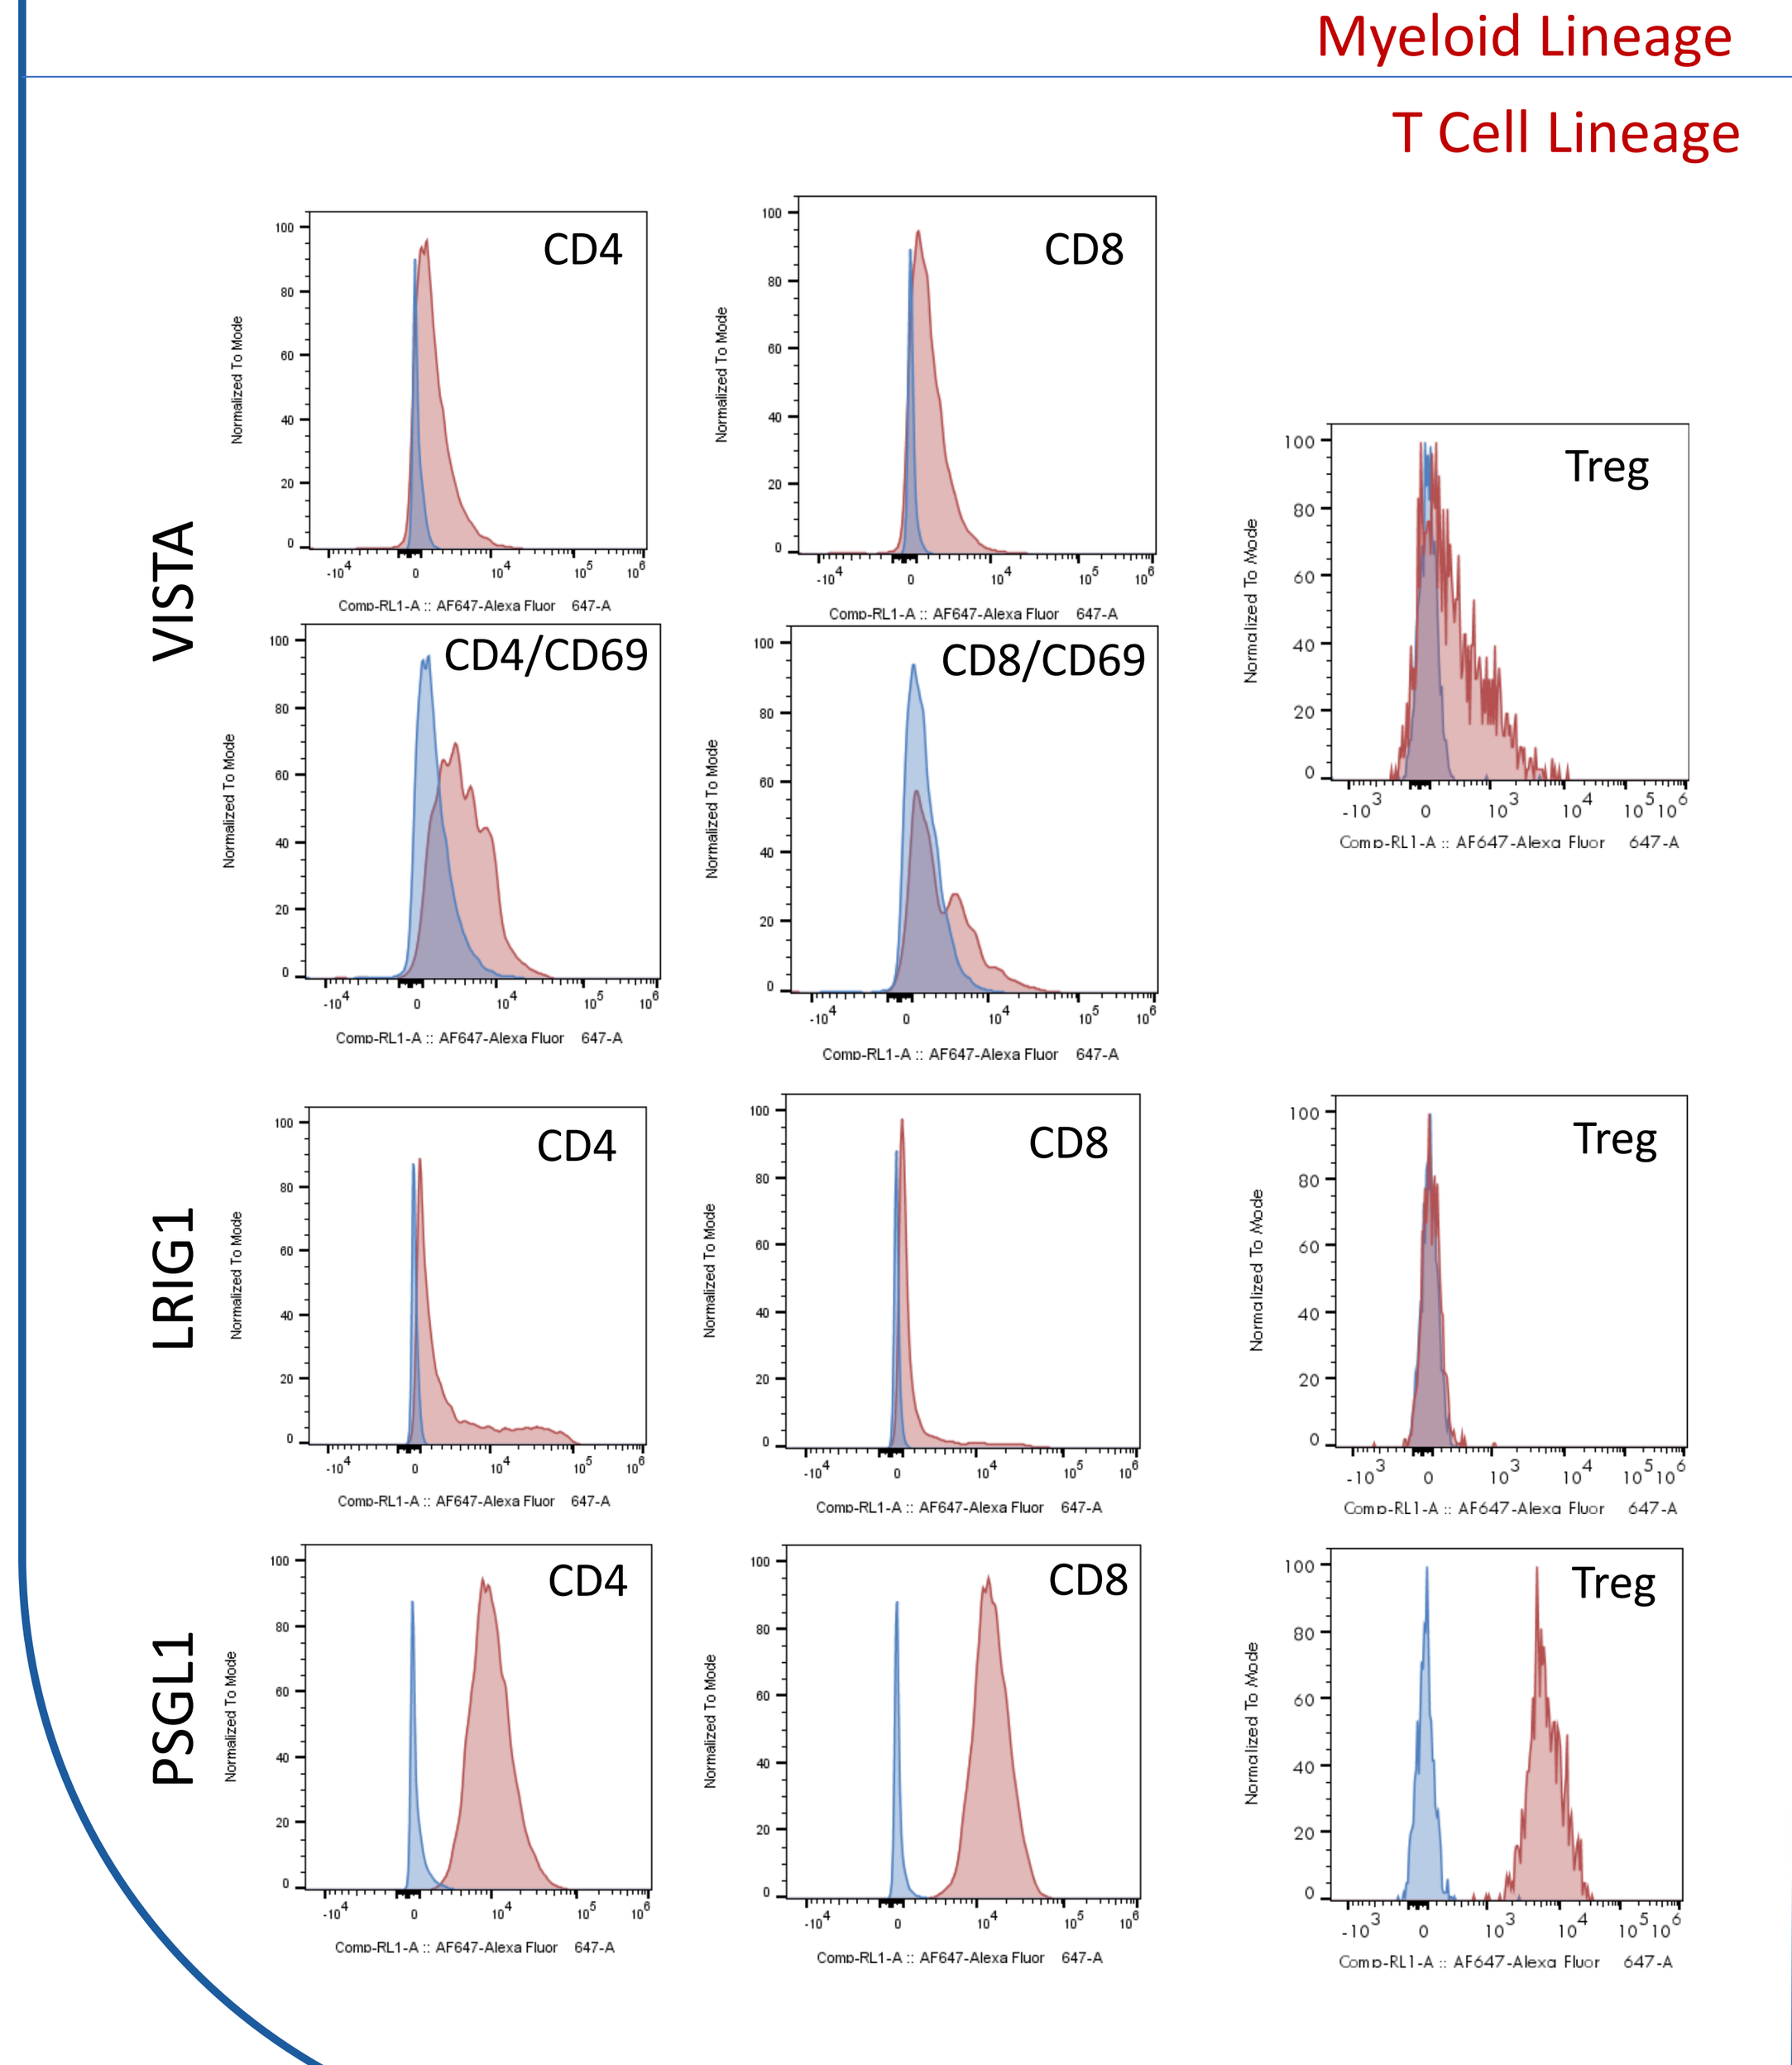
T Cell Lineage
VISTA
LRIG1
PSGL1
CD4
CD8
CD4/CD69
CD8/CD69
Treg

