Highly Potent Fully Human anti-VISTA Antibodies
A New Target Checkpoint Inhibitor against Immunosuppressive Myeloid Cells.

Thierry Guillaudeux[1], Eric Tarcha[1], Tina Albershardt[1], Robert Bader[1], Benjamin Dutzar[1], Nathan Eyde[1], Emily Frazier[1], David Jurchen[1], Remington Lance[1], Cristina Loomis[1], Kurt Lustig[1], Yulia Ovechkina[1], David Peckham[1], Jeff Posakony[1], Rebecca Reeves[1], Shaarwari Sridhar[1], Mei Xu[1] and Shawn Iadonato[1].

[1] Kineta Inc. 219 Terry Av. North, Seattle, WA 98109.

Background

- VISTA (V-domain Ig Suppressor of T cell Activation) is a unique B7 family member that functions as both ligand and receptor
- VISTA is mainly expressed in Immune cells and mostly on Myeloid cells, Neutrophils, NK cells and Treg
- VISTA is highly expressed on MDSC and Treg in the TME
- VISTA is a negative regulator that directly suppresses T cell activation and proliferation
- High VISTA expression correlates with poor survival in cancer patients
- VISTA is a unique Immune checkpoint inhibitor for tumor immunotherapy

VISTA is predominantly expressed in the myeloid lineage within peripheral blood

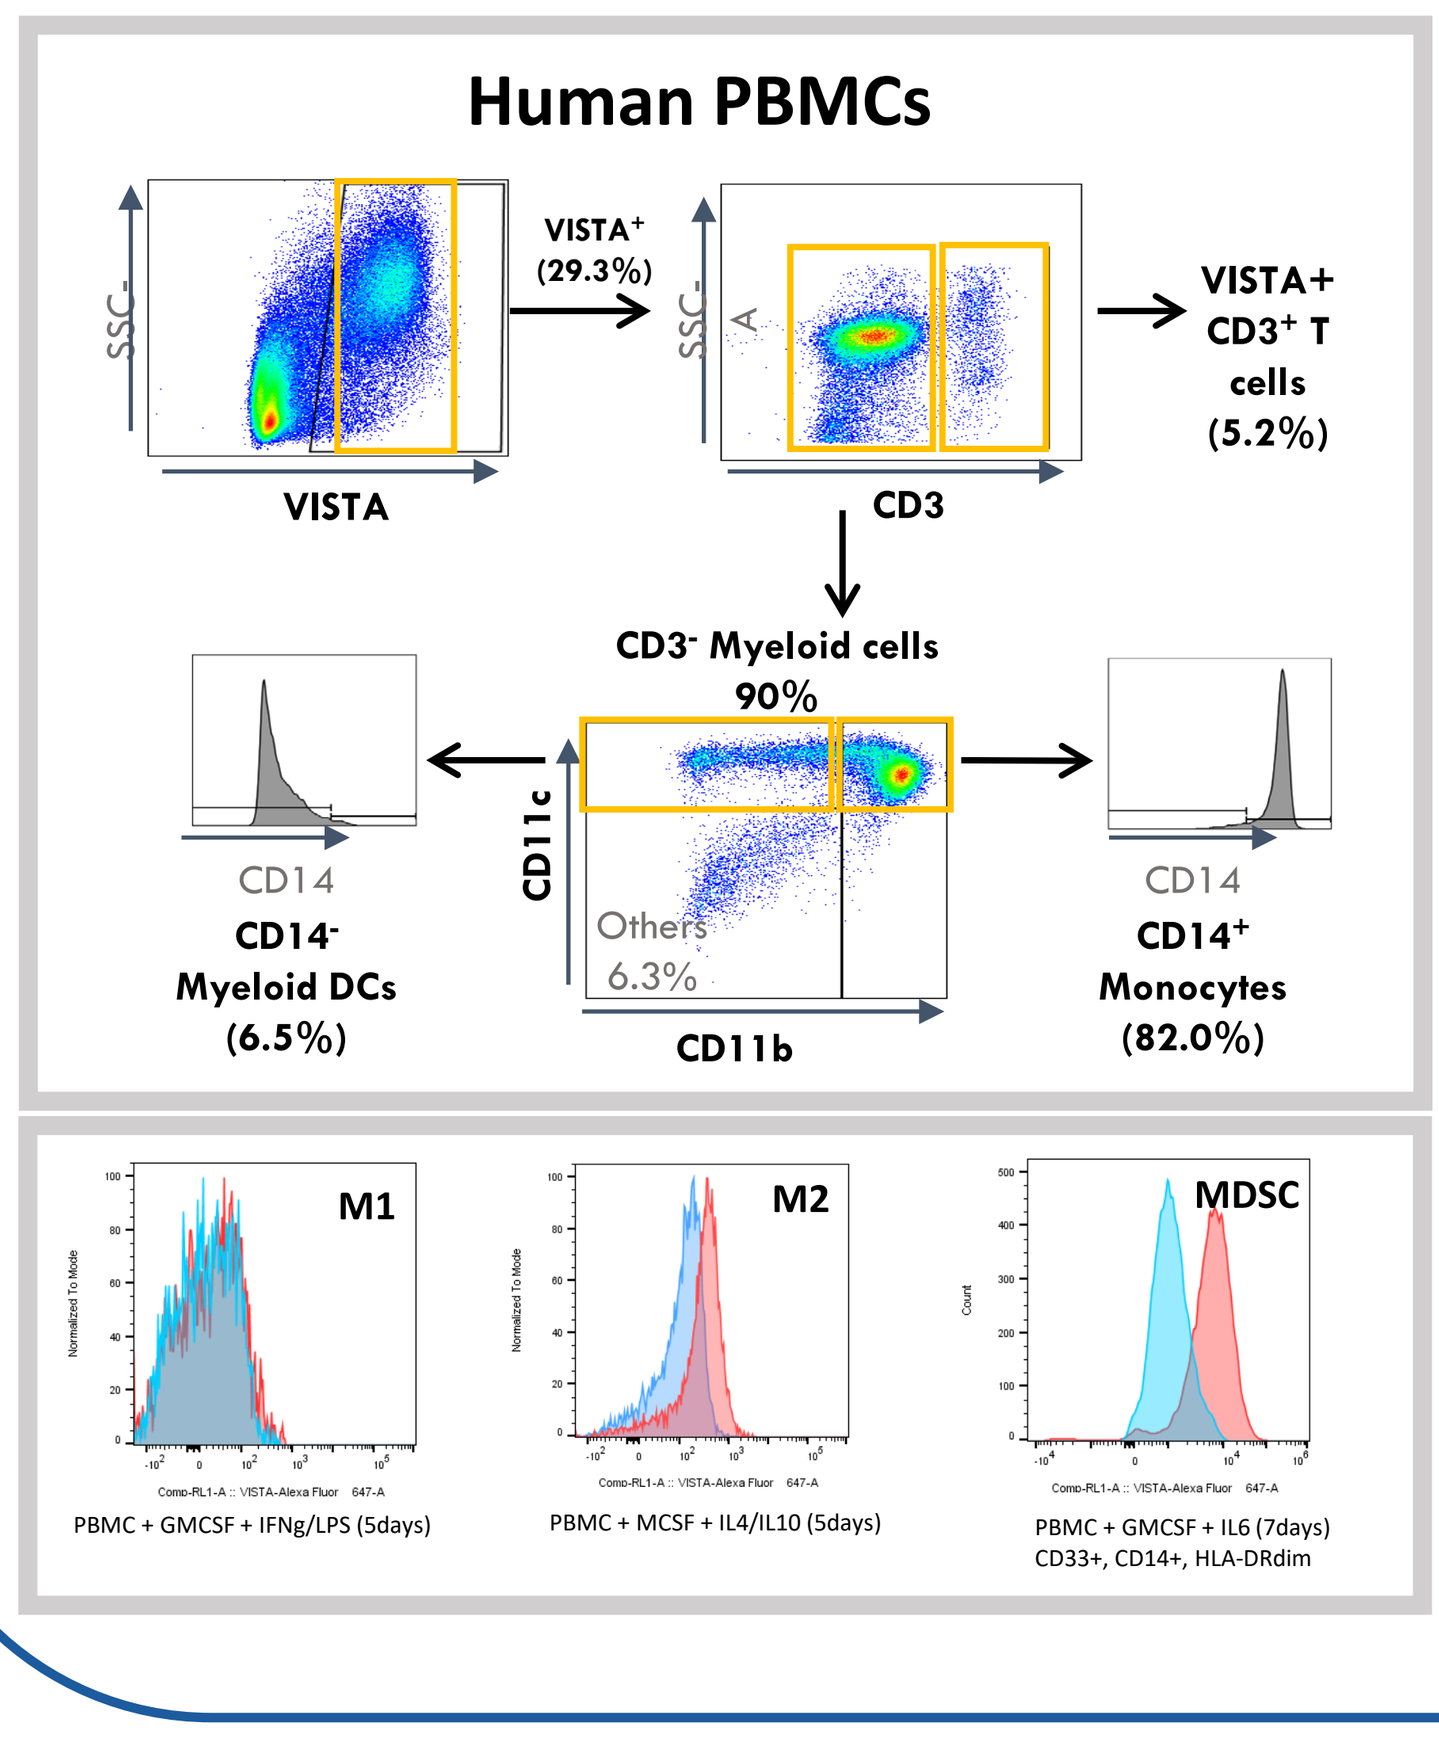
Human PBMCs
VISTA⁺ (29.3%)
VISTA
CD3
VISTA+ CD3⁺ T cells (5.2%)
CD3⁻ Myeloid cells 90%
CD14⁻ Myeloid DCs (6.5%)
CD14⁺ Monocytes (82.0%)
CD11c
CD11b
Others 6.3%
M1
M2
MDSC
PBMC + GMCSF + IFNg/LPS (5days)
PBMC + MCSF + IL4/IL10 (5days)
PBMC + GMCSF + IL6 (7days) CD33+, CD14+, HLA-DRdim