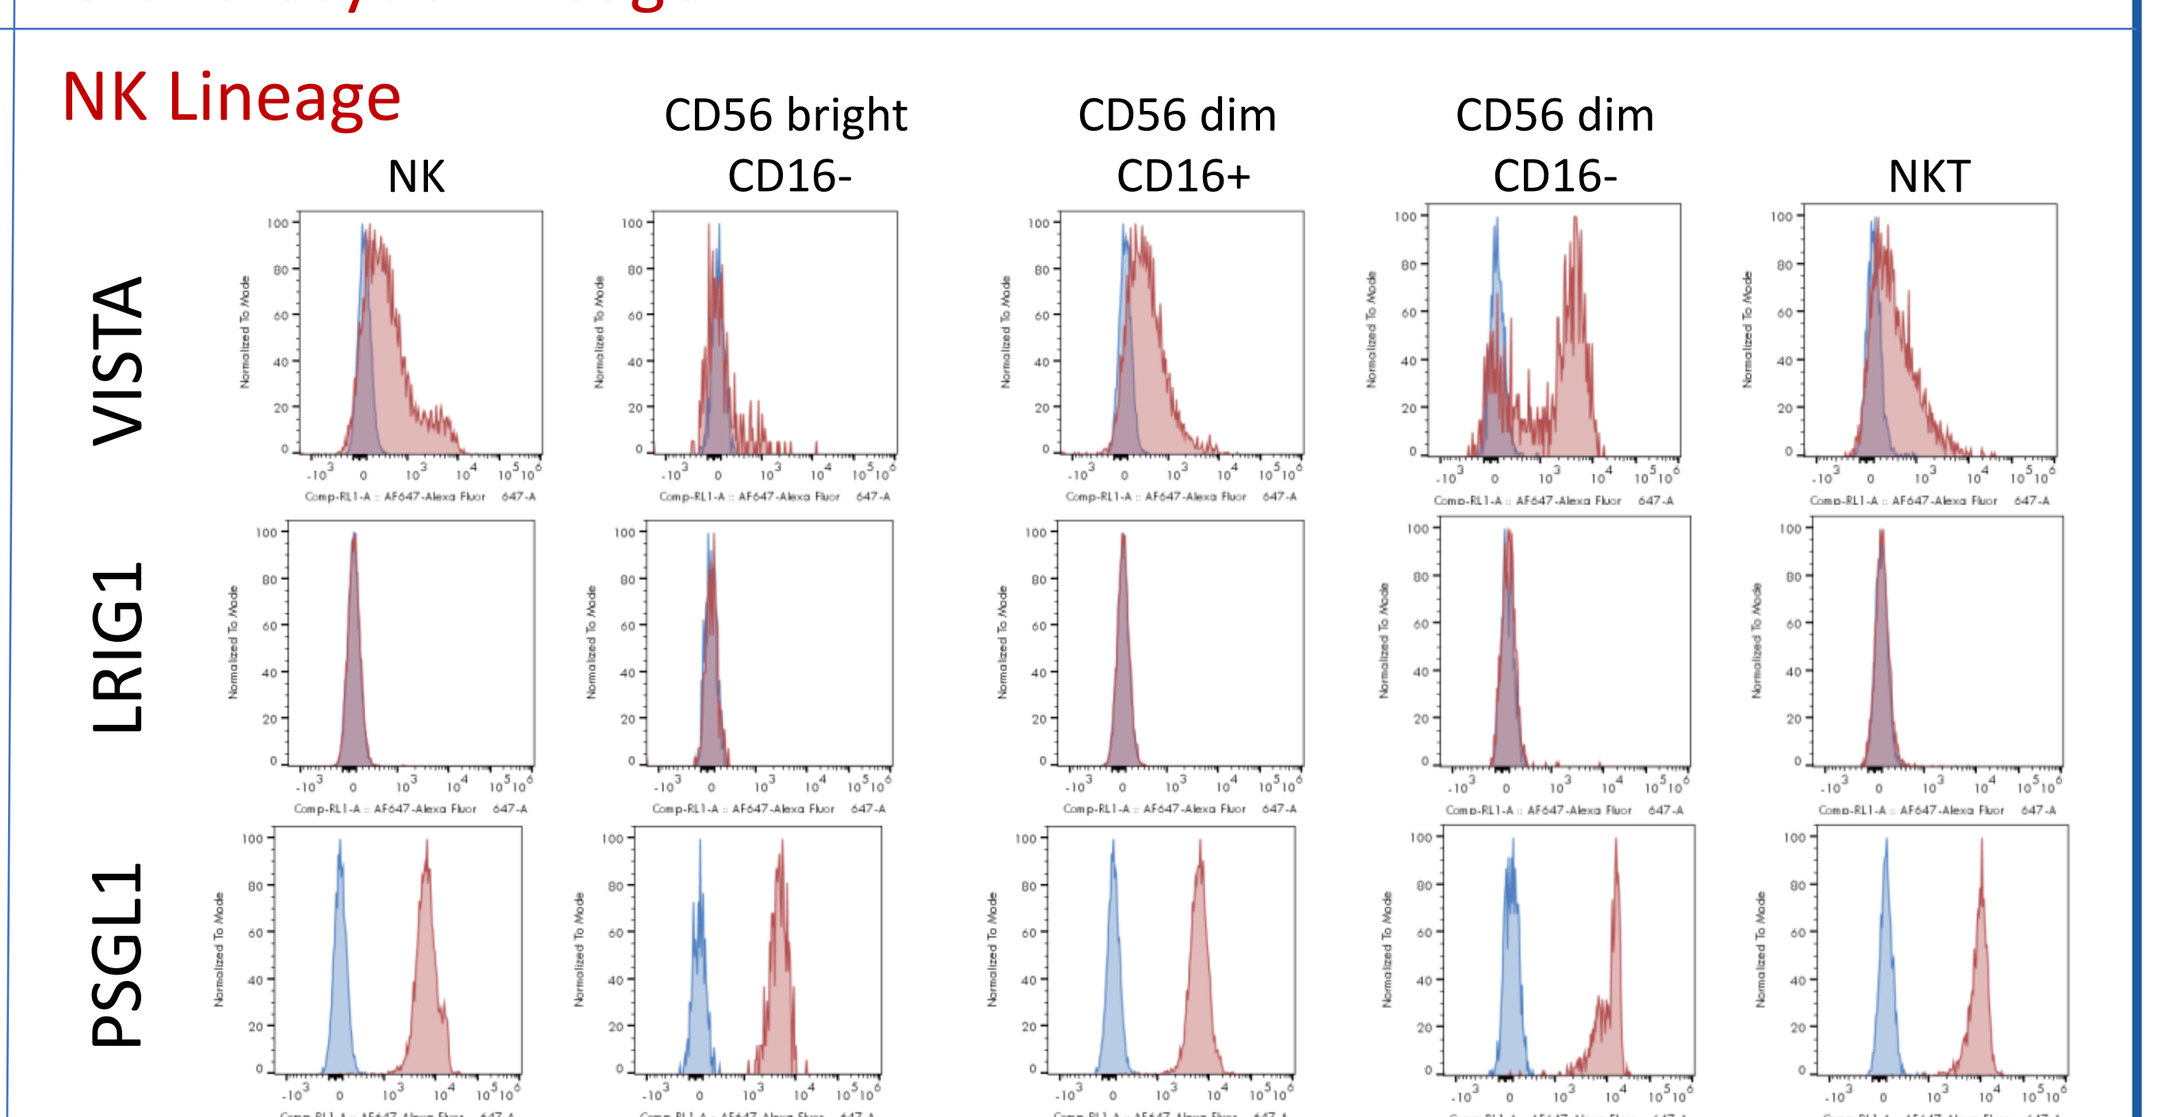
NK Lineage
NK
CD56 bright CD16-
CD56 dim CD16+
CD56 dim CD16-
NKT
VISTA
LRIG1
PSGL1