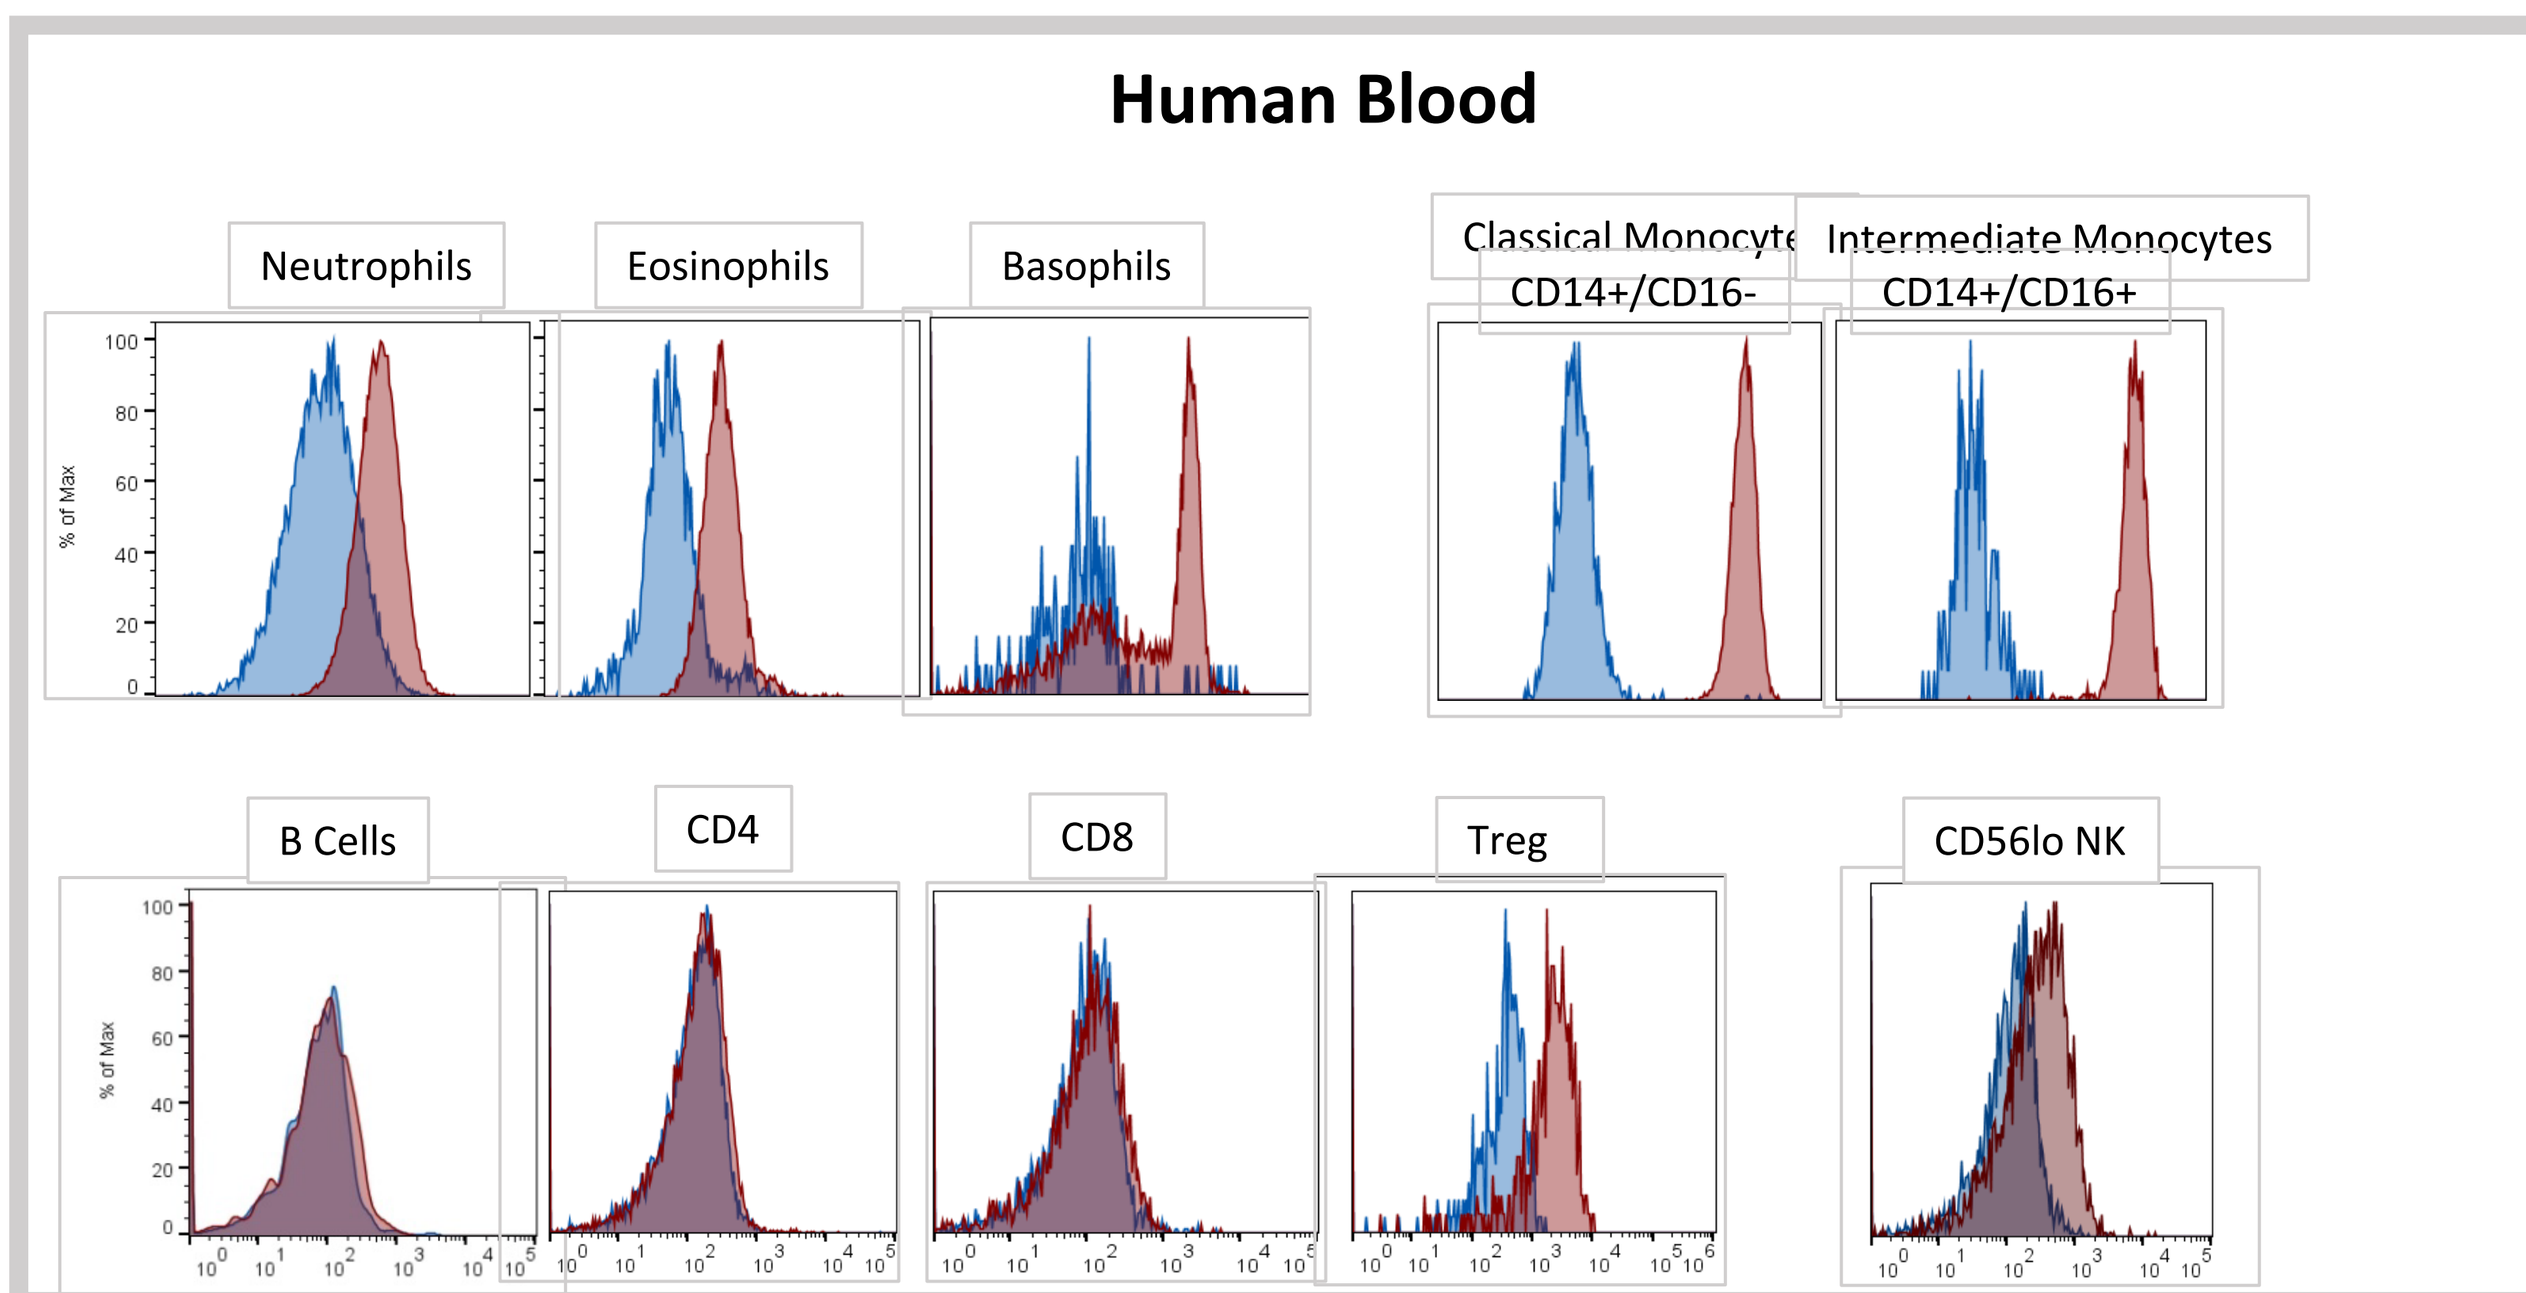
Human Blood
Neutrophils
Eosinophils
Basophils
Classical Monocytes CD14+/CD16-
Intermediate Monocytes CD14+/CD16+
B Cells
CD4
CD8
Treg
CD56lo NK

- Granulocytes, Monocytes, Dendritic cells, Treg and NK cells highly express VISTA on their surface
- M2 and MDSC differentiated cells in vitro highly express VISTA

VISTA Has Profound Negative Regulatory Effect on T Cell Activation and Proliferation

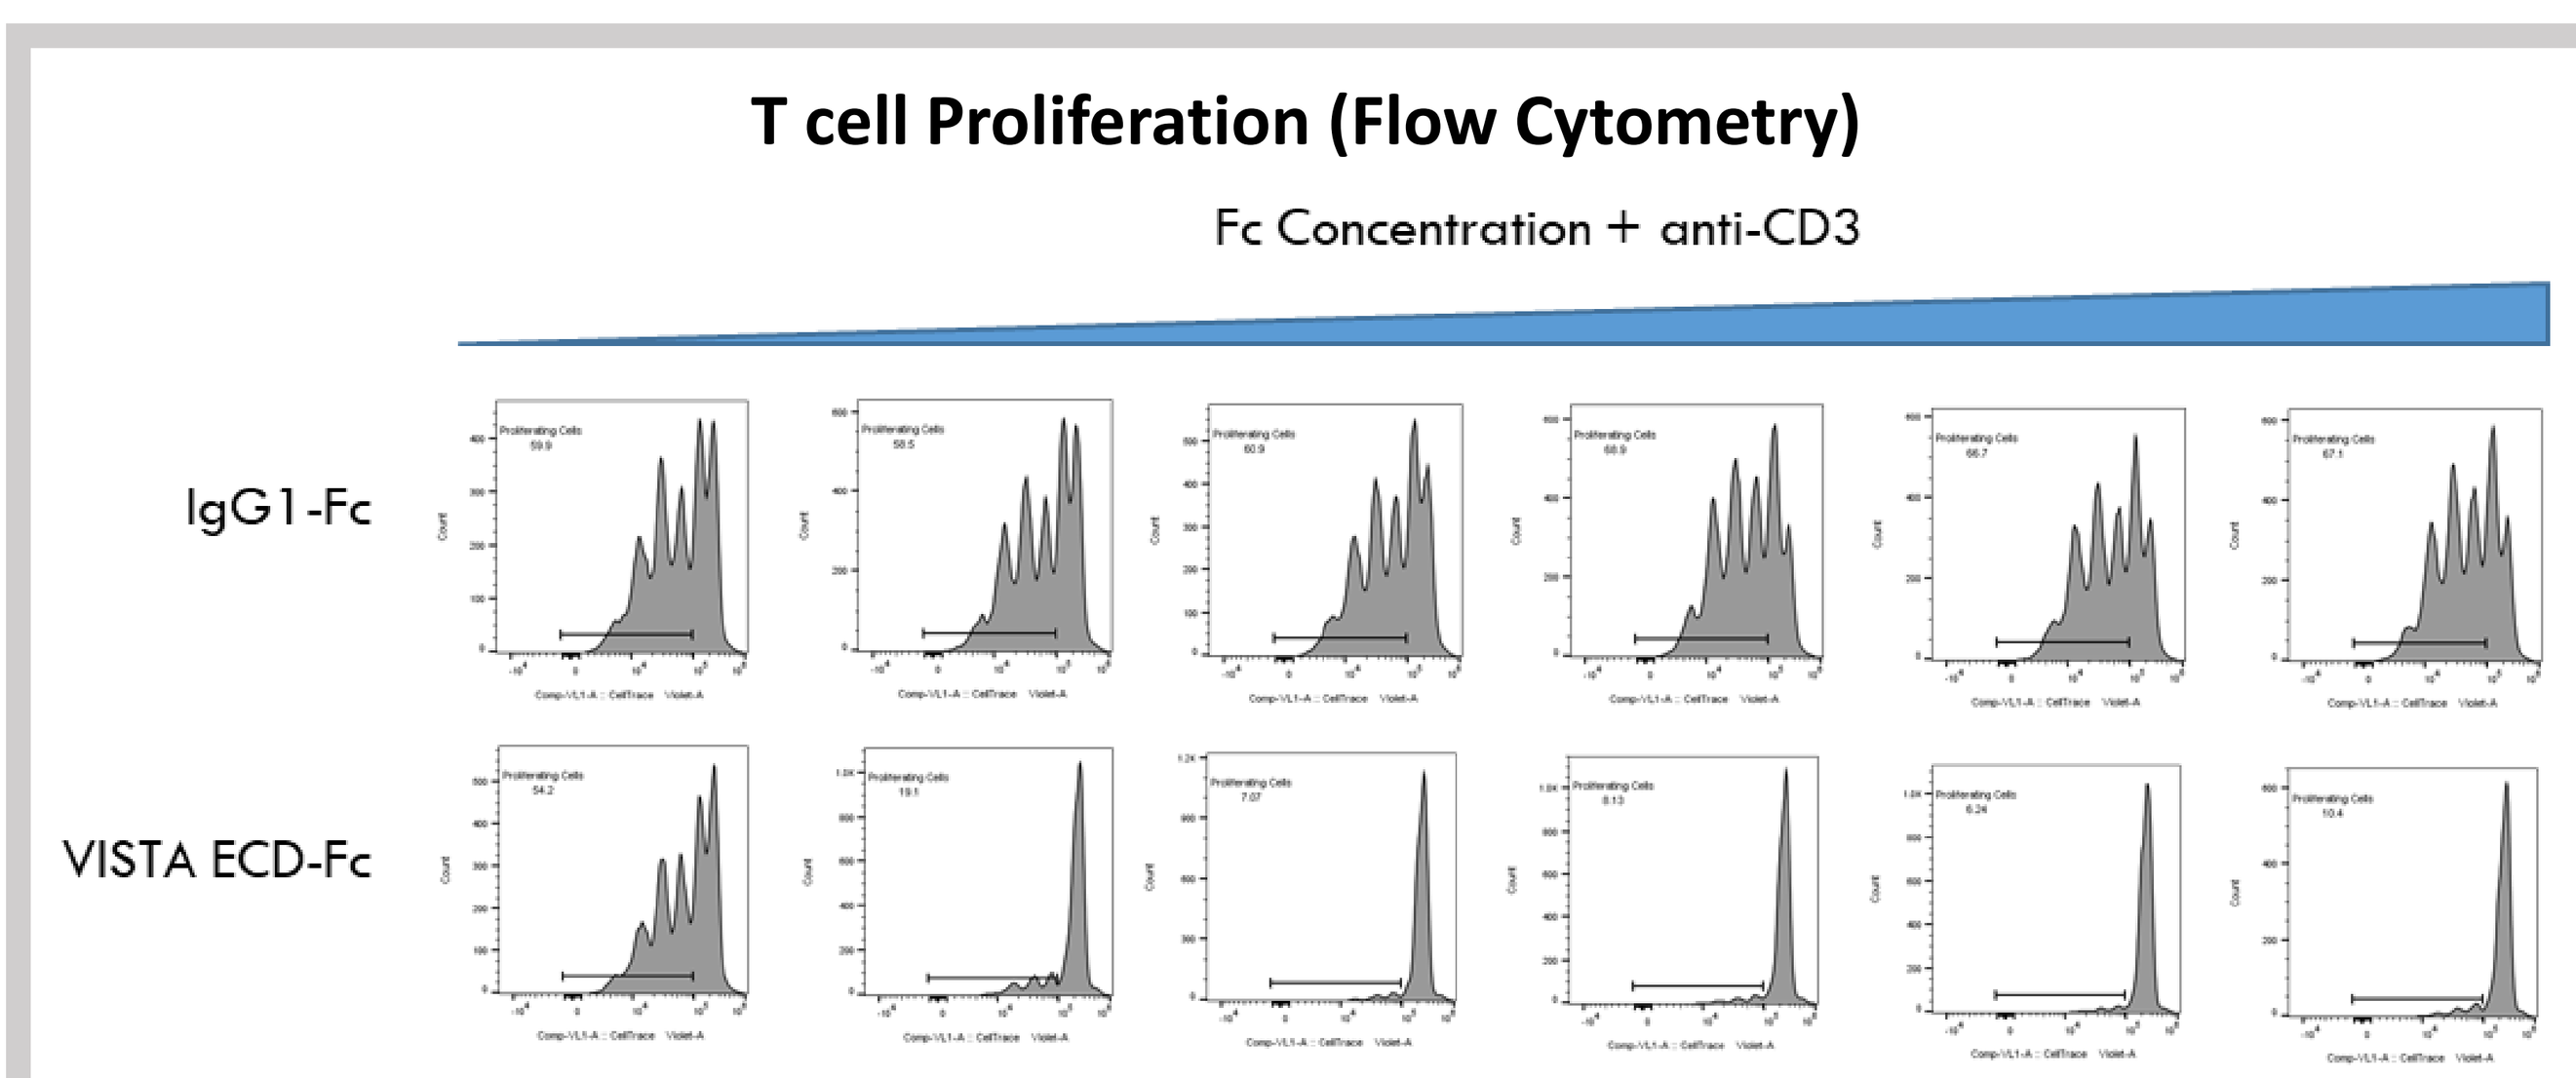
T cell Proliferation (Flow Cytometry)
Fc Concentration + anti-CD3
IgG1-Fc
VISTA ECD-Fc

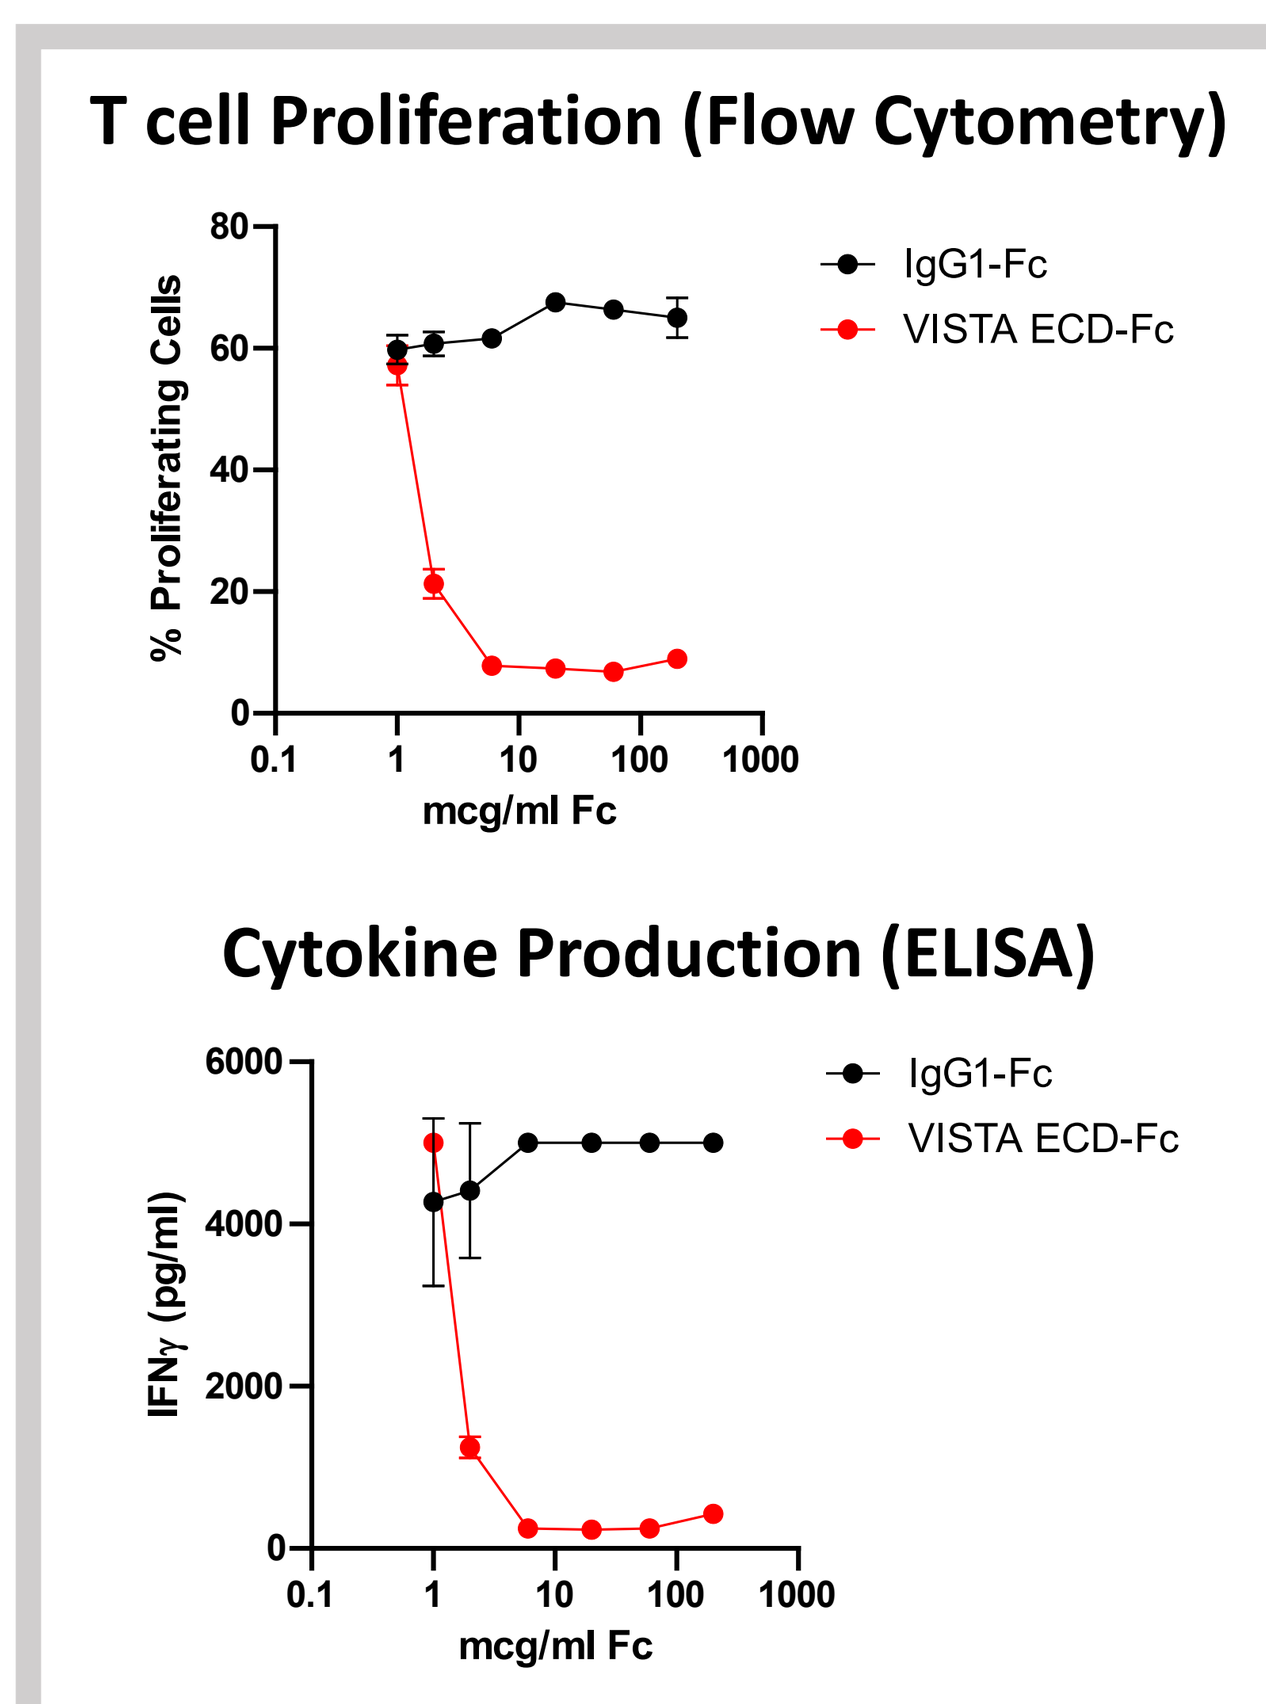
T cell Proliferation (Flow Cytometry)
% Proliferating Cells
mcg/ml Fc
IgG1-Fc
VISTA ECD-Fc
Cytokine Production (ELISA)
IFNγ (pg/ml)