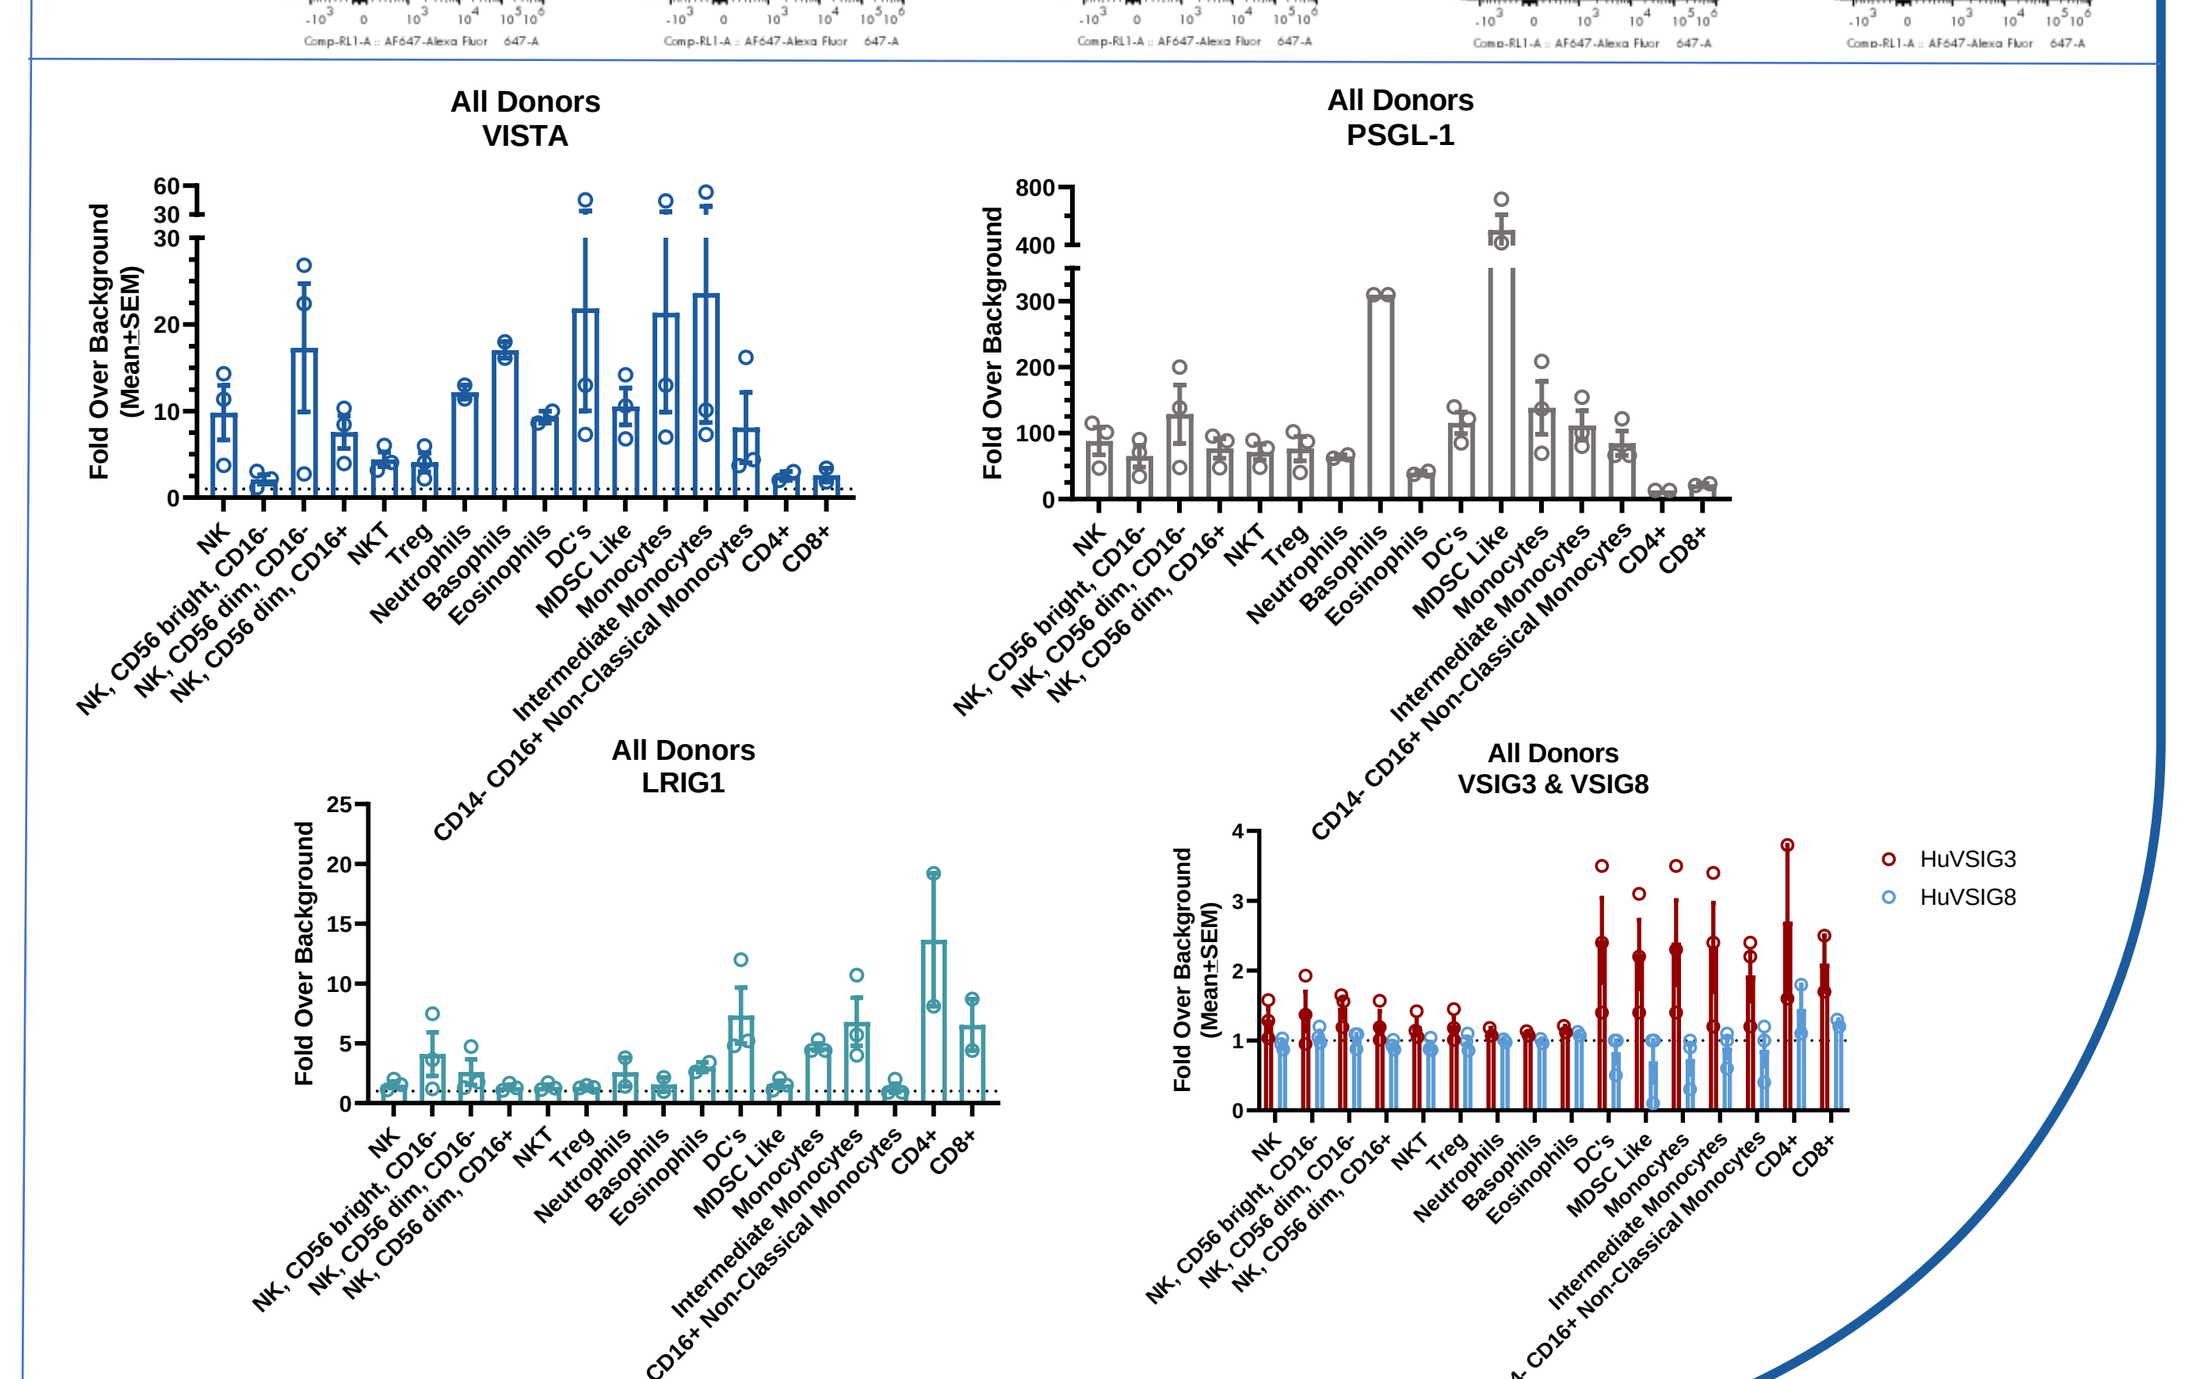
All Donors VISTA
All Donors PSGL-1
All Donors LRIG1
All Donors VSIG3 & VSIG8
HuVSIG3
HuVSIG8