Figure 1: Effect of VISTA-Fc on anti-CD3-induced T cell proliferation. Immobilized VISTA-Fc or IgG1-Fc was incubated on plates with immobilized anti-CD3 (OKT3), and the ability of VISTA to suppress anti-CD3-induced T cell proliferation and cytokine production was assessed. 96-well flat-bottomed plates were coated with 2 mcg/mL anti-CD3 +/- 0, 2, 6, 20 or 60 mcg/ml VISTA-Fc or IgG1-Fc. Human pan-T cells from healthy donor were labeled with CellTrace Violet and plated at 2E5 cells/well. After 4 days of incubation, T cell proliferation was analyzed via flow cytometry; IFNγ concentrations were measured by ELISA. Representative graphs are shown. Means of % proliferating cells or IFNγ production ± SEM are graphed.

Objectives

- Screen for specific fully human anti-VISTA antibodies
- Evaluate and select highly potent VISTA antagonist antibodies in vitro and in vivo

- **Immunization of humanized Trianni® mice**
 - *In vivo* affinity maturation in mouse leads to high affinity antibodies
 - VDJ recombination allows unique fully human variable domains
- Microfluidics-based sequencing of single B cell clones
- Native pairing of heavy and light chains
- Flow-based enrichment using yeast ScFv display libraries