VISTA interacts with its putative receptors at different pH

- VISTA binds strongly to VISTA at pH 7.4 and pH 6.0

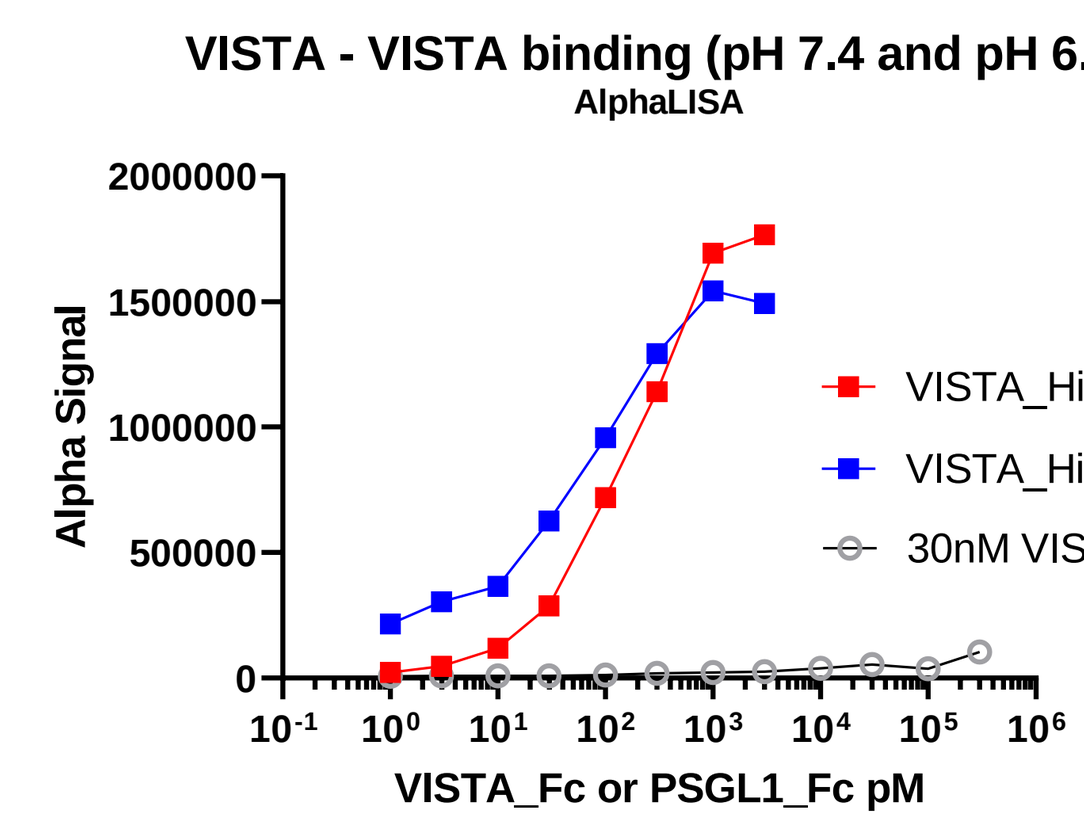
VISTA - VISTA binding (pH 7.4 and pH 6.0)
AlphaLISA
Alpha Signal
VISTA_Fc or PSGL1_Fc pM
VISTA_His_AVI (30nM) pH7.4
VISTA_His_AVI (30nM) pH6
30nM VISTA (PSGL1) pH6

huVISTA_His_Avi and huVISTA_huFc or huPSGL1_muFc used in AlphaLisa binding assay. Anti-human IgG1 or anti-murine IgG1 Acceptor beads (10ug/ml final) and Streptavidin Donor beads (40ug/ml final) were used, and reagents were combined in either Universal AlphaLisa buffer (pH7.4) or MES buffer (pH6). Fluorescent signal was read with CLARIOstar at 615nm.

- VISTA binds to PSGL-1 only at pH 6.0

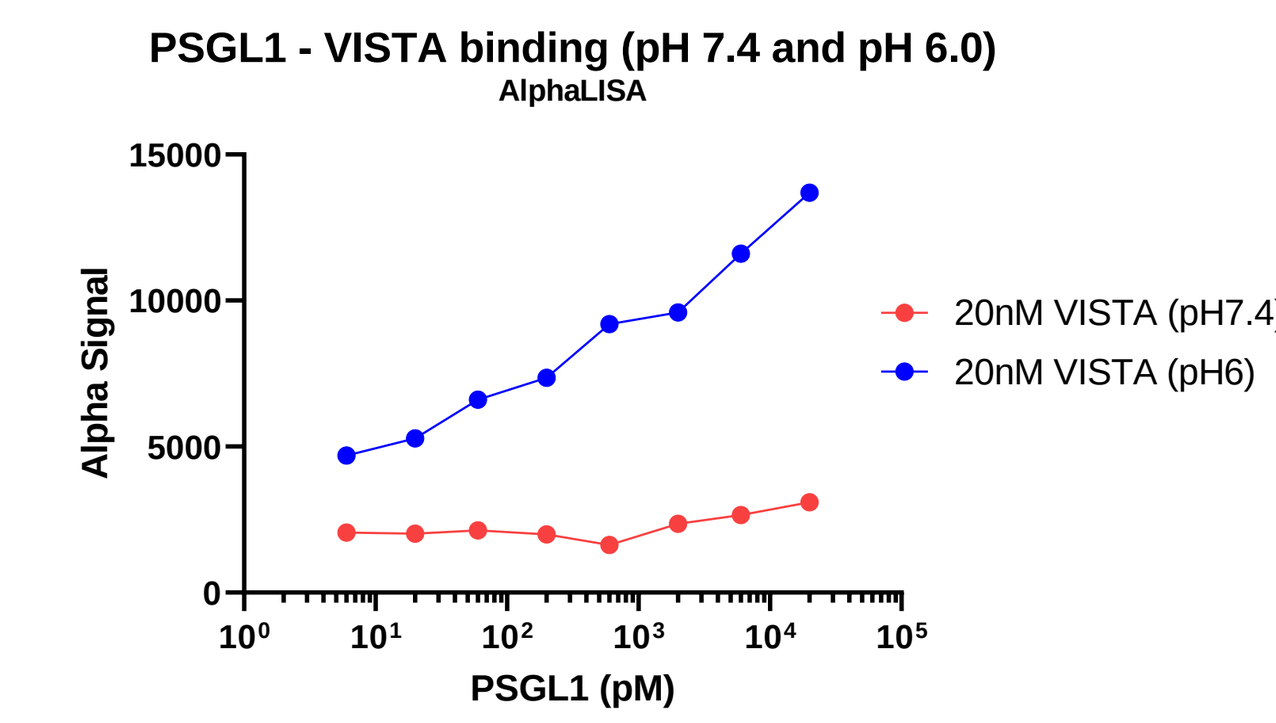
PSGL1 - VISTA binding (pH 7.4 and pH 6.0)
AlphaLISA
Alpha Signal
PSGL1 (pM)
20nM VISTA (pH7.4)
20nM VISTA (pH6)

huVISTA_His_Avi and huPSGL1_muFc used in AlphaLisa binding assay. Anti-murine IgG1 Acceptor beads (10ug/ml final) and Streptavidin Donor beads (40ug/ml final) were used, and reagents combined in either Universal AlphaLisa buffer (pH7.4) or MES buffer (pH6). Fluorescent signal was read with CLARIOstar at 615nm.

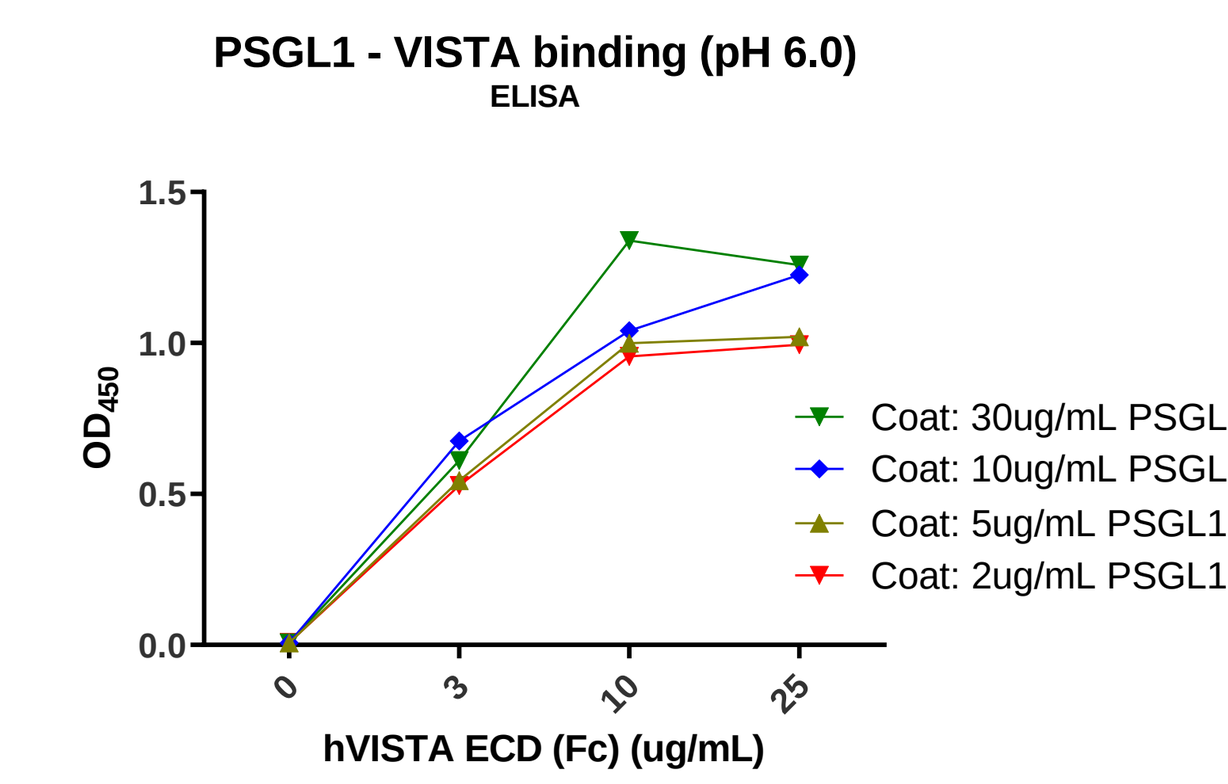
PSGL1 - VISTA binding (pH 6.0)
ELISA
hVISTA ECD (Fc) (ug/mL)
Coat: 30ug/mL PSGL1
Coat: 10ug/mL PSGL1
Coat: 5ug/mL PSGL1
Coat: 2ug/mL PSGL1

Wells were coated with 2, 5, 10, or 30 ug/ml of PSGL1 ECD (Fc). After washing, hVISTA ECD (biotinylated) was diluted to 3, 10, and 25 ug/ml and added to the wells. After incubation at room temperature for 2 hours, plates were washed. Streptavidin-HRP diluted to 1:200 was added for 30 minutes. Plates were then washed and TMB substrate added for 10 minutes, and the reaction stopped with 2N sulfuric acid. Plates were read with the CLARIOstar at 450nm.

- VISTA binds to LRIG1 at pH 7.4

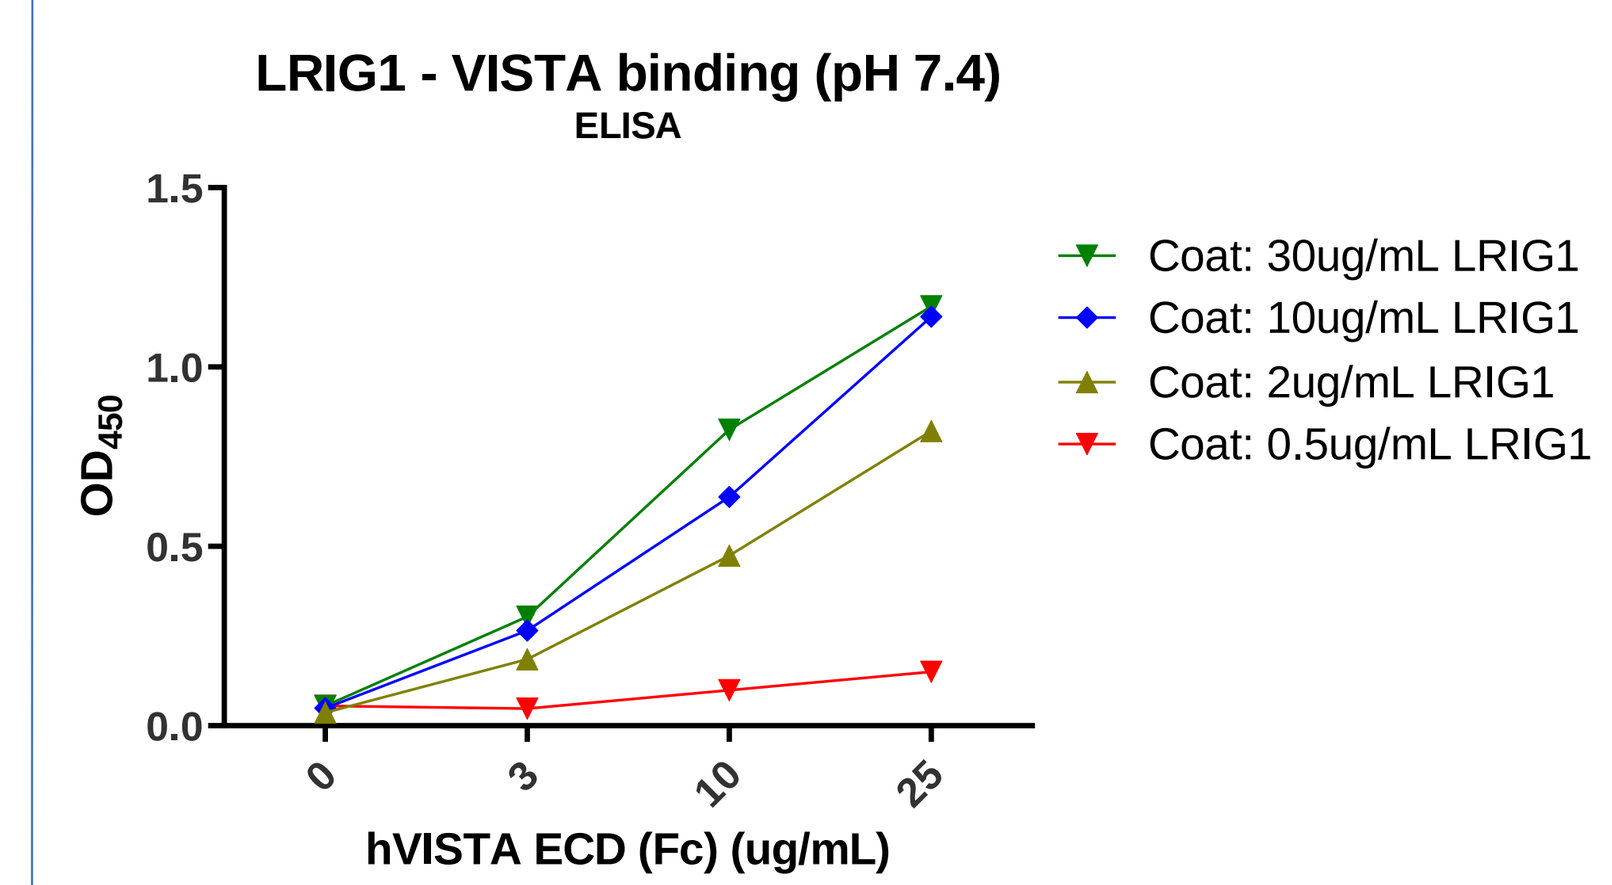
LRIG1 - VISTA binding (pH 7.4)
ELISA
hVISTA ECD (Fc) (ug/mL)
Coat: 30ug/mL LRIG1
Coat: 10ug/mL LRIG1
Coat: 2ug/mL LRIG1
Coat: 0.5ug/mL LRIG1

Wells were coated with 0.5, 2, 10, or 30 ug/ml of LRIG1 ECD (His). After washing, hVISTA ECD (biotinylated) was diluted to 3, 10, and 25 ug/ml and added to the wells. After incubation at room temperature for 2 hours, plates were washed. Streptavidin-HRP diluted to 1:200 was added for 30 minutes. Plates were then washed and TMB substrate added for 10 minutes, and the reaction was stopped with 2N sulfuric acid. Plates were read with the CLARIOstar at 450nm.

- VISTA binds to VSIG3 at pH 7.4

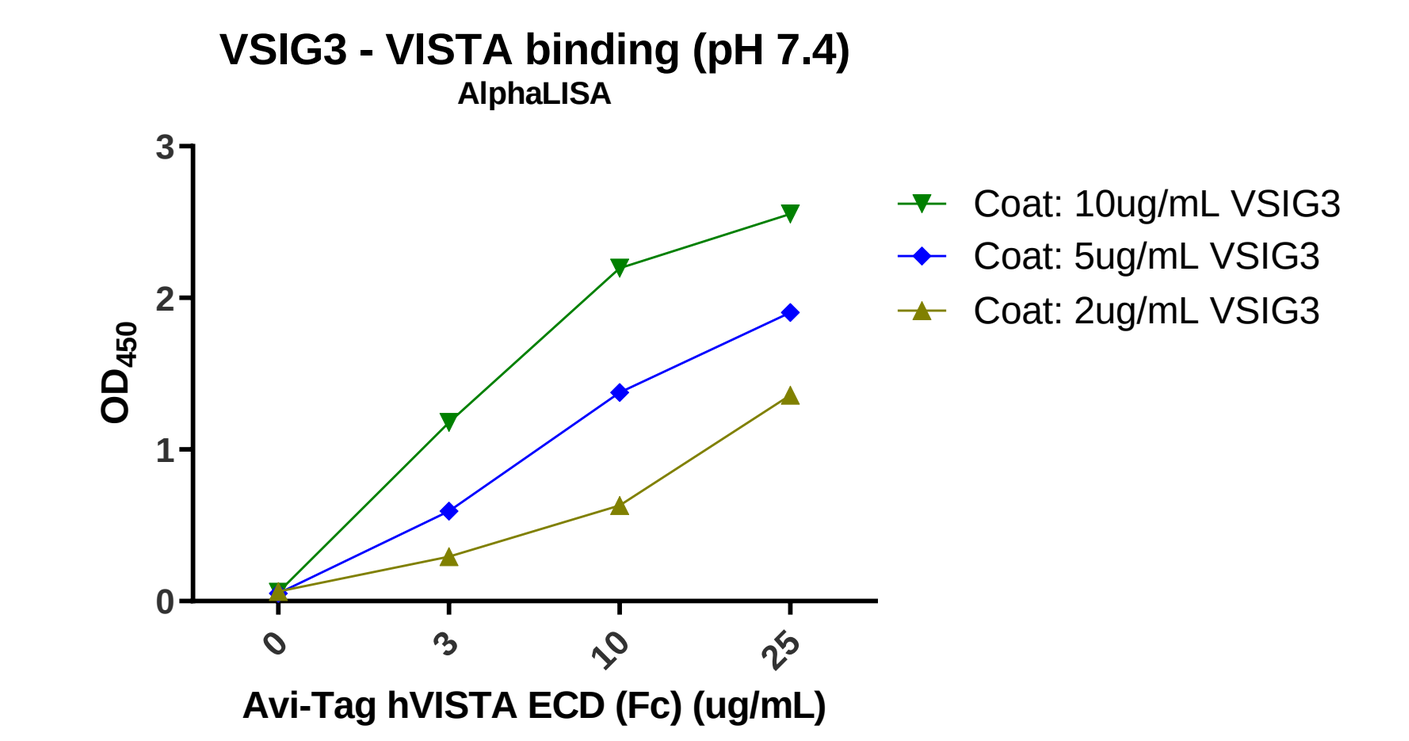
VSIG3 - VISTA binding (pH 7.4)
AlphaLISA
Avi-Tag hVISTA ECD (Fc) (ug/mL)
Coat: 10ug/mL VSIG3
Coat: 5ug/mL VSIG3
Coat: 2ug/mL VSIG3

Wells were coated with 2, 5, or 10 ug/ml of VSIG3 ECD (Fc). After washing, hVISTA ECD (AviTag) was diluted to 3, 10, and 25 ug/ml and added to the wells. After incubation at room temperature for 2 hours, plates were washed. Streptavidin-HRP diluted to 1:200 was added for 30 minutes. Plates were then washed and TMB substrate added for 10 minutes, and the reaction was stopped with 2N sulfuric acid. Plates were read with the CLARIOstar at 450nm.