Results

Exceptional Antibody Diversity in Both Heavy and Light Chain

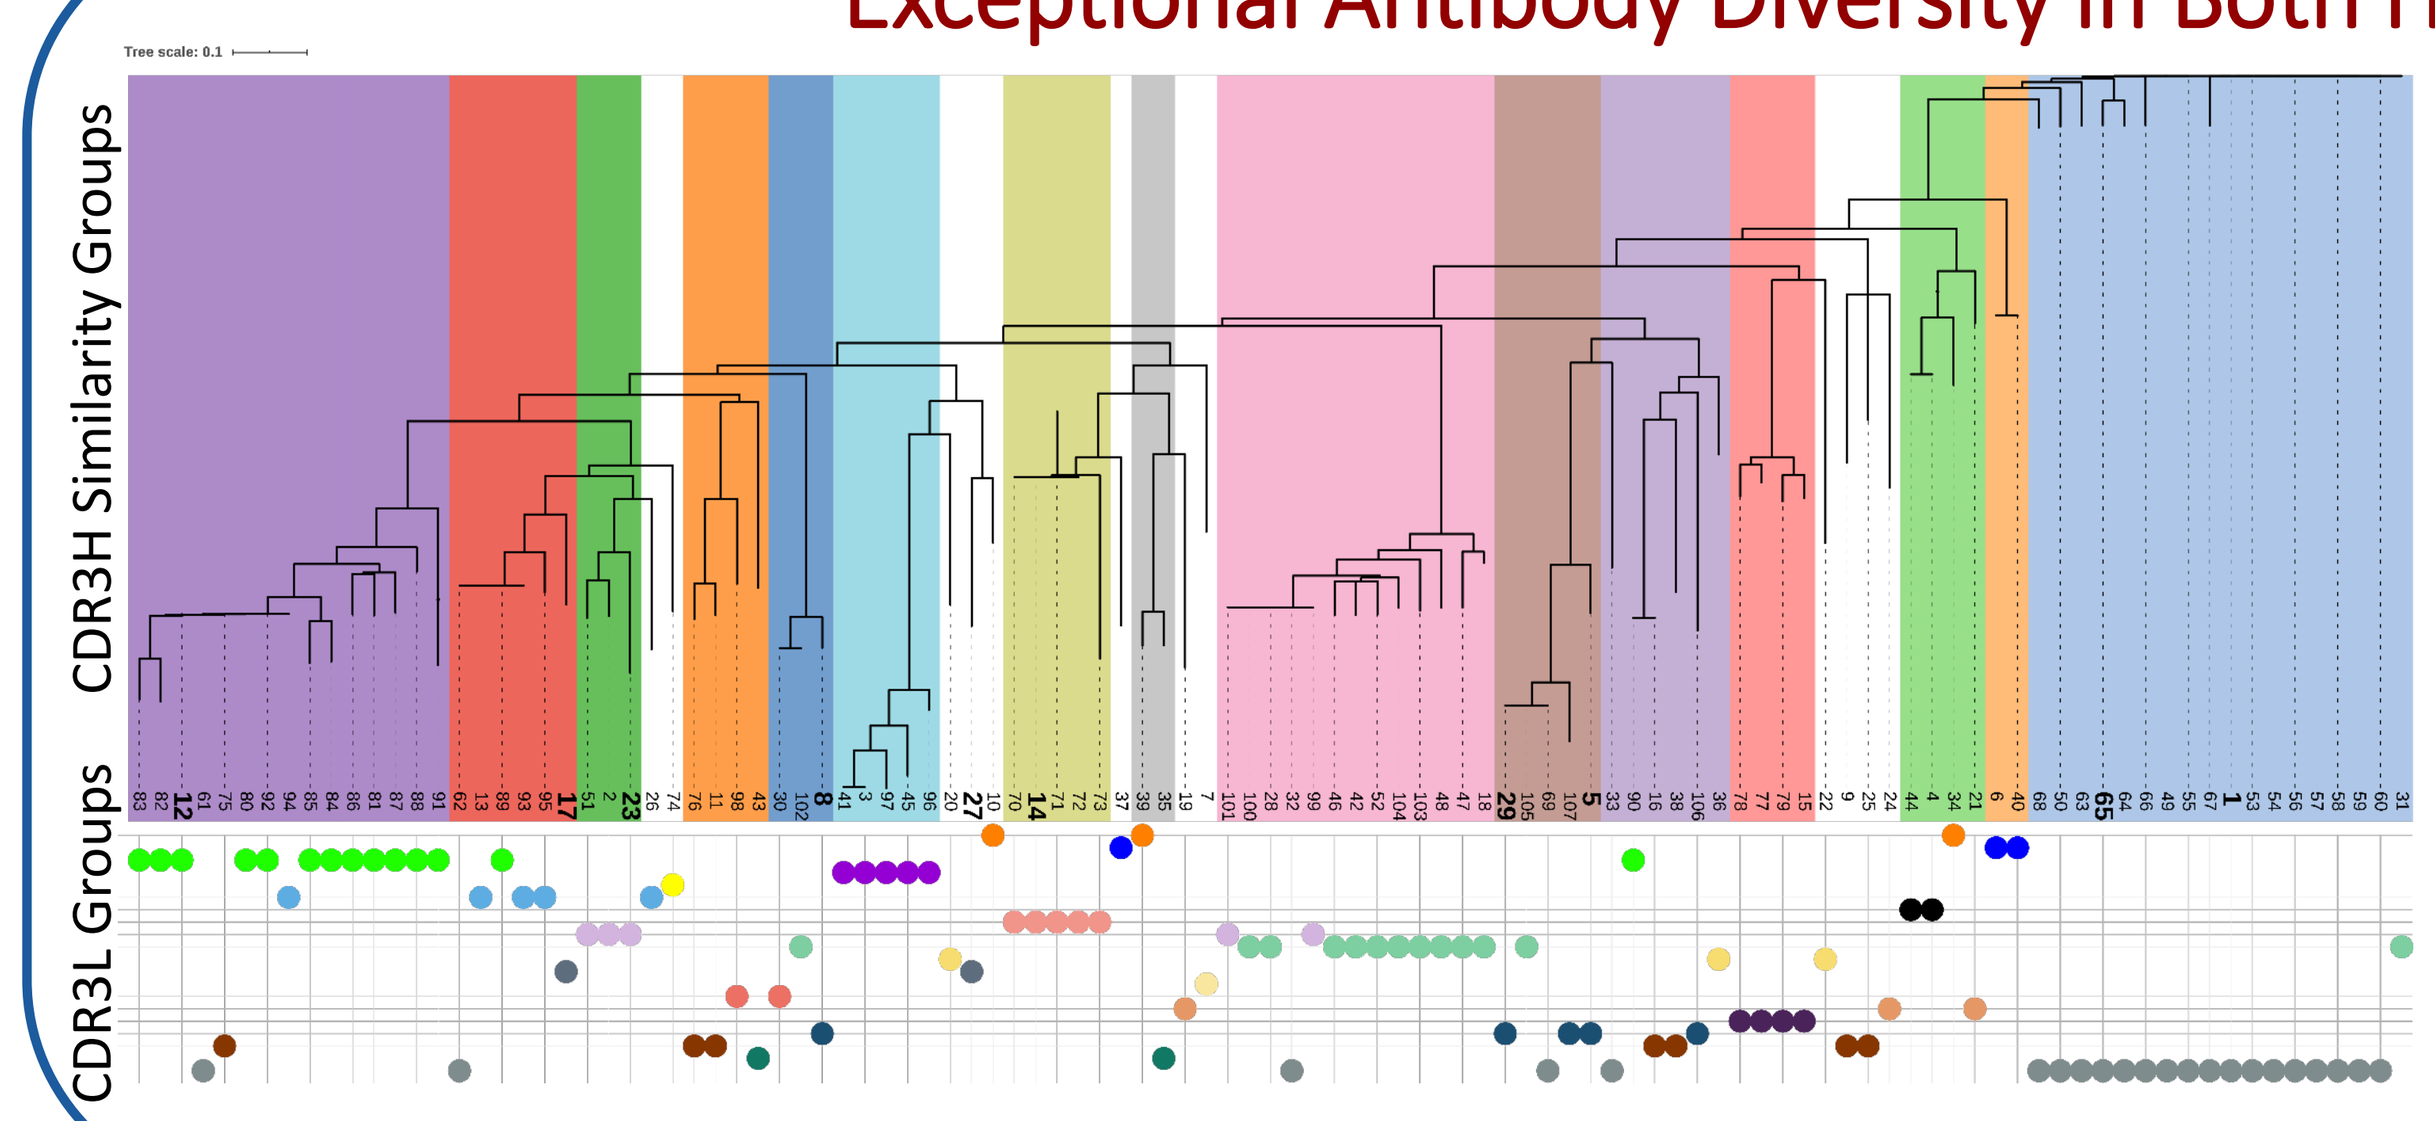
CDR3H Similarity Groups
CDR3L Groups

107 fully human ScFv antibodies directed against Human VISTA were generated

- 15 V_H diversity groups
- 15 V_L diversity groups
- Highest diversity in CDR3H

Kineta's anti-VISTA Antibodies Bind Cyno VISTA-ECD but not Mouse VISTA-ECD

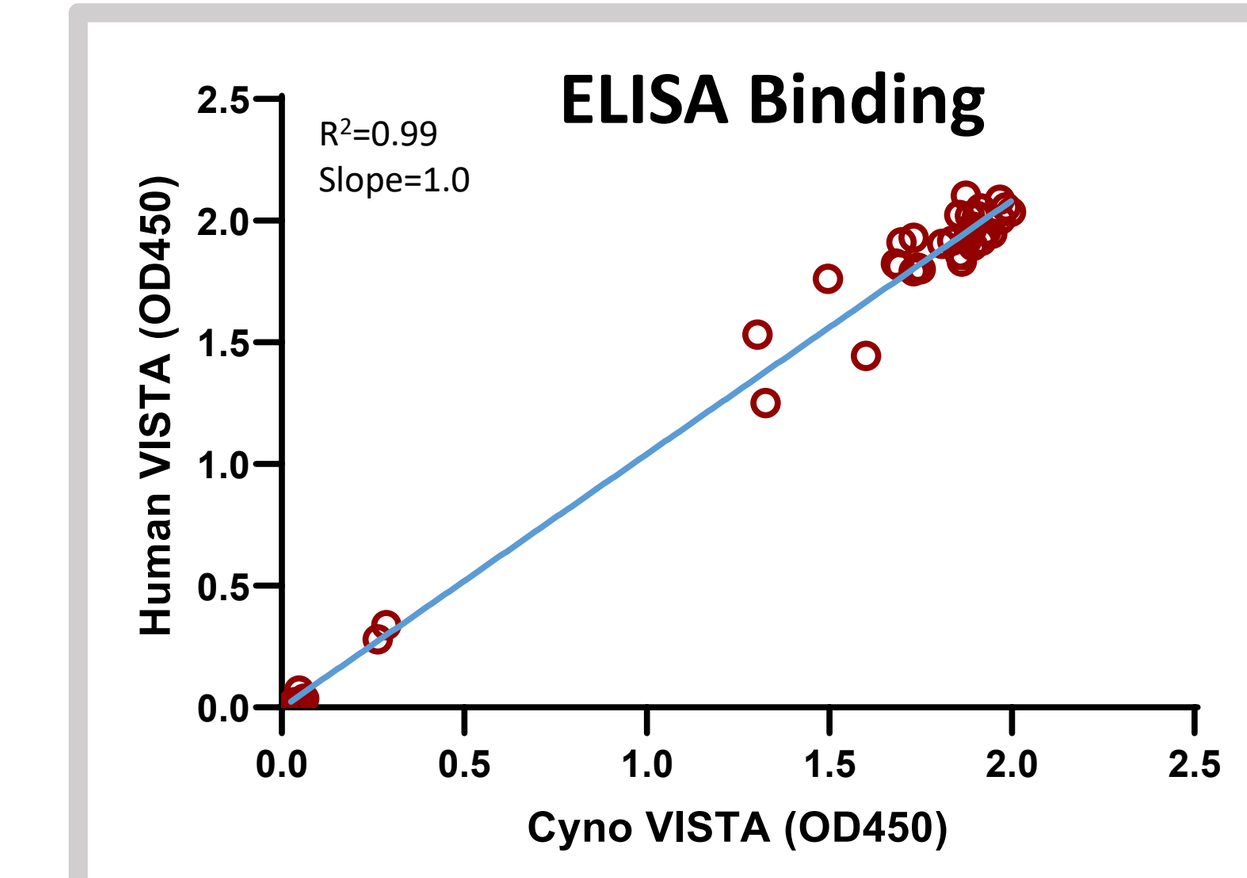
ELISA Binding
R²=0.99
Slope=1.0
Human VISTA (OD450)
Cyno VISTA (OD450)

Figure 5 : Forty-two (42) fully human monoclonal antibodies were tested for cross-species binding to human and cynomolgus monkey VISTA. Scatter plot of log mean OD_{450} values of plate-bound human or cyno VISTA ECD (1mcg/mL) with anti-VISTA antibodies (2mcg/mL) are represented.