- VISTA binds to VSIG8 at pH 7.4

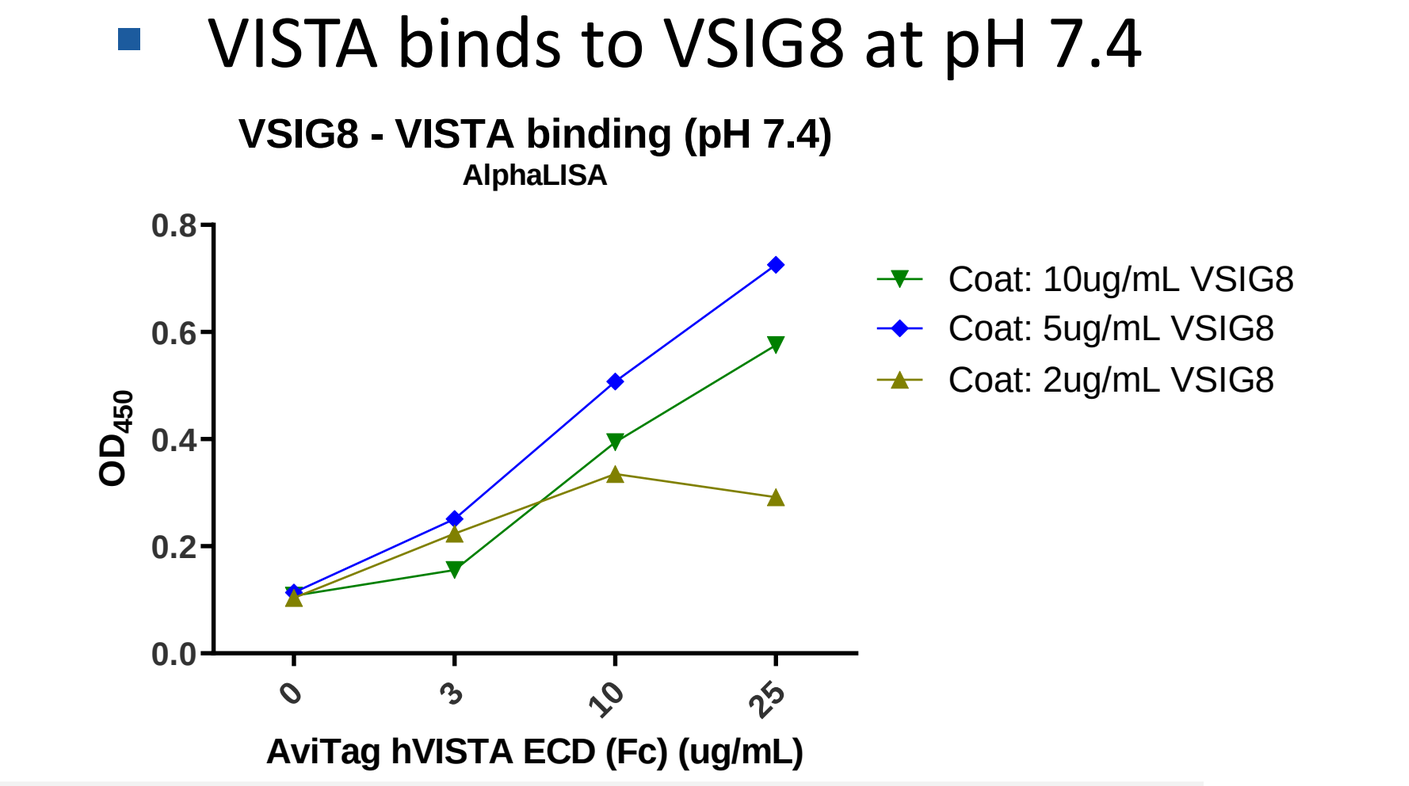
VSIG8 - VISTA binding (pH 7.4)
AlphaLISA
AviTag hVISTA ECD (Fc) (ug/mL)
Coat: 10ug/mL VSIG8
Coat: 5ug/mL VSIG8
Coat: 2ug/mL VSIG8

Wells were coated with 2, 5, or 10 ug/ml of VSIG8 ECD (Fc). After washing, hVISTA ECD (AviTag) was diluted to 3, 10, and 25 ug/ml and added to the wells. After incubation at room temperature for 2 hours, plates were washed. Streptavidin-HRP diluted to 1:200 was added for 30 minutes. Plates were then washed and TMB substrate added for 10 minutes, and the reaction was stopped with 2N sulfuric acid. Plates were read with the CLARIOstar at 450nm.

Kineta's VISTA antibodies block VISTA interaction with PSGL-1, VSIG3, VSIG8 and LRIG1

- KVA Abs Disrupt VISTA-PSGL1 Interaction

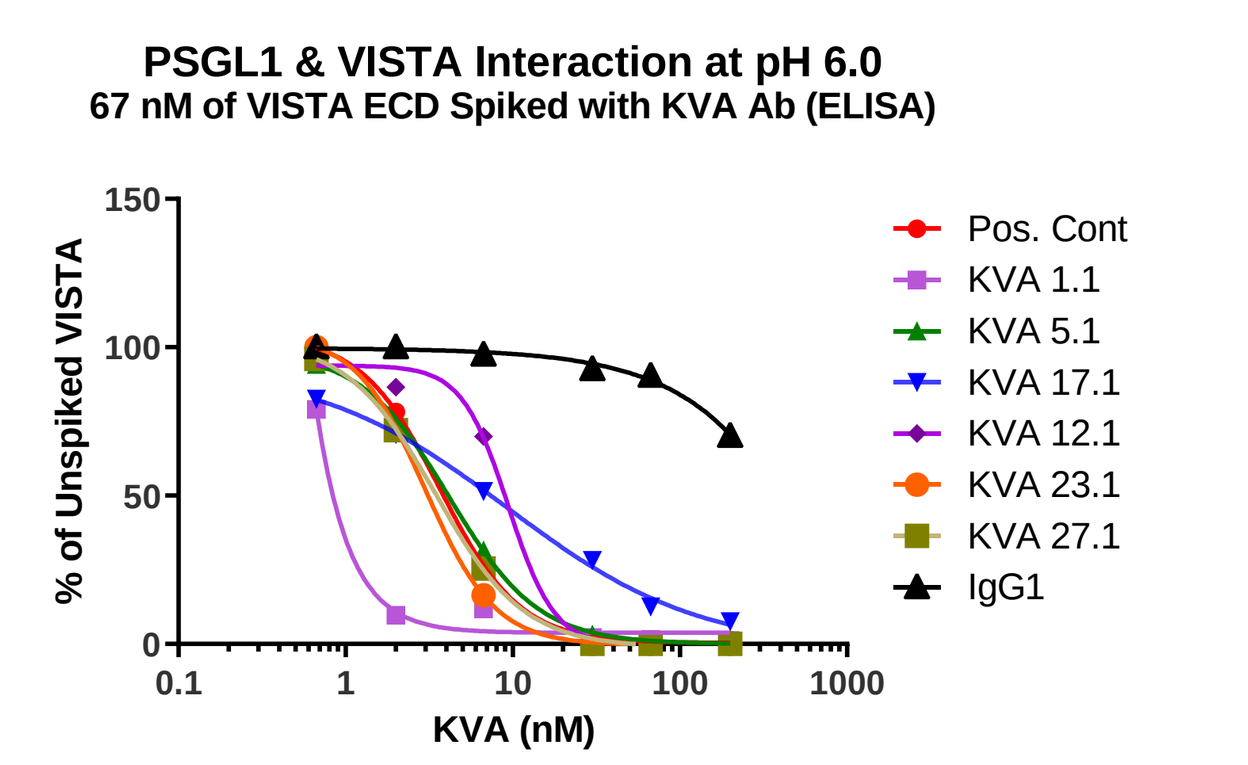
PSGL1 & VISTA Interaction at pH 6.0
67 nM of VISTA ECD Spiked with KVA Ab (ELISA)
% of Unspiked VISTA
KVA (nM)
Pos. Cont
KVA 1.1
KVA 5.1
KVA 17.1
KVA 12.1
KVA 23.1
KVA 27.1
IgG1

Ab	EC50 (nM)
Pos. Cont	3.7
KVA 1.1	~ 0.04
KVA 5.1	4.2
KVA 17.1	9.3
KVA 12.1	9.3
KVA 23.1	2.8
KVA 27.1	3.5
IgG1	N/A

Wells were coated with 2 ug/ml of PSGL1 ECD (Fc). HuVISTA ECD (biotinylated) at 3 ug/ml was spiked with anti-VISTA antibodies and incubated for 1h at RT. HuVISTA ECD spiked with anti-VISTA antibodies were added to the wells. After incubation at RT for 2 hours, plates were washed and Streptavidin-HRP diluted to 1:200 was added for 30 minutes. After developing for 10 minutes, the reaction was stopped with 2N sulfuric acid and the plates were read with the CLARIOstar microplate reader at 450nm.