Kineta's anti-VISTA Antibodies are Highly Potent

ELISA Binding

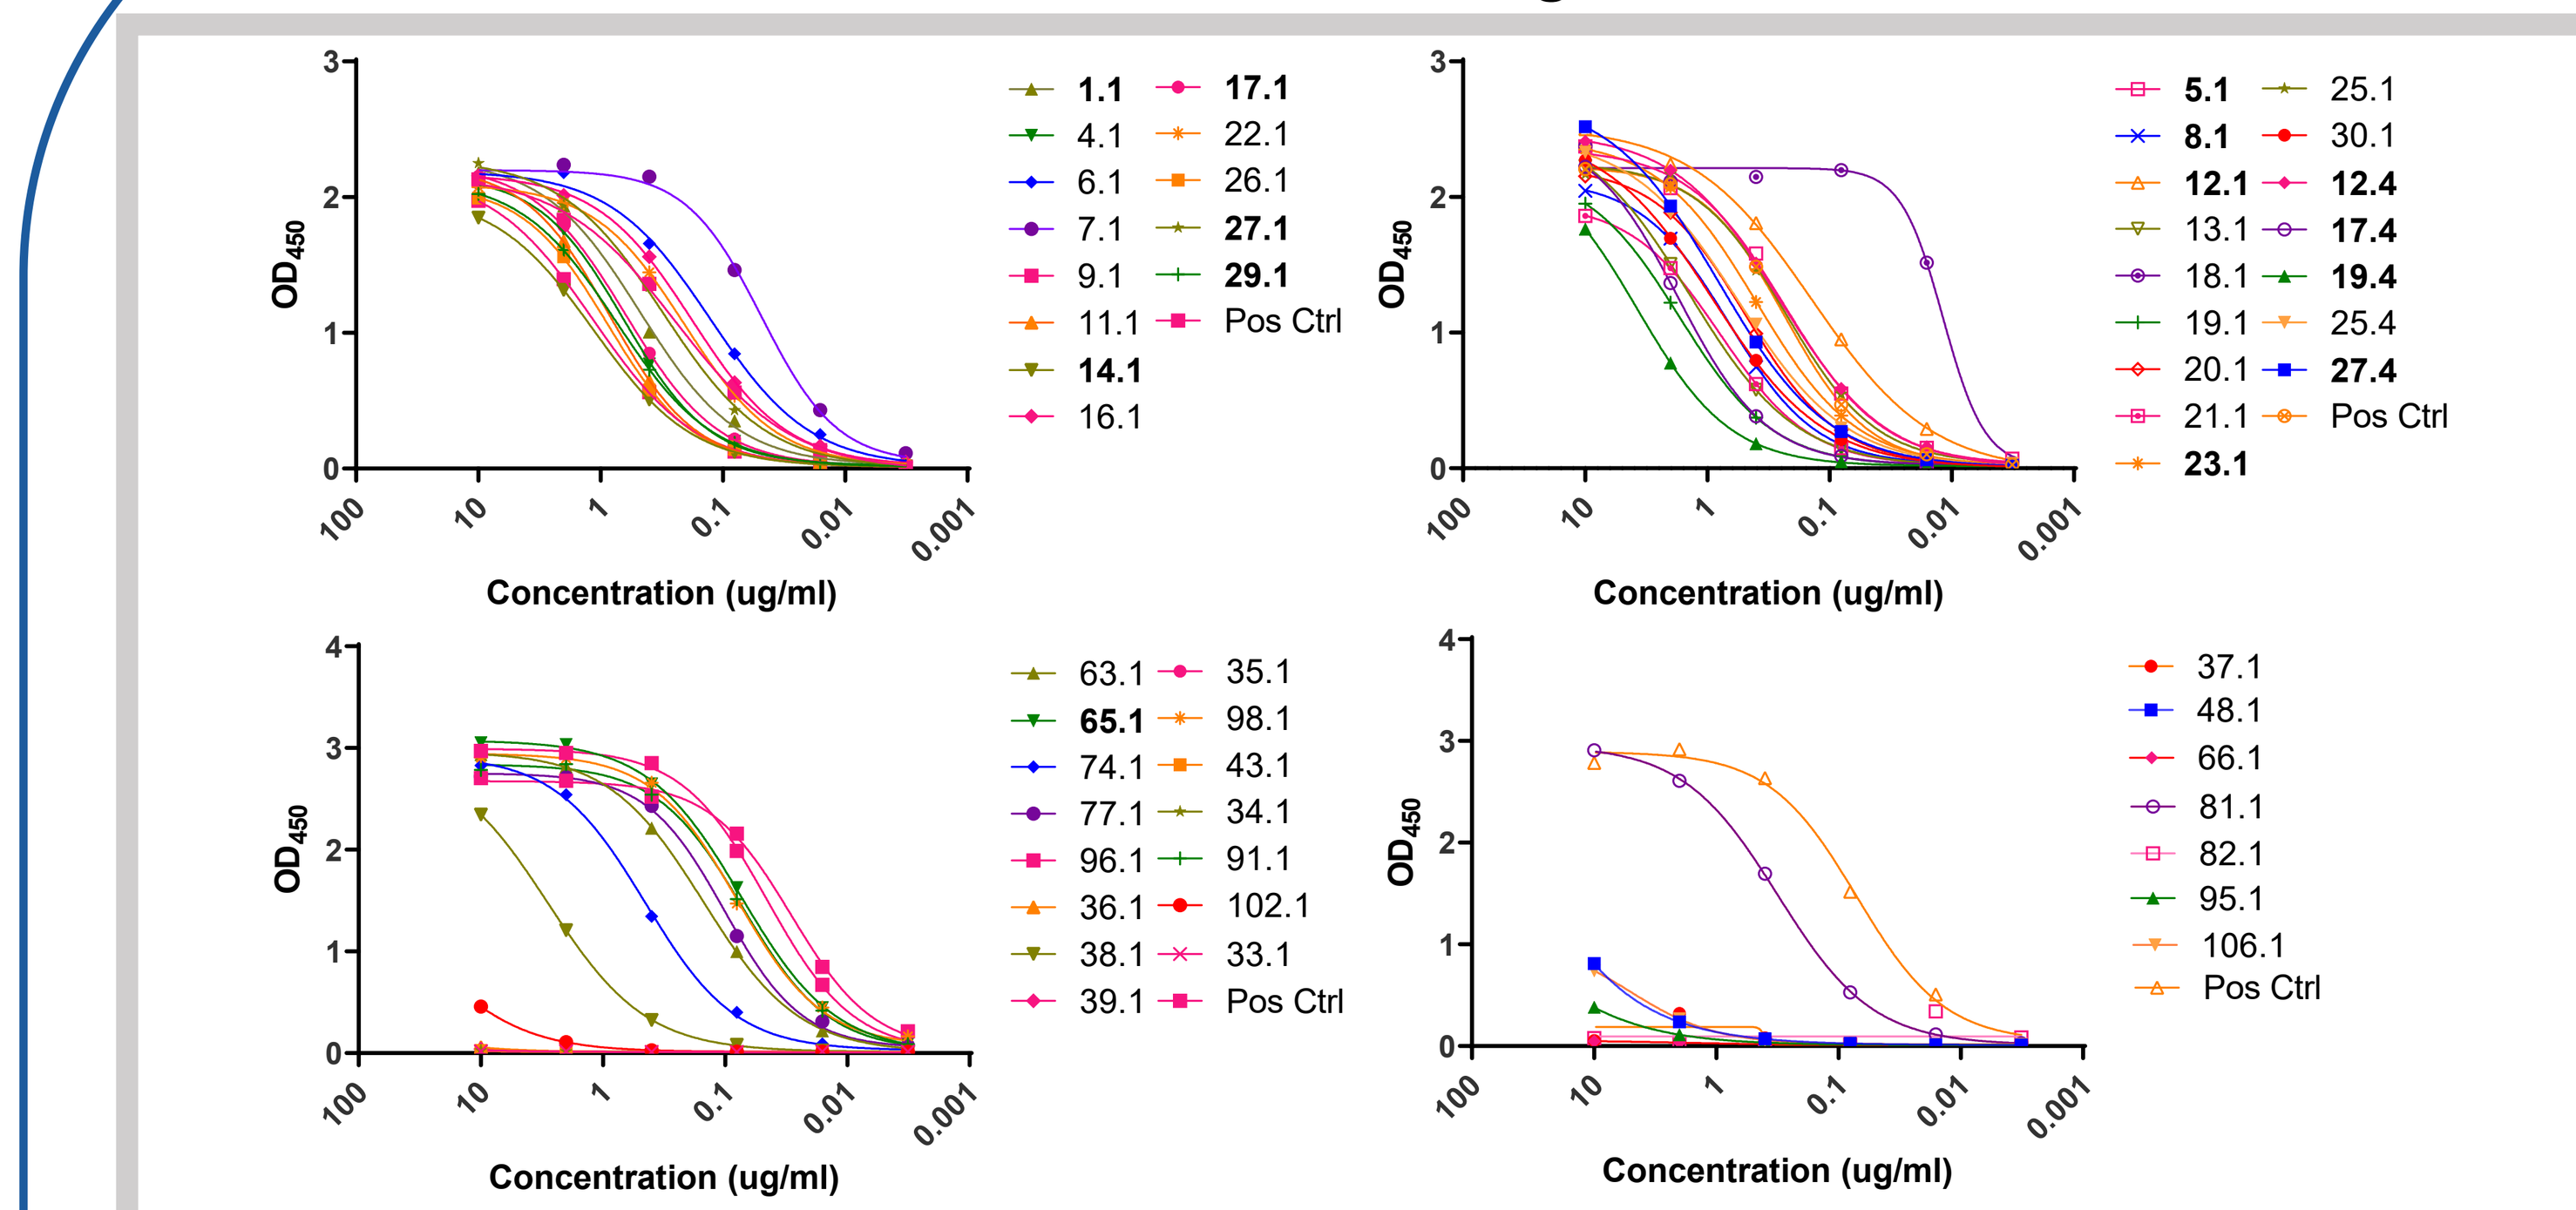
OD450
Concentration (ug/ml)
Pos Ctrl

Figure 2 : Anti-VISTA antibodies were evaluated for binding to human VISTA-ECD. Antibodies were tittered (10 mcg/ml – 0.003 mcg/ml, 67 – 0.020 nM) on a nickel ELISA plate coated with 50 ng/well human VISTA-ECD (his-tag). Biotinylated anti-human IgG(H+L) followed by HRP-streptavidin were used to detect the anti-VISTA antibodies. Plot of OD_{450} values of anti-VISTA antibodies.

Loading Sample ID	Conc. (nM)	KD (nM)	k_{on}(1/Ms)	k_{dis}(1/s)
KVA1.1 at 25 ug/mL Scouting	50	2.9	1.3E+05	3.8E-04
KVA5.1 at 25 ug/ml Full kinetics	125-15.6	4.0	1.1E+05	4.3E-04
Scouting	50	2.4	9.6E+04	2.3E-04
KVA8.1 at 25 ug/mL Scouting	50	87.5	4.3E+04	3.8E-03
KVA12.1 at 25 ug/ml Full kinetics	125-15.6	2.2	1.4E+05	3.1E-04
Scouting	50	1.9	1.7E+05	3.2E-04
KVA14.1 at 25 ug/mL Scouting	50	19.0	1.3E+05	2.5E-03
KVA17.1 at 25 ug/ml Full kinetics	250-15.6	1.7	1.5E+05	2.5E-04
Scouting	50	2.8	1.0E+05	2.9E-04
KVA23.1 at 25 ug/ml Full kinetics	125-15.6	2.0	1.4E+05	2.7E-04
Scouting	50	1.5	1.5E+05	2.1E-04
KVA27.1 at 25 ug/ml Full kinetics	250-15.6	0.7	1.7E+05	1.2E-04
Scouting	50	1.3	1.7E+05	2.2E-04
KVA29.1 at 25 ug/mL Scouting	50	**5.2**	**9.7E+04**	**5.0E-04**
KVA65.1 at 25 ug/mL Scouting	50	2.2	2.3E+05	4.9E-04

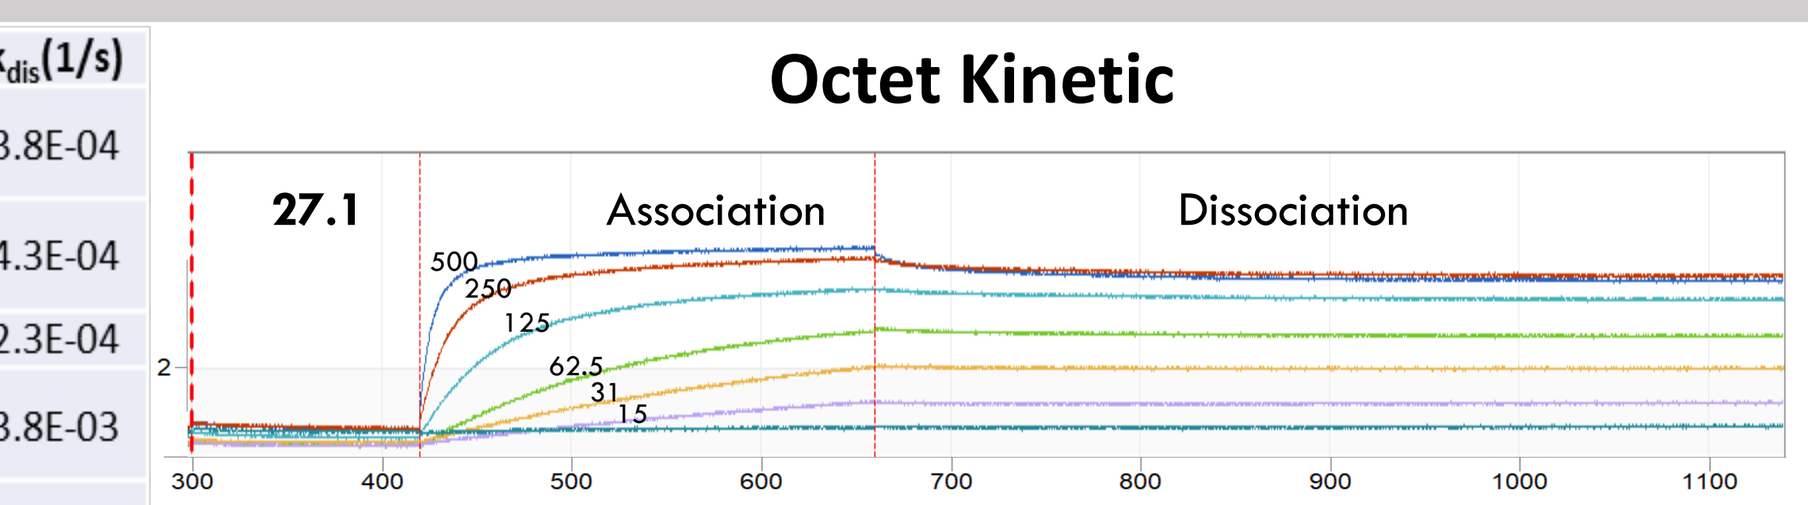
Octet Kinetic
27.1
Association
Dissociation

Figure 3 : Kinetic binding studies were performed as follows: the purified hu.Abs were captured at 25 mcg/ml using Anti-human IgG Fc Capture (AHC) dip and read biosensors. Sensors were dipped human or cyno VISTA-ECD in assay buffer (PBS, pH7.4) at 500, 250, 125, 62.5 31.3, 15.6 and 0 nM. Probes were dipped into assay buffer and the dissociation rate was measured. 1:1 curve fitting analysis was performed to determine k_{on}, k_{off}, K_D. N=1 each. Scouting data at 50 nM are included.