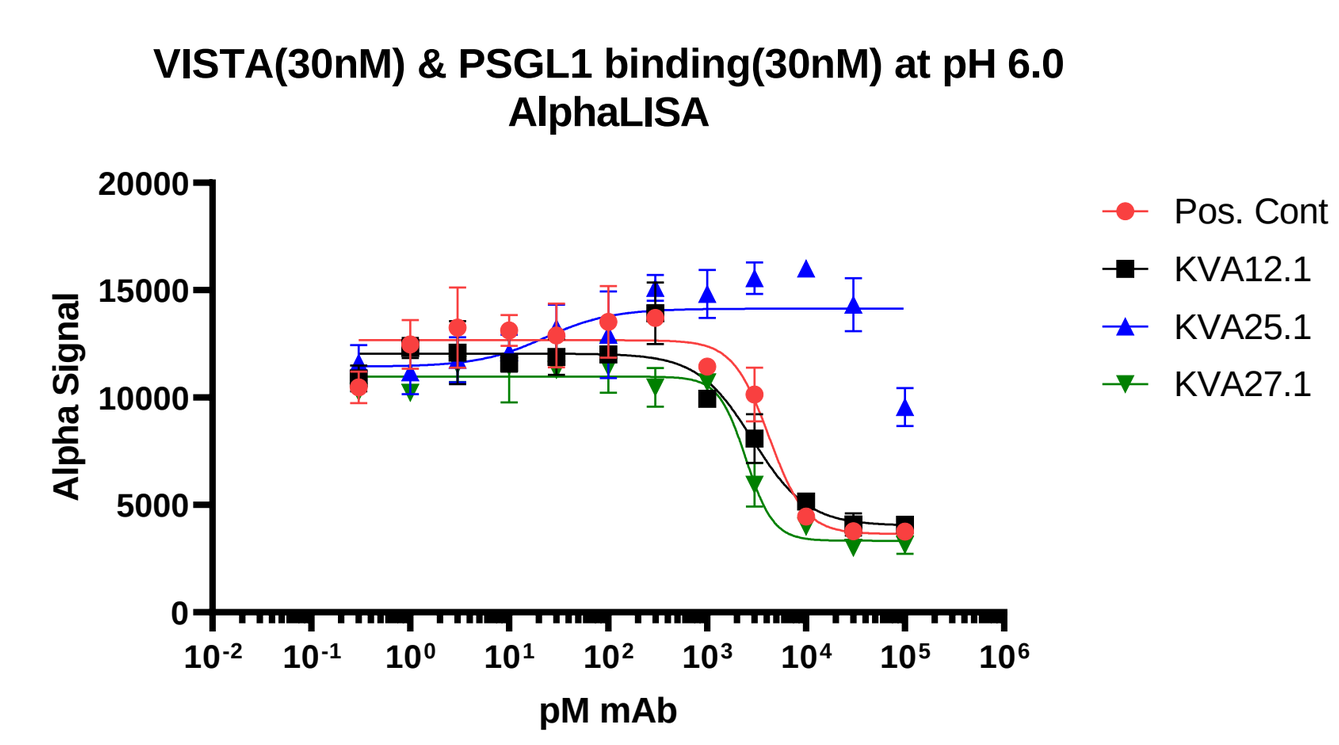
VISTA(30nM) & PSGL1 binding(30nM) at pH 6.0
AlphaLISA
Alpha Signal
pM mAb
Pos. Cont
KVA12.1
KVA25.1
KVA27.1

	Pos. Cont	KVA12.1	KVA25.1	KVA27.1
IC50 nM	4.2	2.8	NA	2.4

10ul mAbs (dilute from 0.5uM, 100nM final) was added to 30nM VISTA ECD and 10ul PSGL1 (30nM Final) and incubated for 1hr. Then, 15ul anti-Ms IgG1 Acceptor (33.3ug/ml, 10ug/ml final) was added and incubated for 1hr. Finally, 10ul of Streptavidin Donor (133.3ug/ml, 40ug/ml final) was added and incubated for 1hr and fluorescence was read at 615nm using a CLARIOstar spectrometer.

- KVA Abs Disrupt VISTA-VSIG3 Interaction

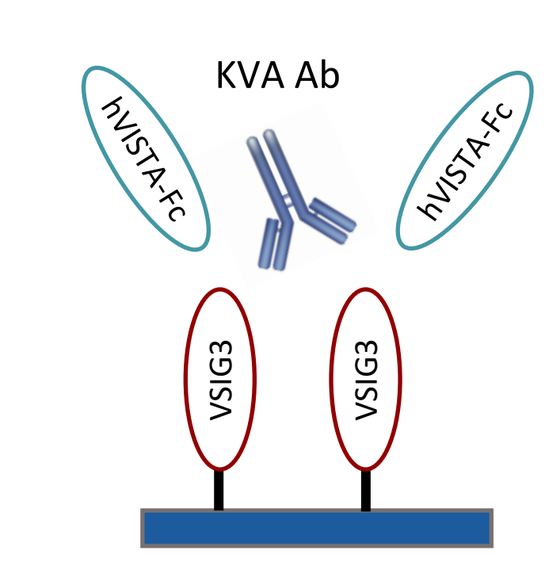
KVA Ab
hVISTA-Fc
hVISTA-Fc
VSIG3
VSIG3

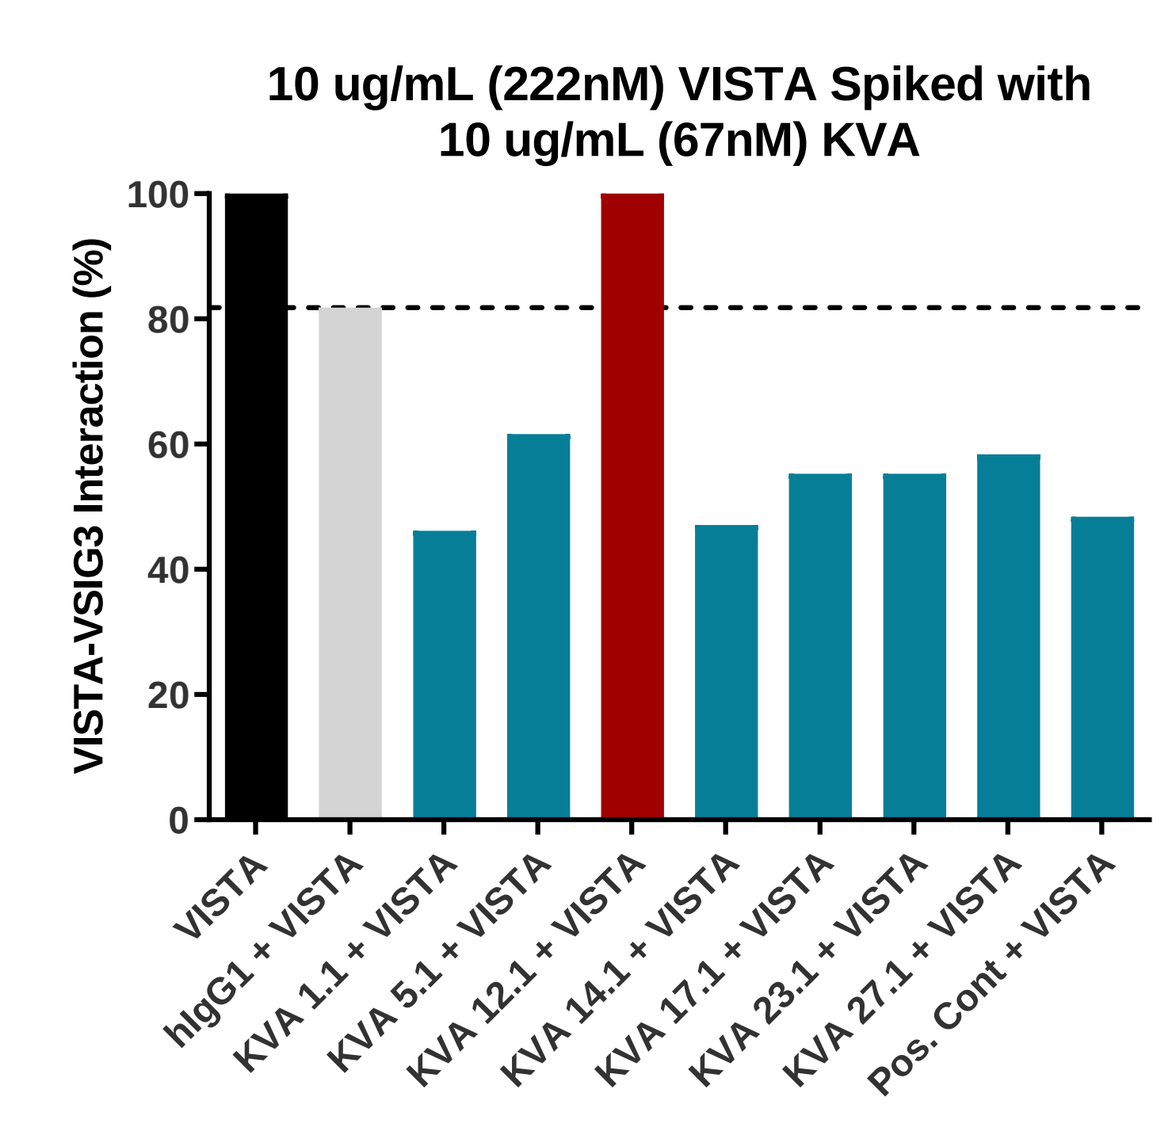
10 ug/mL (222nM) VISTA Spiked with 10 ug/mL (67nM) KVA
VISTA-VSIG3 Interaction (%)
VISTA
hIgG1 + VISTA
KVA 1.1 + VISTA
KVA 5.1 + VISTA
KVA 12.1 + VISTA
KVA 14.1 + VISTA
KVA 17.1 + VISTA
KVA 23.1 + VISTA
KVA 27.1 + VISTA
Pos. Cont + VISTA

Spiked Ab	% of Unspiked VISTA
10ug/ml Ab	
KVA 1.1	46%
KVA 5.1	62%
KVA 12.1	100%
KVA 14.1	47%
KVA 17.1	55%
KVA 23.1	55%
KVA 27.1	58%
Pos. Cont	48%
IgG1	82%

Spiking 10 ug/ml KVA (KVA 1.1, 5.1, 14.1, 17.1, 23.1, 27.1, Pos. Cont) into 10 ug/ml VISTA Fc blocked binding of VISTA to VISG3