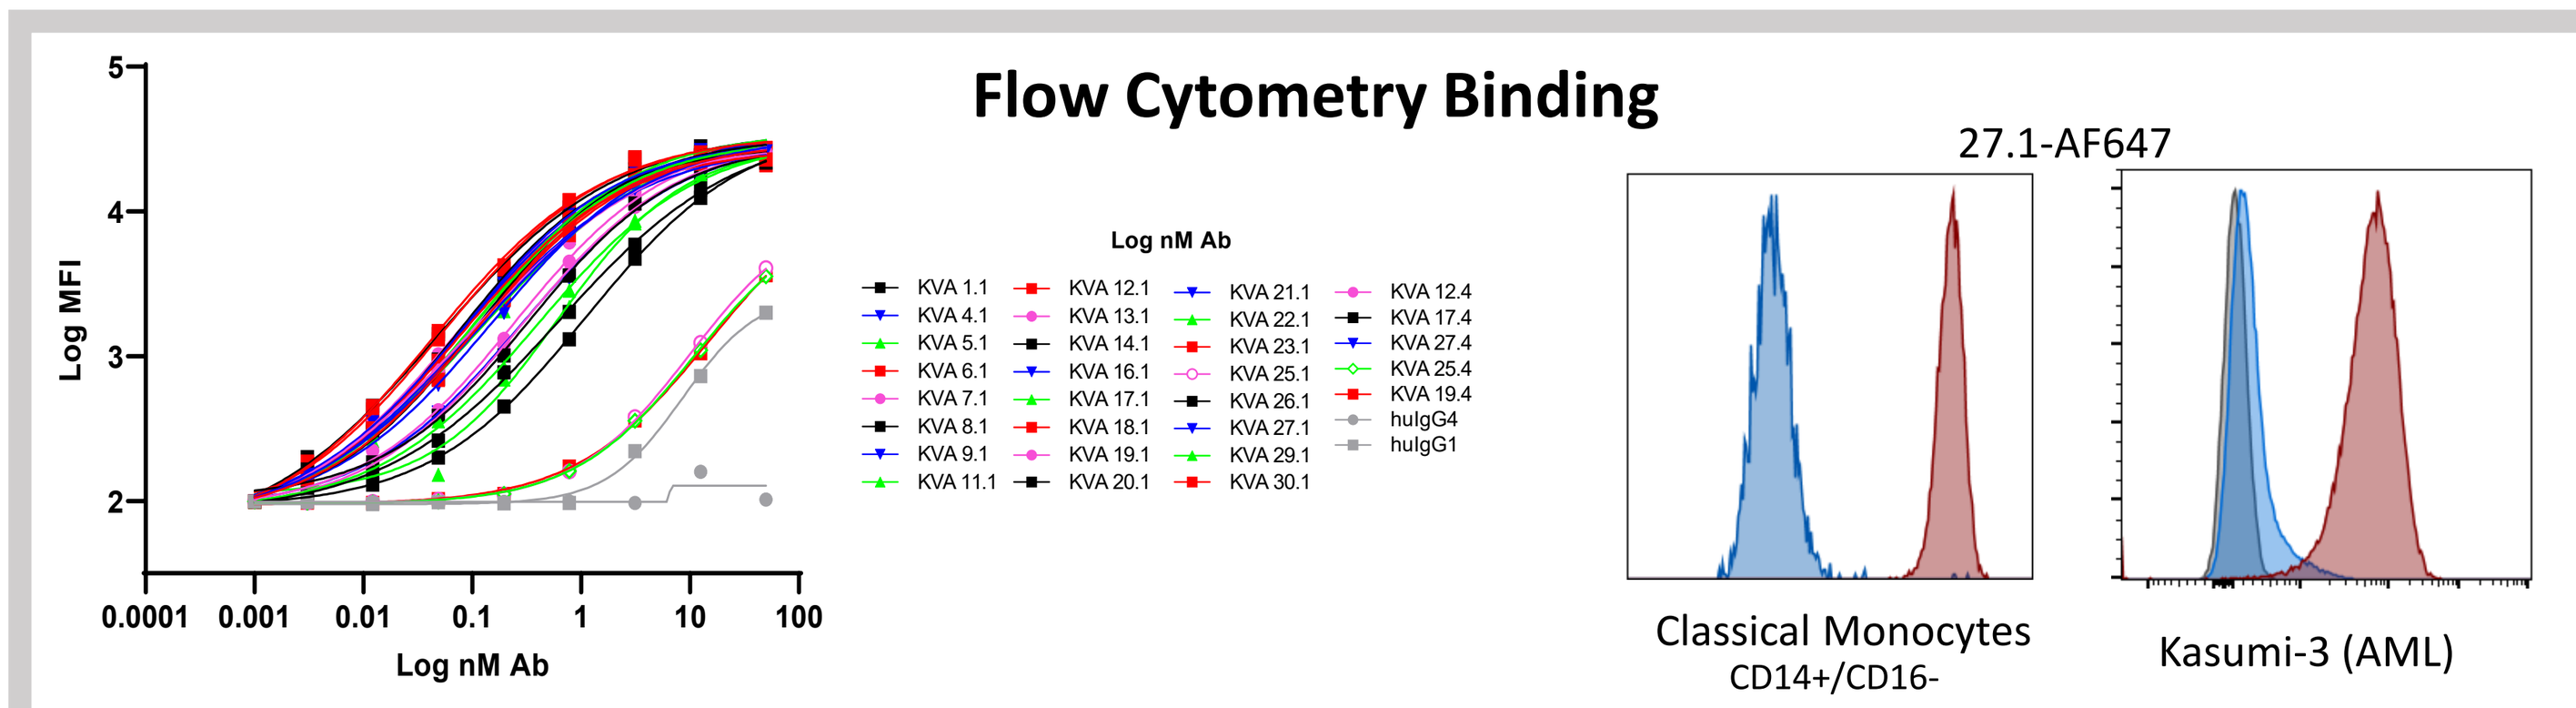
Flow Cytometry Binding
Log MFI
Log nM Ab
27.1-AF647
Classical Monocytes CD14+/CD16-
Kasumi-3 (AML)

Figure 4: Dose dependent binding of Kineta's mAbs to CHO cell surface expressed human and cyno VISTA by flow cytometry (Left). Prototypic Kineta's mAb binding on Human Classical Monocytes as well as Kasumi-3 cell line (Right).

VISTA Antagonist Antibody Induces Strong Anti-tumor Response as a Single Agent or in Combo-therapies

CT26 mouse model

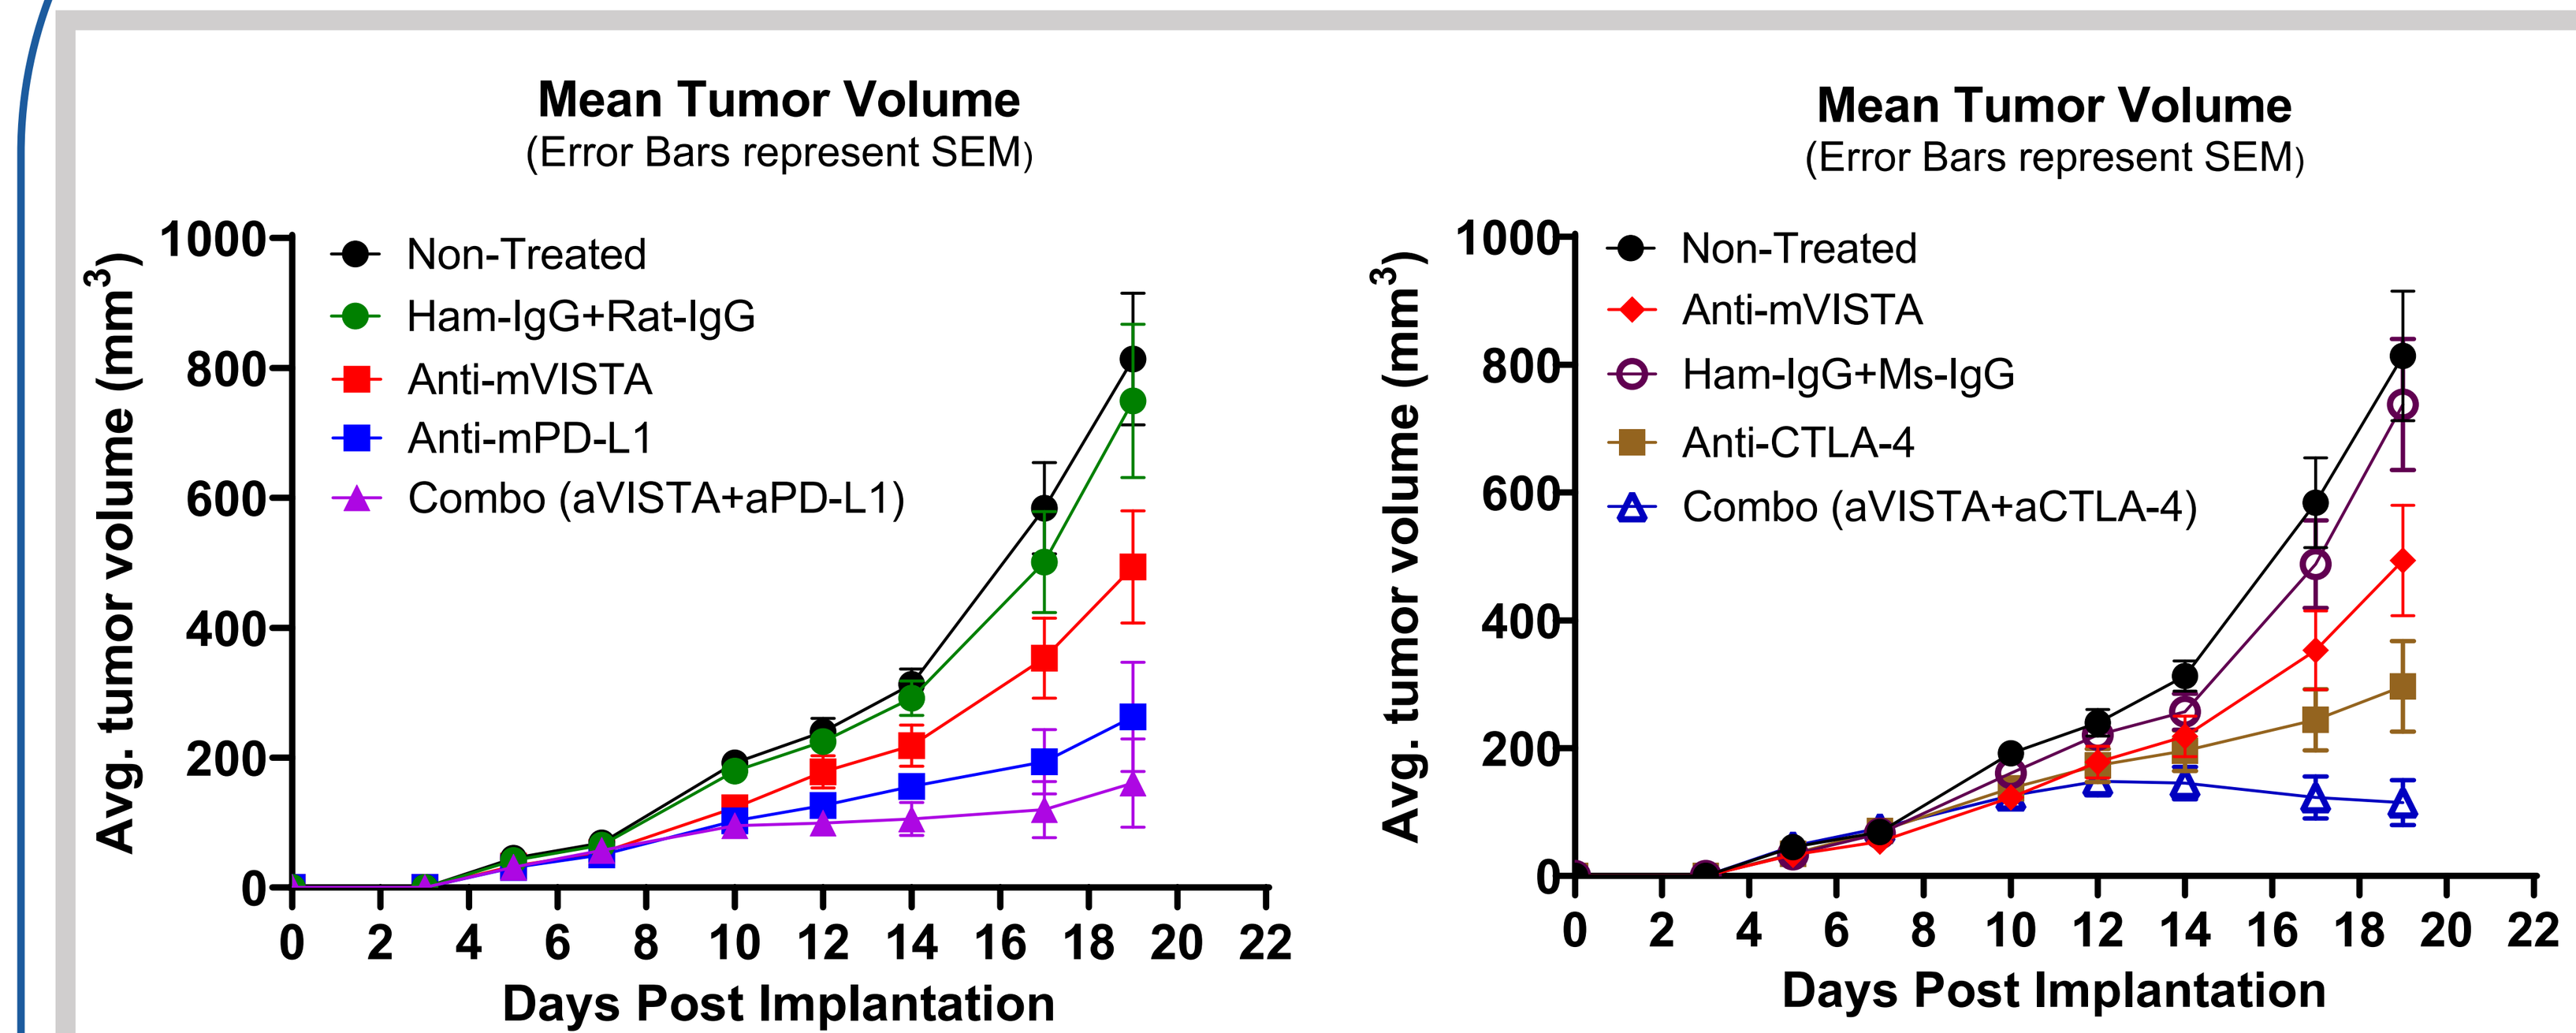
Mean Tumor Volume
(Error Bars represent SEM)
Avg. tumor volume (mm³)
Days Post Implantation
Non-Treated
Ham-IgG+Rat-IgG
Anti-mVISTA
Anti-mPD-L1
Combo (aVISTA+aPD-L1)
Ham-IgG+Ms-IgG
Anti-CTLA-4
Combo (aVISTA+aCTLA-4)

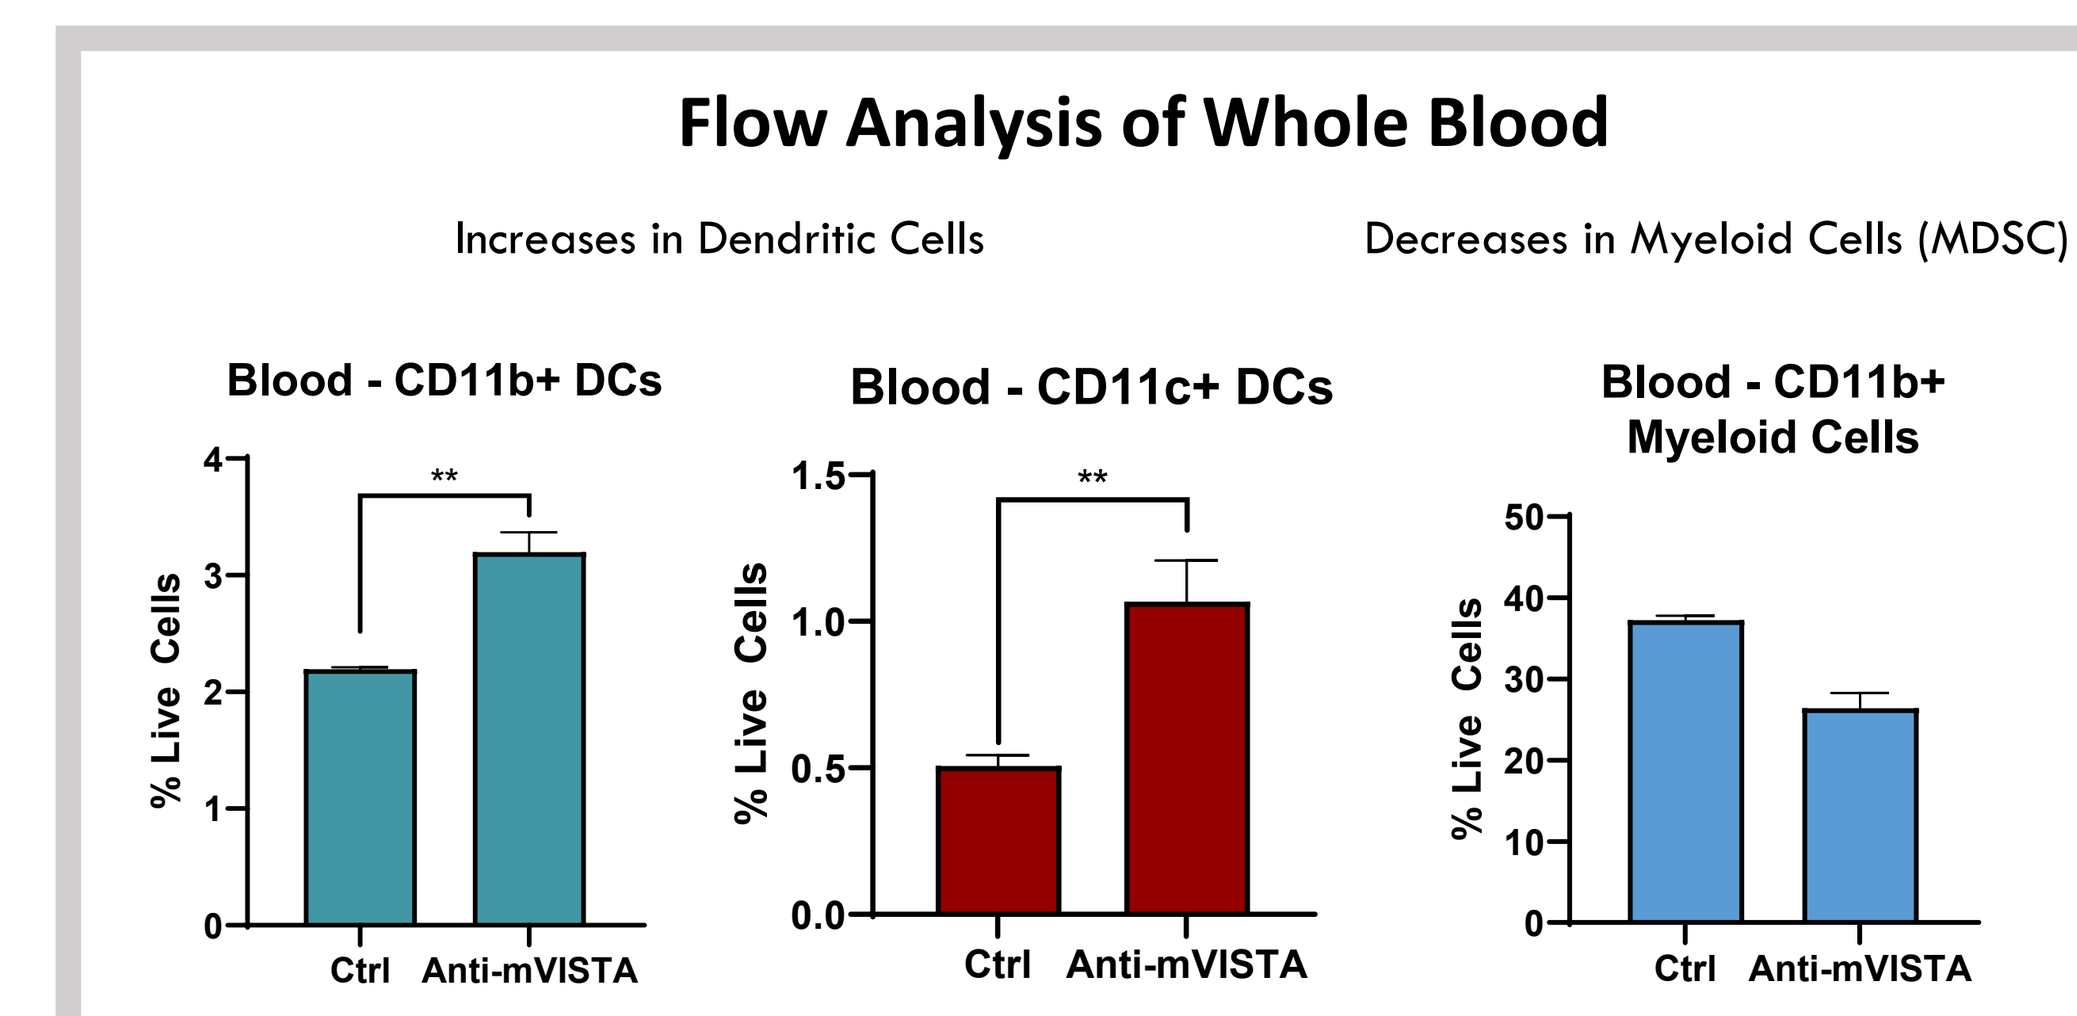
Flow Analysis of Whole Blood
Increases in Dendritic Cells
Decreases in Myeloid Cells (MDSC)
Blood - CD11b+ DCs
Blood - CD11c+ DCs
Blood - CD11b+ Myeloid Cells
% Live Cells
Ctrl
Anti-mVISTA

Figure 6 : CT26 Tumor Model. 5.E5 CT26 cells were inoculated in PBS subcutaneously to female BALB/c mice. Tumor volume (L x W^2 x 0.5) and bodyweight were measured 3X/wk. Beginning on Day 3, animals were treated by IP injection of 200 mcg control antibodies (hamster + rat or mouse isotype ctrl), anti-mVISTA (13F3, hamster) and/or anti-mPD-L1 or anti-CTLA-4 3X/wk for 3 weeks. Mean tumor volume is plotted. Whole blood was collected for immunophenotyping on Day 14 by flow cytometry for the following immune populations: CD45, CD3, CD4, CD8, CD11b, CD11c, and NKp46.

Conclusions

- **107 fully human ScFv anti-VISTA antibodies were generated and analyzed**
- **Kineta's anti-VISTA antibodies carry unique sequences and the selected lead candidates exhibit potencies in the subnanomolar range**
- **Kineta's human anti-VISTA antibodies are highly specific and cross-react with Cyno-VISTA but not Mouse-VISTA**
- **VISTA antagonist antibody induces strong anti-tumor response as a single agent or in combo-therapies with anti-PDL1 or anti-CTLA4 in CT26 tumor models**
- **VISTA antagonist antibody activates Dendritic Cells in the blood and reduces MDSCs**