- KVA Abs Disrupt VISTA-LRIG1 Interaction

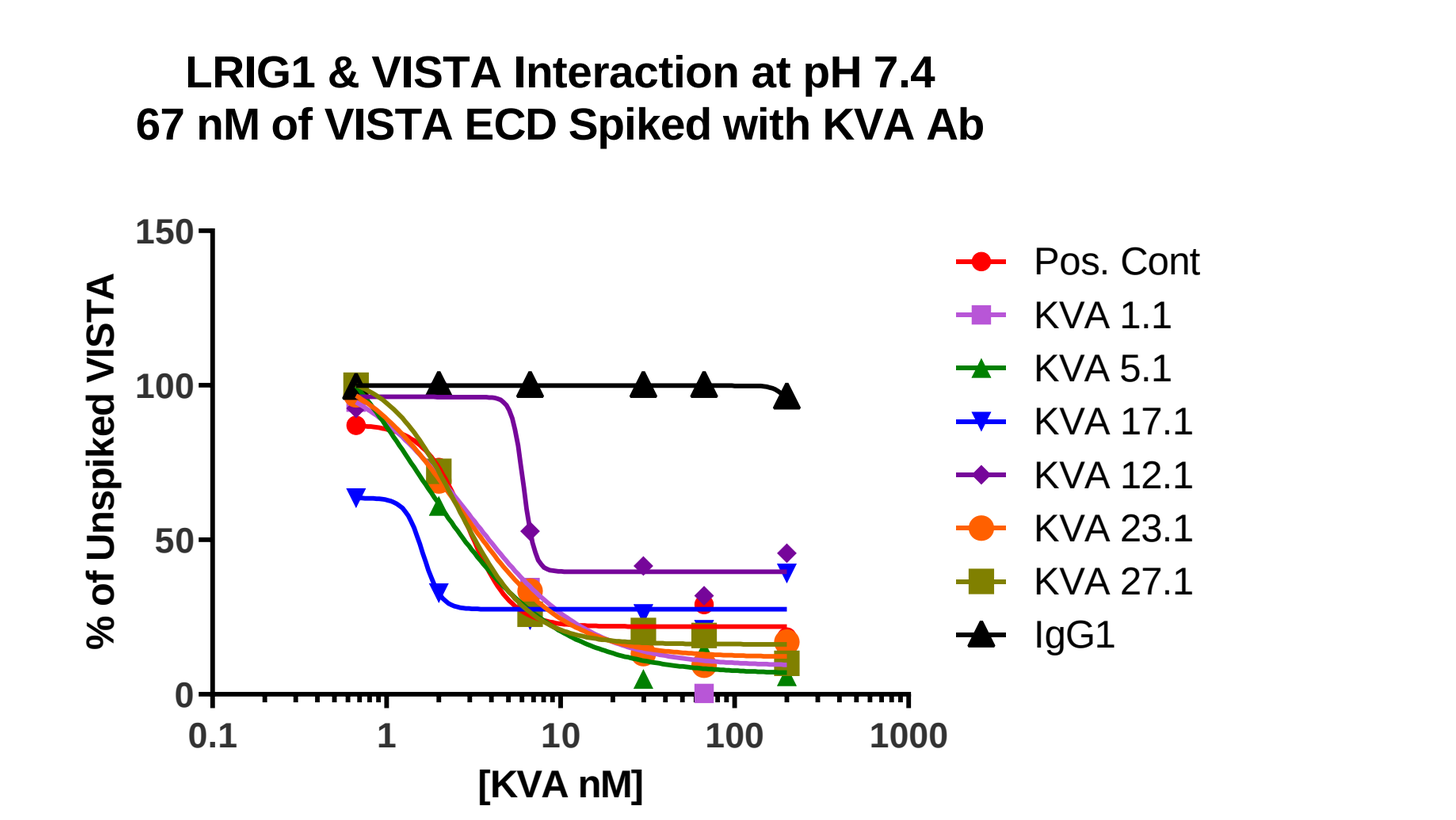
LRIG1 & VISTA Interaction at pH 7.4
67 nM of VISTA ECD Spiked with KVA Ab
% of Unspiked VISTA
[KVA nM]
Pos. Cont
KVA 1.1
KVA 5.1
KVA 17.1
KVA 12.1
KVA 23.1
KVA 27.1
IgG1

Ab	EC50 (nM)
Pos. Cont	2.9
KVA 1.1	3.0
KVA 5.1	1.5
KVA 17.1	N/A
KVA 12.1	~ 6.1
KVA 23.1	2.6
KVA 27.1	2.5
IgG1	N/A

Wells were coated with 10 ug/ml of LRIG1 ECD (His). HuVISTA ECD (biotinylated) at 3 ug/ml was spiked with anti-VISTA antibodies and incubated for 1h at RT. HuVISTA ECD spiked with anti-VISTA antibodies were added to the wells. After incubation at RT for 2 hours, plates were washed and Streptavidin-HRP diluted to 1:200 was added for 30 minutes. After developing for 10 minutes, the reaction was stopped with 2N sulfuric acid and the plates were read with the CLARIOstar microplate reader at 450nm.

- KVA Abs Disrupt VISTA-VSIG8 Interaction

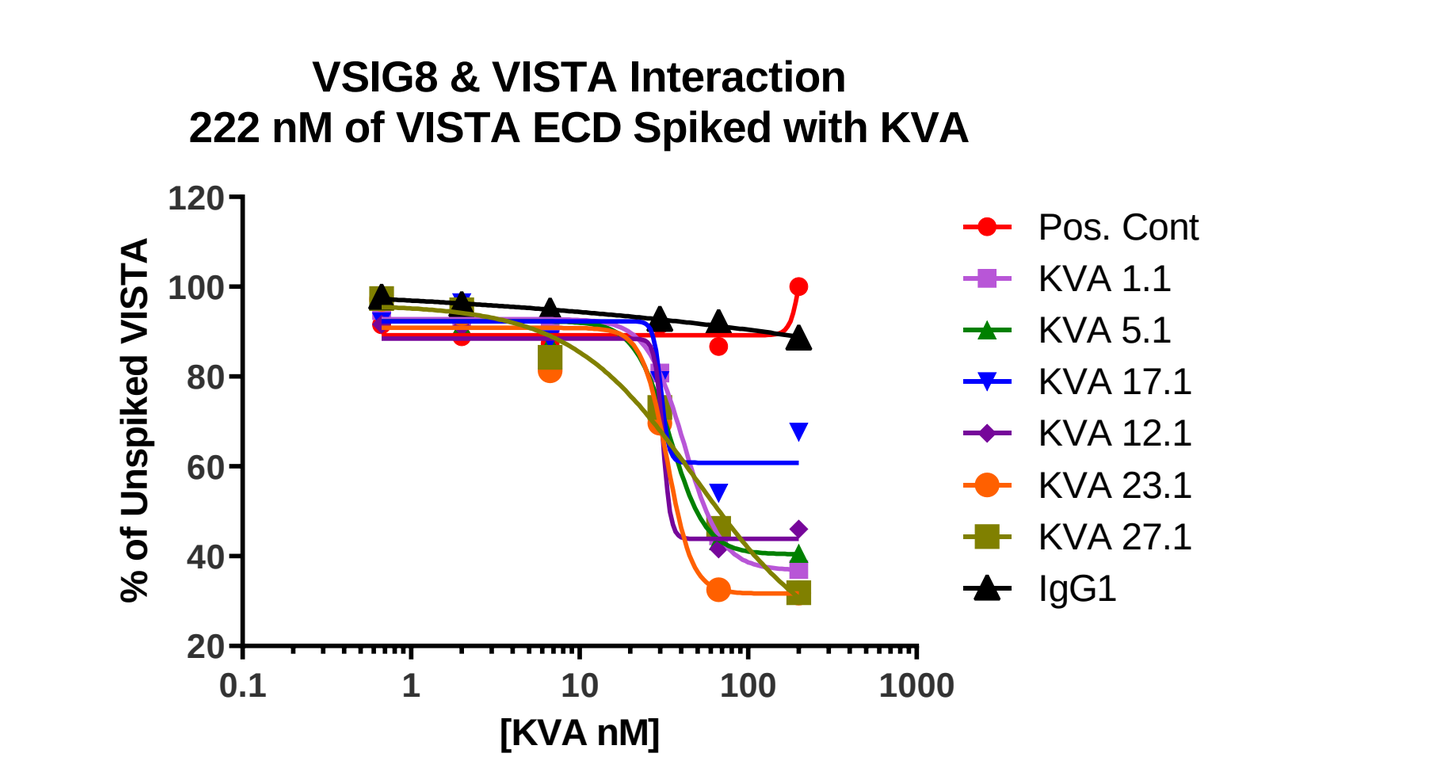
VSIG8 & VISTA Interaction
222 nM of VISTA ECD Spiked with KVA
% of Unspiked VISTA
[KVA nM]
Pos. Cont
KVA 1.1
KVA 5.1
KVA 17.1
KVA 12.1
KVA 23.1
KVA 27.1
IgG1

Ab	EC50 (nM)
Pos. Cont	N/A
KVA 1.1	41.81
KVA 5.1	34.46
KVA 17.1	~ 30.23
KVA 12.1	~ 31.23
KVA 23.1	33.14
KVA 27.1	49.63
IgG1	N/A

Wells were coated with 5 ug/ml of VSIG8 ECD (Fc). HuVISTA ECD (biotinylated) at 10 ug/ml was spiked with anti-VISTA antibodies and incubated for 1h at RT. HuVISTA ECD spiked with anti-VISTA antibodies were added to the wells. After incubation at RT for 2 hours, plates were washed and Streptavidin-HRP diluted to 1:200 was added for 30 minutes. After developing for 10 minutes, the reaction was stopped with 2N sulfuric acid and the plates were read with the CLARIOstar microplate reader at 450nm.

Conclusion

- VISTA is highly expressed on Myeloid cells as well as Granulocytes, NK and NKT cells.
- VISTA is also expressed on Treg and modestly on CD4 and CD8 cells. CD4/CD69 activated T cells seems to be high expresser.
- PSGL-1 is highly expressed on all immune cells.
- LRIG-1 has low level expression on Myeloid cells as well as CD4+ T cells.
- VISTA binds to the 5 putative receptors already identified including itself at neutral or acidic pH.
- Kineta's anti-VISTA antibodies selectively inhibit these interactions with different potencies.